Increased FNDC5 (irisin) expression in subcutaneous adipose tissue in obese patients with type 2 diabetes one year after bariatric surgery.

Julia Muenzker[1], Alexandra Harger[1], Alexandru Tuca[1], Lisa Lindheim[1], Verena Zachhuber[1], Eva Svehlikova[1], Oana Freisinger[2], Johanna Fruhmann[2], Friedrich Tadler[3], Thomas R. Pieber[1], Barbara Obermayer-Pietsch[1]

[1] Clinical Department for Endocrinology and Metabolism, Medical University of Graz, Austria

[2] Clinical Department of Surgery, Medical University of Graz, Austria

[3] Clinical Department of Surgery, Krankenhaus der Elisabethinen, Graz, Austria

Background & Aim

Gastric bypass surgery improves glycaemic control, but the underlying mechanisms are incompletely understood. The aim of this study was to investigate potentially causal or associated alterations in gene expression of adipokines, myokines and hormones in the subcutaneous and visceral adipose tissue of diabetic and non-diabetic obese subjects before and after gastric bypass.

Materials & Methods

Biopsy specimens from the adipose tissue of 12 diabetic and 12 non-diabetic patients at (subcutaneous, visceral) and one year after (subcutaneous) gastric bypass were analyzed with quantitative real-time PCR for the expression levels of various adipokines, myokines and hormones such as adiponectin, leptin, irisin (FNDC5), osteocalcin and osteopontin.

Study Work Plan

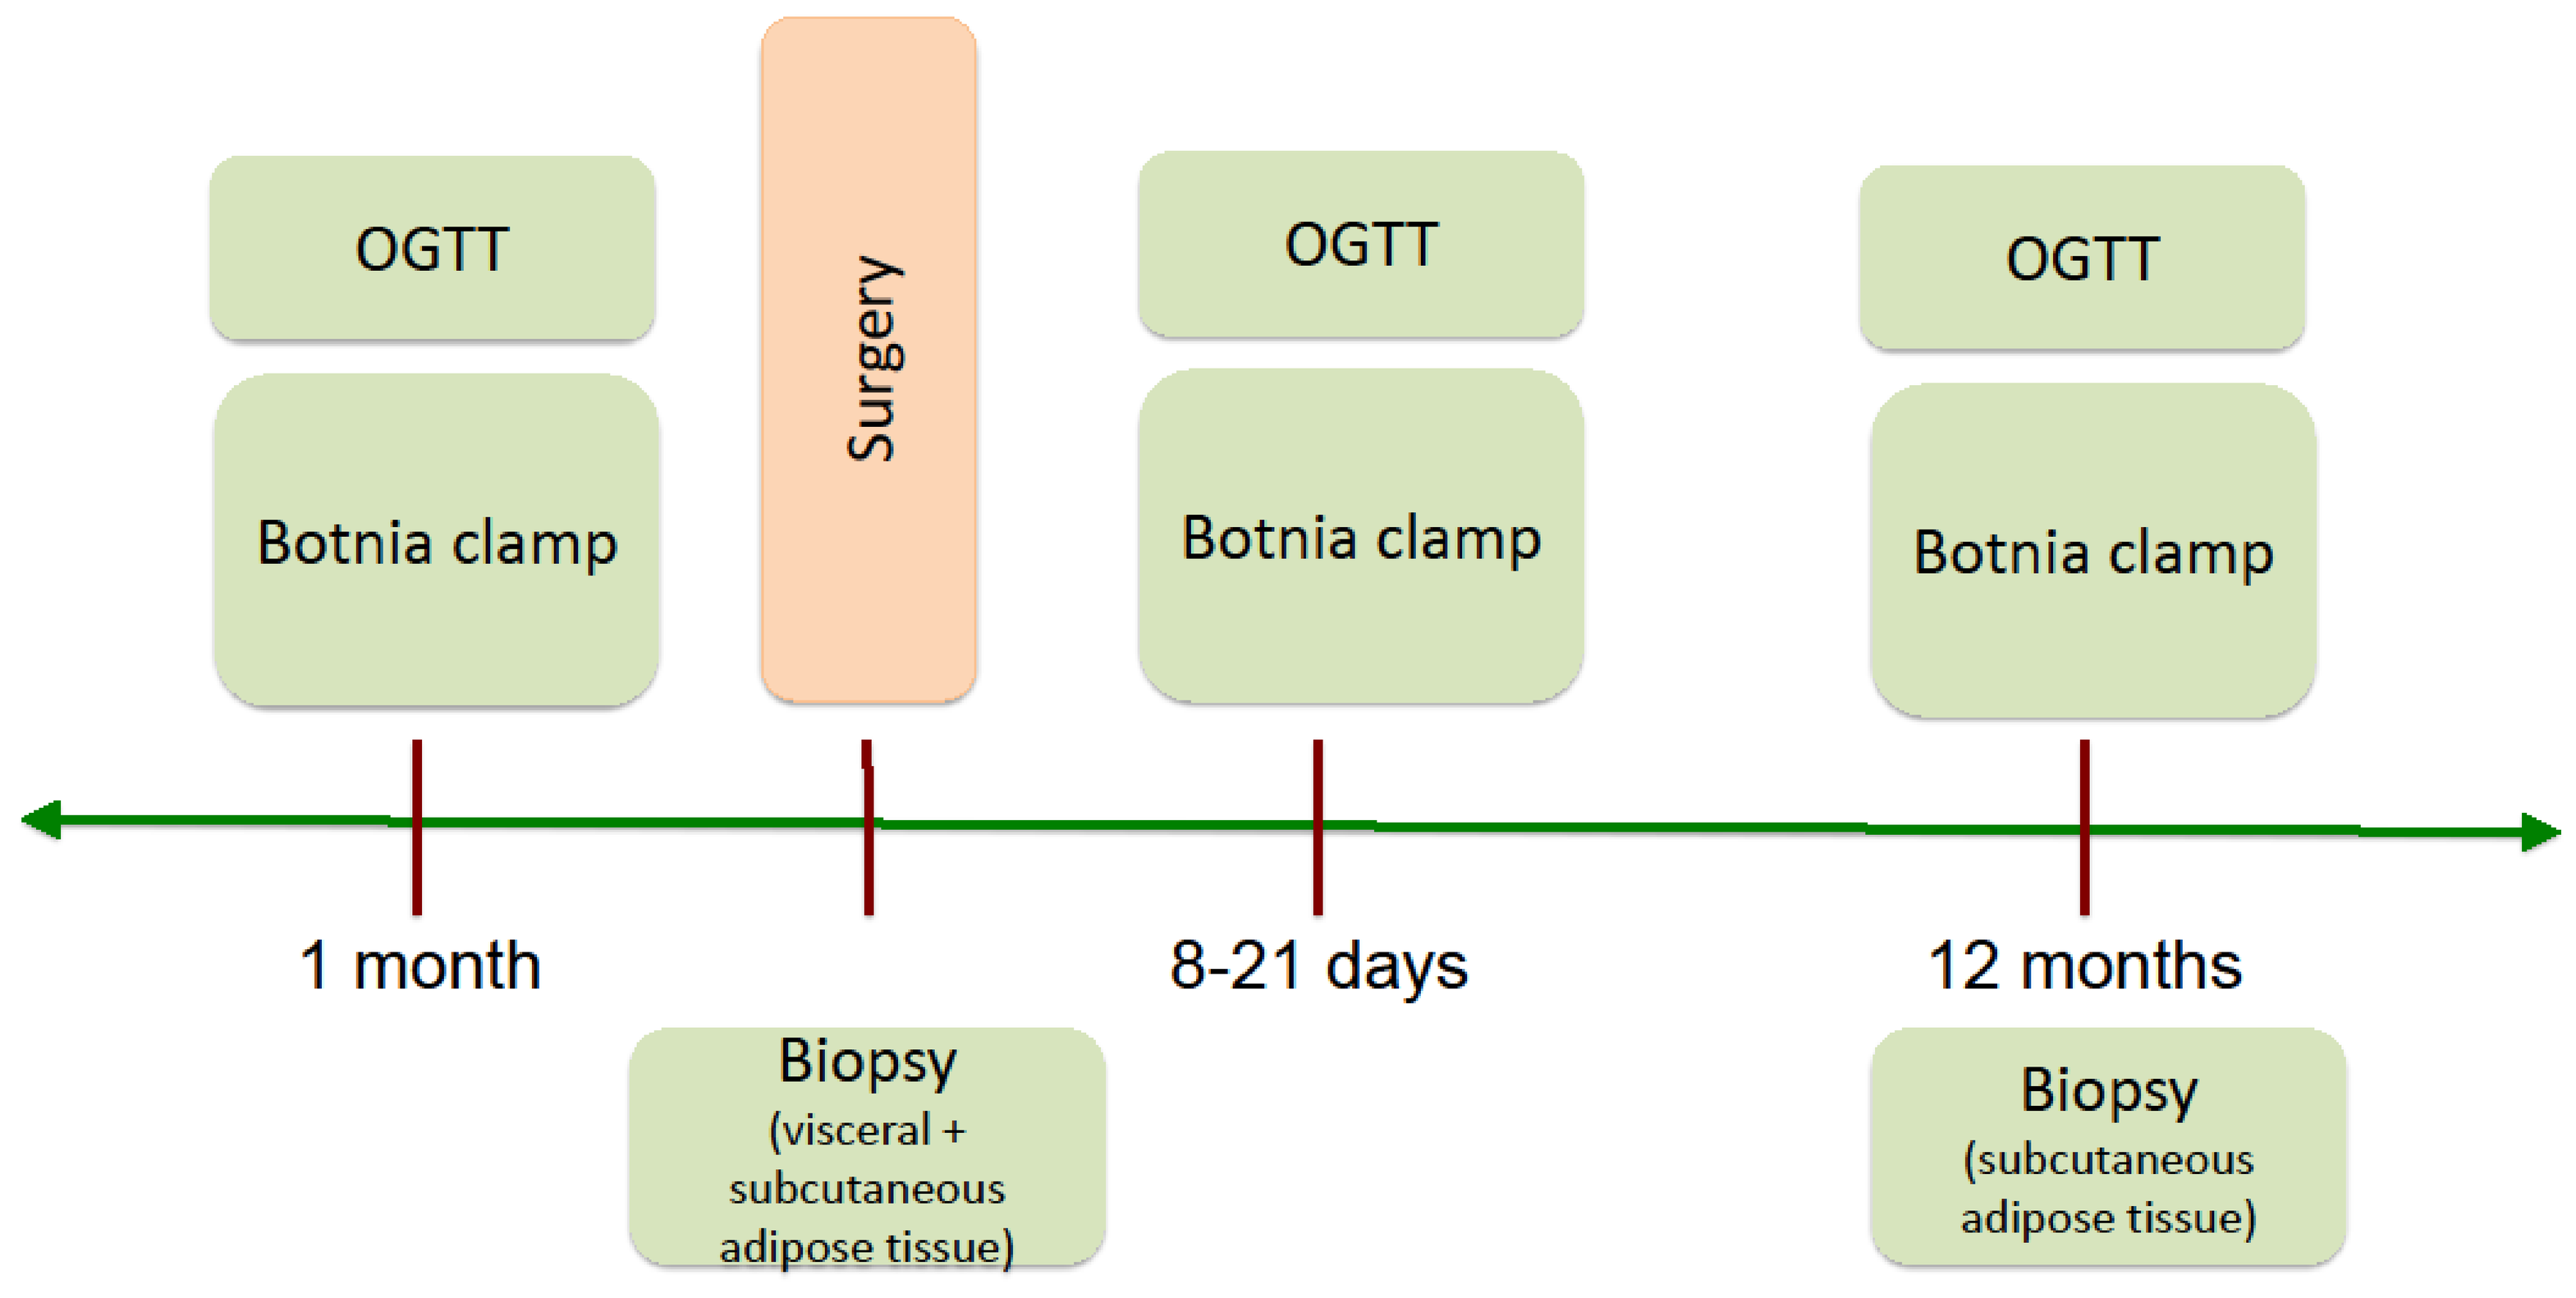
OGTT
Botnia clamp
Surgery
OGTT
Botnia clamp
OGTT
Botnia clamp
1 month
8-21 days
12 months
Biopsy
(visceral + subcutaneous adipose tissue)
Biopsy
(subcutaneous adipose tissue)

OGTT: oral glucose tolerance test; 180 min, 9 sampling points (assessment of glucose, insulin, C-peptide, proinsulin)

Botnia clamp: IVGTT (intravenous glucose tolerance test; 60 min) and subsequent hyperinsulinaemic- euglycaemic clamp (120 min)

	Diabetics			Non-Diabetics		
	pre-OP	1 year postsurgery	p-value	pre-OP	1 year postsurgery	p-value
Age (years)	48 ± 11			46 ± 8		
Weight (kg)	129.63 ± 18.52	90.93 ± 16.04	<0.001	125.17 ± 18.08	88.17 ± 14.15	<0.001
BMI (kg/m2)	46.06 ± 5.57	32.32 ± 5.28	<0.001	44.07 ± 5.26	30.81 ± 4.19	<0.001
HbA1c (%)	6.98 ± 1.25	5.98 ± 0.78	0.02	5.44 ± 0.24	5.24 ± 0.25	0.02
Fasting glucose (mmol/l)	7.65 ± 1.65	5.57 ± 1.36	<0.001	5.15 ± 0.73	5.40 ± 1.02	<0.001
Fasting c-peptide (ng/ml)	2.98 ± 0.85	1.48 ± 0.43	<0.001	2.28 ± 0.85	1.07 ± 0.29	<0.001
Fasting insulin (mmol/l)	16.77 ± 10.87	4.67 ± 2.85	<0.001	9.76 ± 3.30	3.03 ± 2.24	<0.001
Fasting proinsulin (pmol/l)	33.06 ± 18.93	6.59 ± 2.56	<0.001	17.65 ± 11.26	4.10 ± 1.60	0.04
HOMA	5.47 ± 3.27	1.41 ± 1.10	0.03	2.28 ± 0.95	0.65 ± 0.57	0.16
Insulin Sensitivity Index (ISI)	-0.01 ± 0.03	0.07 ± 0.03	<0.001	0.04 ± 0.02	0.10 ± 0.01	<0.001
Early Insulin Response (EIR)	0.17 ± 0.17	0.28 ± 0.14	<0.001	0.56 ± 0.24	0.67 ± 0.34	<0.001
Diabetes duration (years)	8.3 ± 6.32					

Clinical characteristics of patients at time of bariatric surgery and 1 year afterwards. Data are expressed as mean SD.

Results

Non-diabetic subjects were found to express significantly higher levels of FNDC5 in the subcutaneous adipose tissue than diabetic subjects before gastric bypass ($p = 0.0167$). FNDC5 expression in the subcutaneous adipose tissue of diabetic patients was significantly increased one year after surgery ($p < 0.001$). In non-diabetic subjects, there was no significant increase observable ($p = 0.158$). The increase in FNDC5 in diabetic subjects correlated with reduction in BMI (Pearson's $r = 0.718$, $p = 0.009$). No correlation was observed between FNDC5 expression and early insulin response or insulin sensitivity index.

A

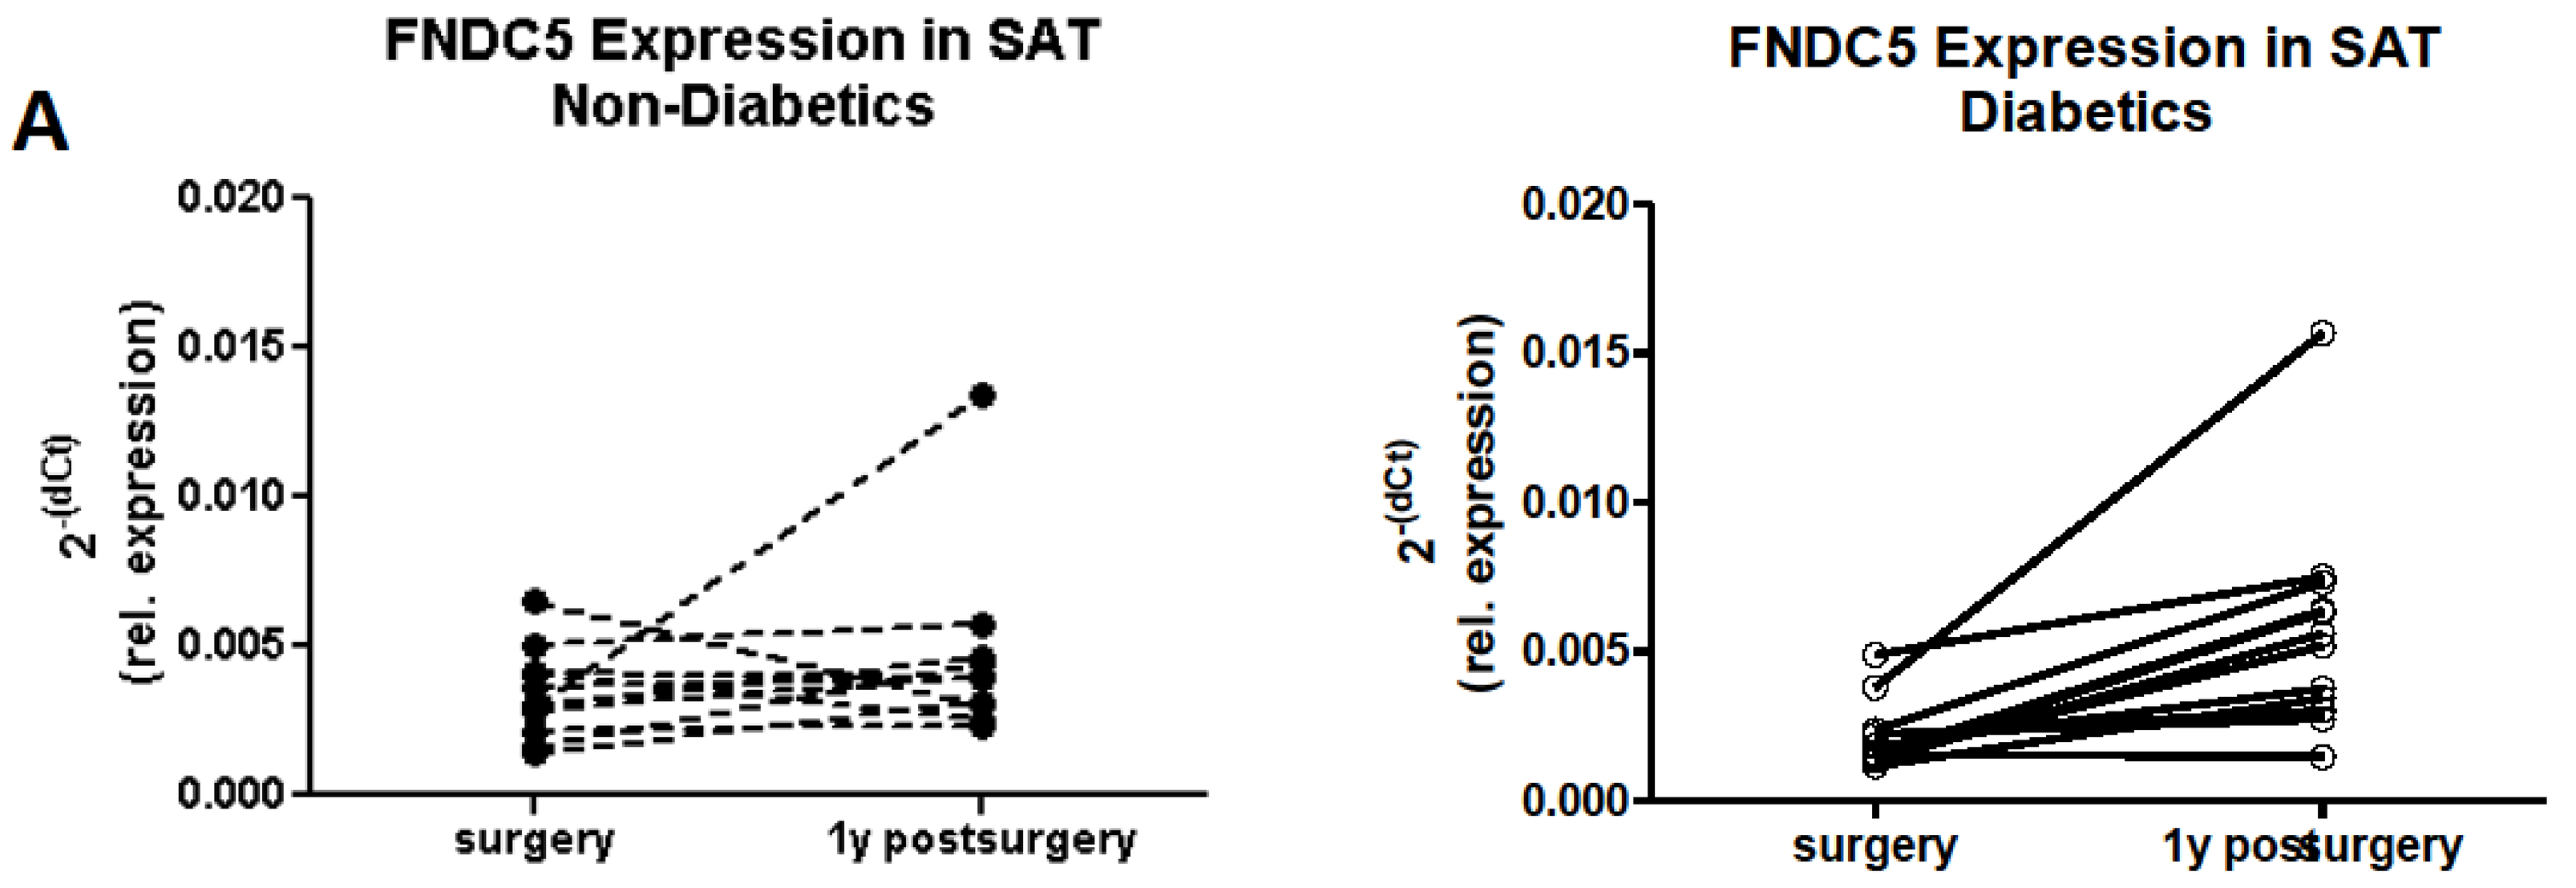
FNDC5 Expression in SAT
Non-Diabetics
FNDC5 Expression in SAT
Diabetics
2^-(dCt) (rel. expression)
0.020
0.015
0.010
0.005
0.000
surgery
1y postsurgery

		OP	post-OP	p-value
FNDC5 (Irisin)	T2D	0,00170	0,00541	**0,001***
	non-T2D	0,00332	0,00394	0,11

B

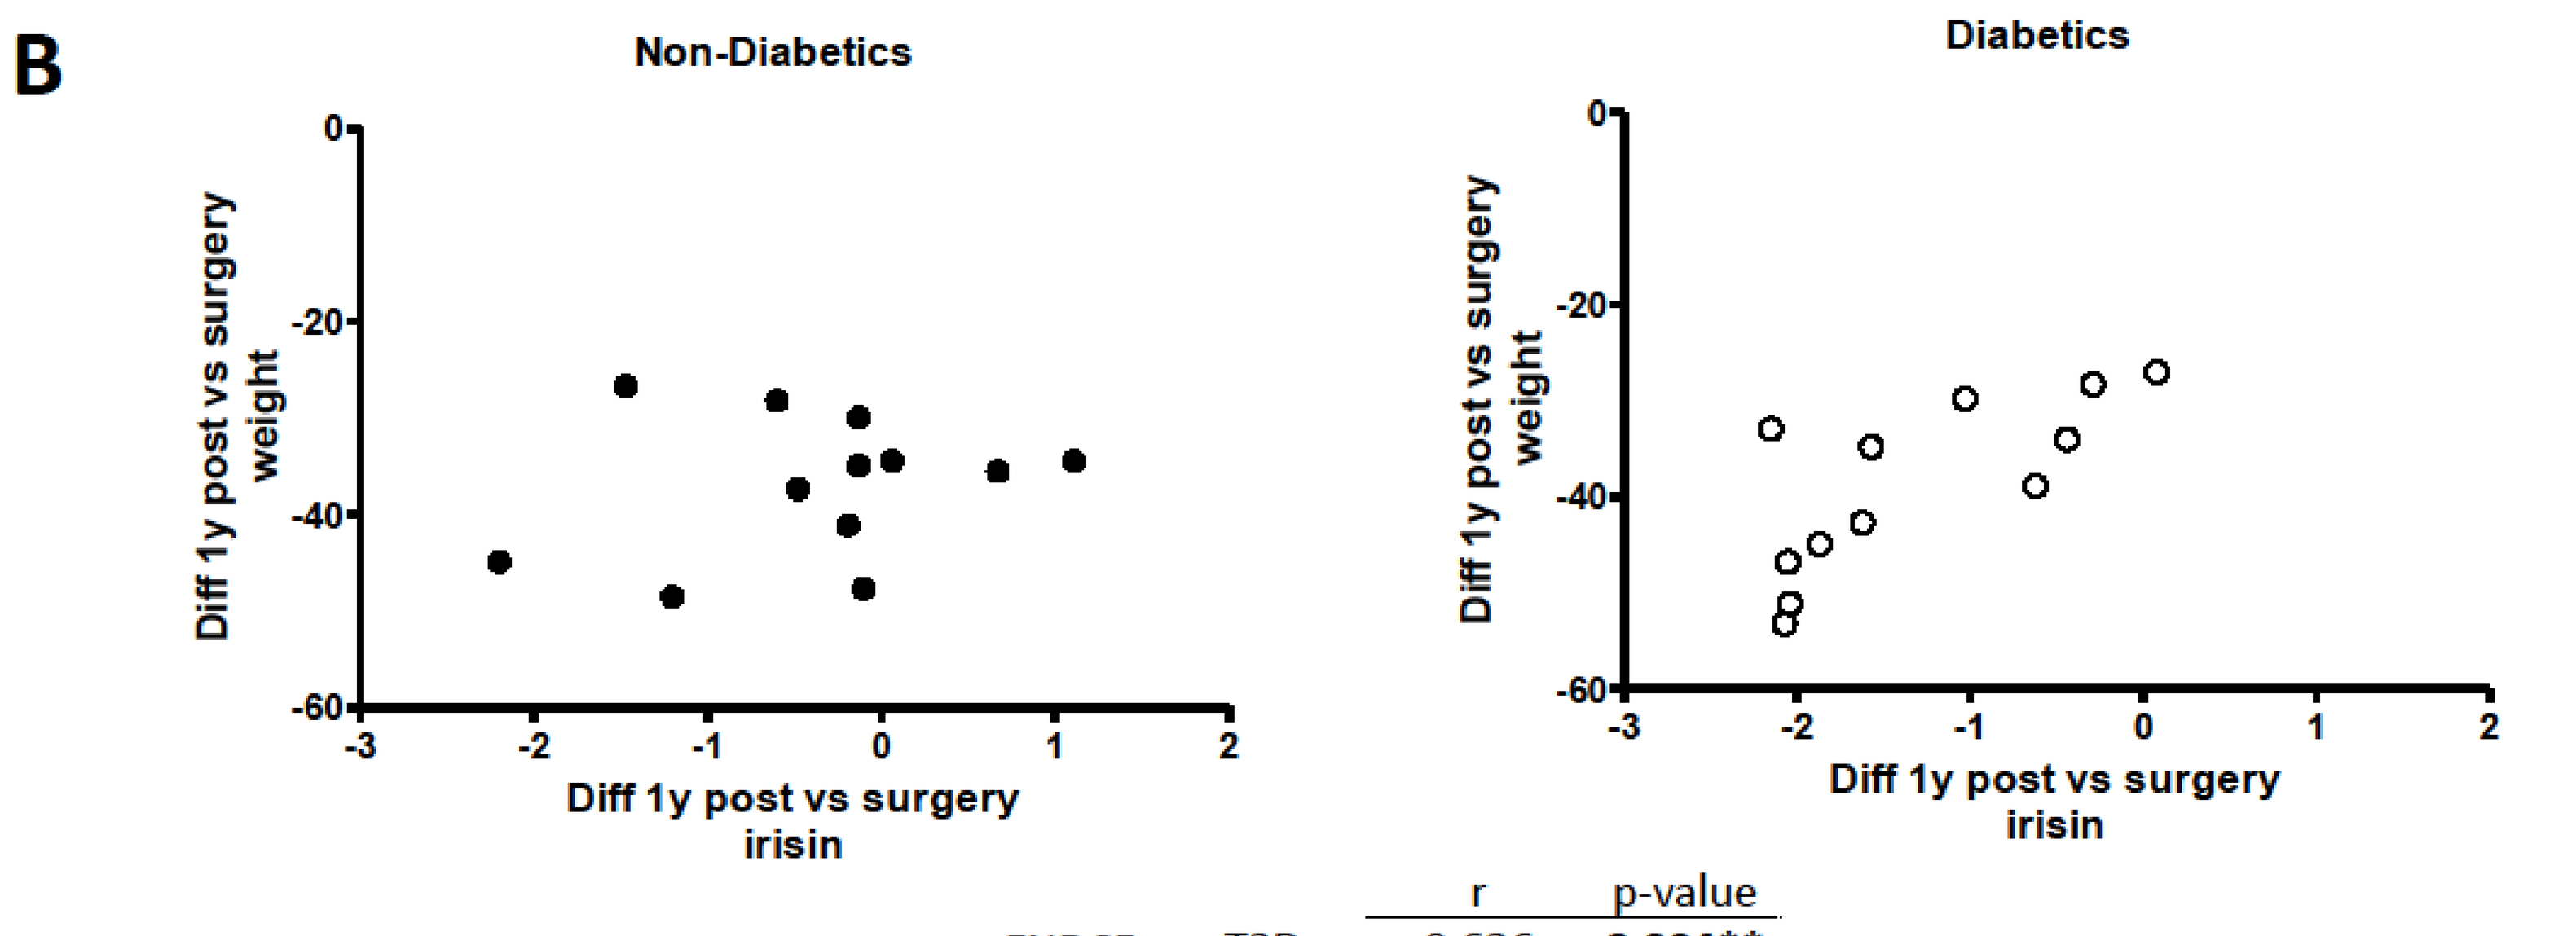
Non-Diabetics
Diabetics
Diff 1y post vs surgery weight
0
-20
-40
-60
-3
-2
-1
0
1
2
Diff 1y post vs surgery irisin

		r	p-value
FNDC5 (Irisin)	T2D	0,636	**0,004****
	non-T2D	0,076	0,731

FNDC5 expresion in subcutaneous adipose tissue (SAT) at time of surgery and 1 year postsurgery (A), and correlation of weight reduction with difference in FNDC5 expression levels before and after surgery (B) .

Conclusion

To our knowledge, this is the first study to investigate the role of the novel myokine irisin in bariatric surgery mediated weight loss. The results of this study demonstrate that exercise-independent weight loss increases (FNDC5) irisin expression levels in subcutaneous adipose tissue of diabetic subjects.

Acknowledgements

Contact:
julia.muenzker@medunigraz.at

Supported by
EFSD/MSD Clinical Research Grant 2009
Clinical Trial Registration: NCT 01271062

17 ECE 622--EP
Obesity and cardiovascular endocrinology
Julia Muenzker
DOI: 10.3252/pso.eu.17ece.2015
Poster presented at: ECE2015 16-20 May 2015 DUBLIN, IRELAND 17th EUROPEAN CONGRESS OF ENDOCRINOLOGY