IN VIVO CHARACTERISATION OF SKELETAL MUSCLE METABOLISM IN GROWTH HORMONE DEFICIENT ADULTS USING ^{31}P MAGNETIC RESONANCE SPECTROSCOPY

BES 2013, Harrogate

Akash Sinha[1,2], Steve Ball[1,3], Kieren Hollingsworth[1], Tim Cheetham[1,2]

Newcastle University[1], Paediatric Endocrinology NUTH[2], Endocrinology NUTH[3]

Poster No 0069

Background

- Adults with growth hormone deficiency (GHD) experience fatigue and reduced energy levels[1]. Although these symptoms improve with GH replacement, their pathophysiology is poorly understood.
- Fatigue can be classified as central or peripheral in origin.
- GH parameters have been reported to be associated with skeletal muscle mitochondrial capacity in adult men[2].
- Phosphorus-31 magnetic resonance spectroscopy is a non-invasive tool used to assess skeletal muscle mitochondrial oxidative capacity and pH recovery (proton handling) *in vivo* following exercise.
- There have been no detailed investigations into the role of GH in functional muscle metabolism and its link with fatigue. Phosphorus-31 magnetic resonance spectroscopy is a technique that may provide insight into skeletal muscle pathophysiology in the GH-deficient state.

Aims

- To characterise and compare *in vivo* skeletal muscle metabolism in **untreated GHD adults** with **treated GHD adults** and **healthy controls.**
- To compare perception of fatigue using specific domains within QoL-AGHDA, a GHD specific well-being assessment tool, across the 3 groups.

Untreated GHD | Treated GHD | Healthy controls

Methods

- Cross-sectional study.
- 22 untreated GHD, 23 treated GHD & 20 healthy controls (matched for age, gender and physical activity) were recruited.
- All subjects underwent ^{31}P MRS, biochemical, auxological and body composition assessment. They also completed Qol-AGHDA questionnaires.
- REC approval obtained.
- Statistical tests as described (Minitab v16).

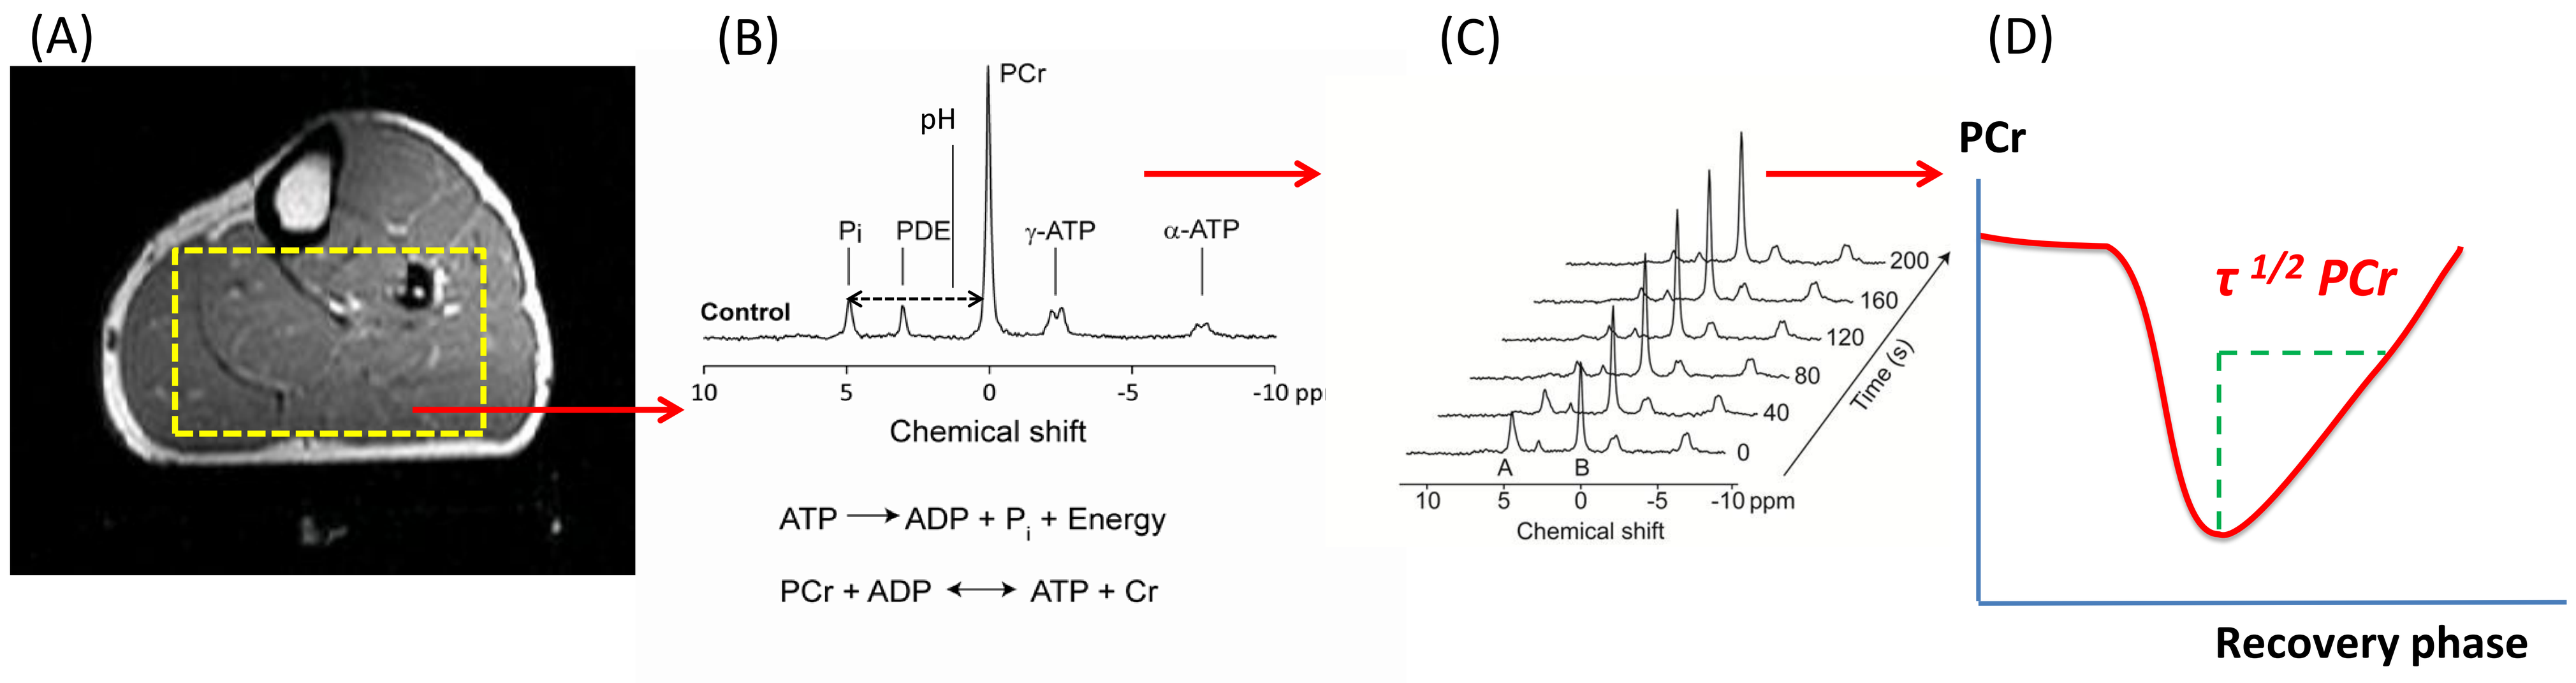
(A)
(B)
PCr
pH
Pi
PDE
γ-ATP
α-ATP
Control
Chemical shift
ATP → ADP + Pi + Energy
PCr + ADP ↔ ATP + Cr
(C)
Time (s)
Chemical shift
(D)
PCr
τ 1/2 PCr
Recovery phase

Fig 1: The phosphorus spectrum (B) from the gastro-soleus compartment (A) showing relative concentrations of (Pi)inorganic phosphate,(PCr)phosphocreatine and ATP. Serial spectra (C) were acquired following dynamic exercise during the recovery phase. Exponential fits of the recovery data (D) were made to estimate half-times for recovery to equilibrium ($\tau^{1/2}$PCr), a measure of mitochondrial oxidative phosphorylation.

Results

Mean ±SD	OFF GH	ON GH	Controls	P value
Total No	22	23	20	
Age	27.85±9.34	29.76±10.07	31.05±7.93	0.54
Sex ratio	59.2% males	65.2% males	60% males	
Aetiology of GHD:				
MPHD / Isolated GHD	17/5	20/3	NA	0.40
Childhood /Adult onset	18/4	15/8	NA	0.21
Surgery	14	9	NA	0.48
Radiotherapy	16	8	NA	0.48
Chemotherapy	6	1	NA	0.48
Endocrine parameters:				
IGF-1 (nmol/L)	14.0±11.2	38.1±19.9	30.7±9.5	<0.001
Thyroxine fT4 (pmol/L)	18.2±3.7	19.0±3.9	16.3±2.1	0.09
Testosterone (nmol/L)	20.6±9.5	15.9±7.9	20.7±5.5	0.25
25OHD (nmol/L)	57.9±34.42	53.9±20.2	44.2±29.4	0.42

Table 1: above shows the subject characteristics including aetiology and endocrine profiles

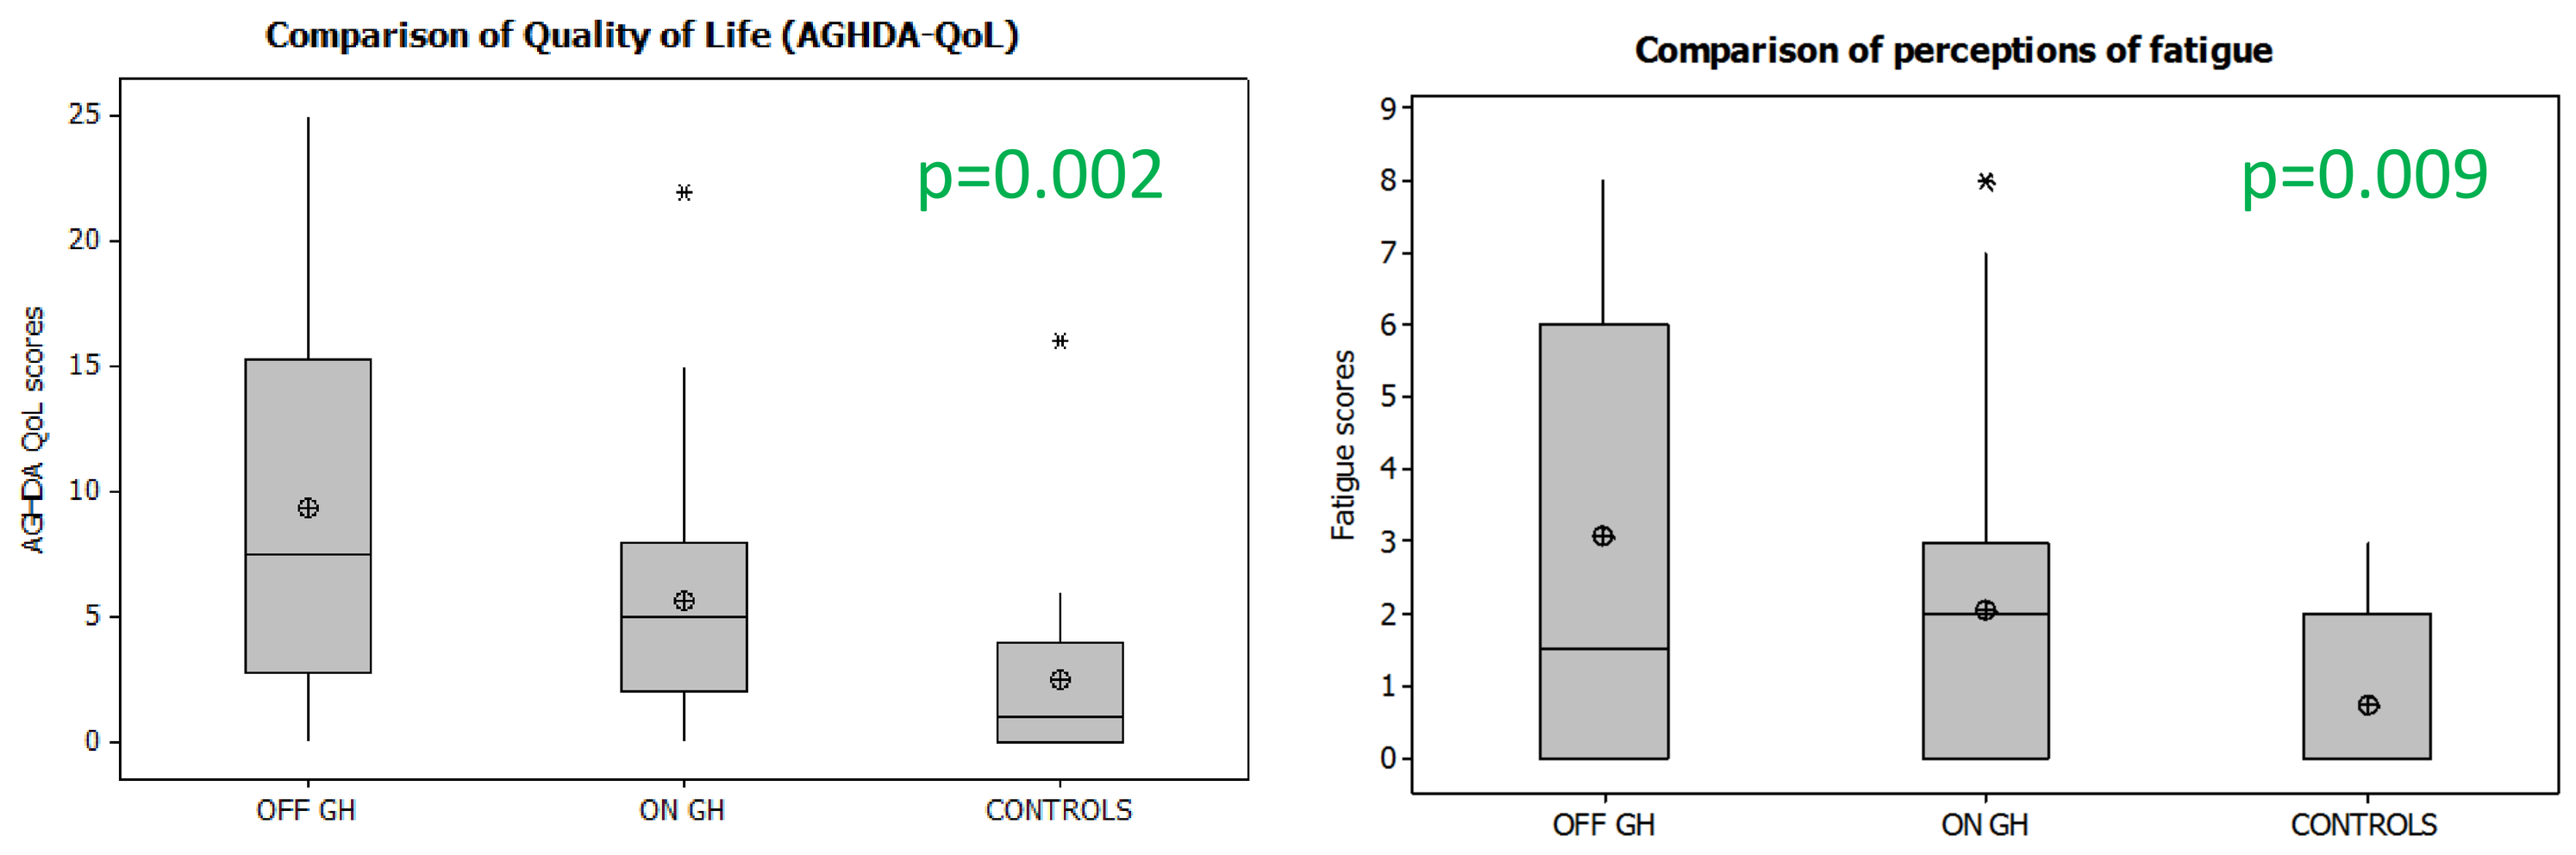
Comparison of Quality of Life (AGHDA-QoL)
AGHDA QoL scores
p=0.002
OFF GH
ON GH
CONTROLS
Comparison of perceptions of fatigue
Fatigue scores
p=0.009
OFF GH
ON GH
CONTROLS

Fig 2: The individual value plots above shows that the highest perception of fatigue (A) and impaired QoL (B) was reported by the untreated GHD group and the least fatigue and normal QoL was reported by the healthy controls (ANOVA)

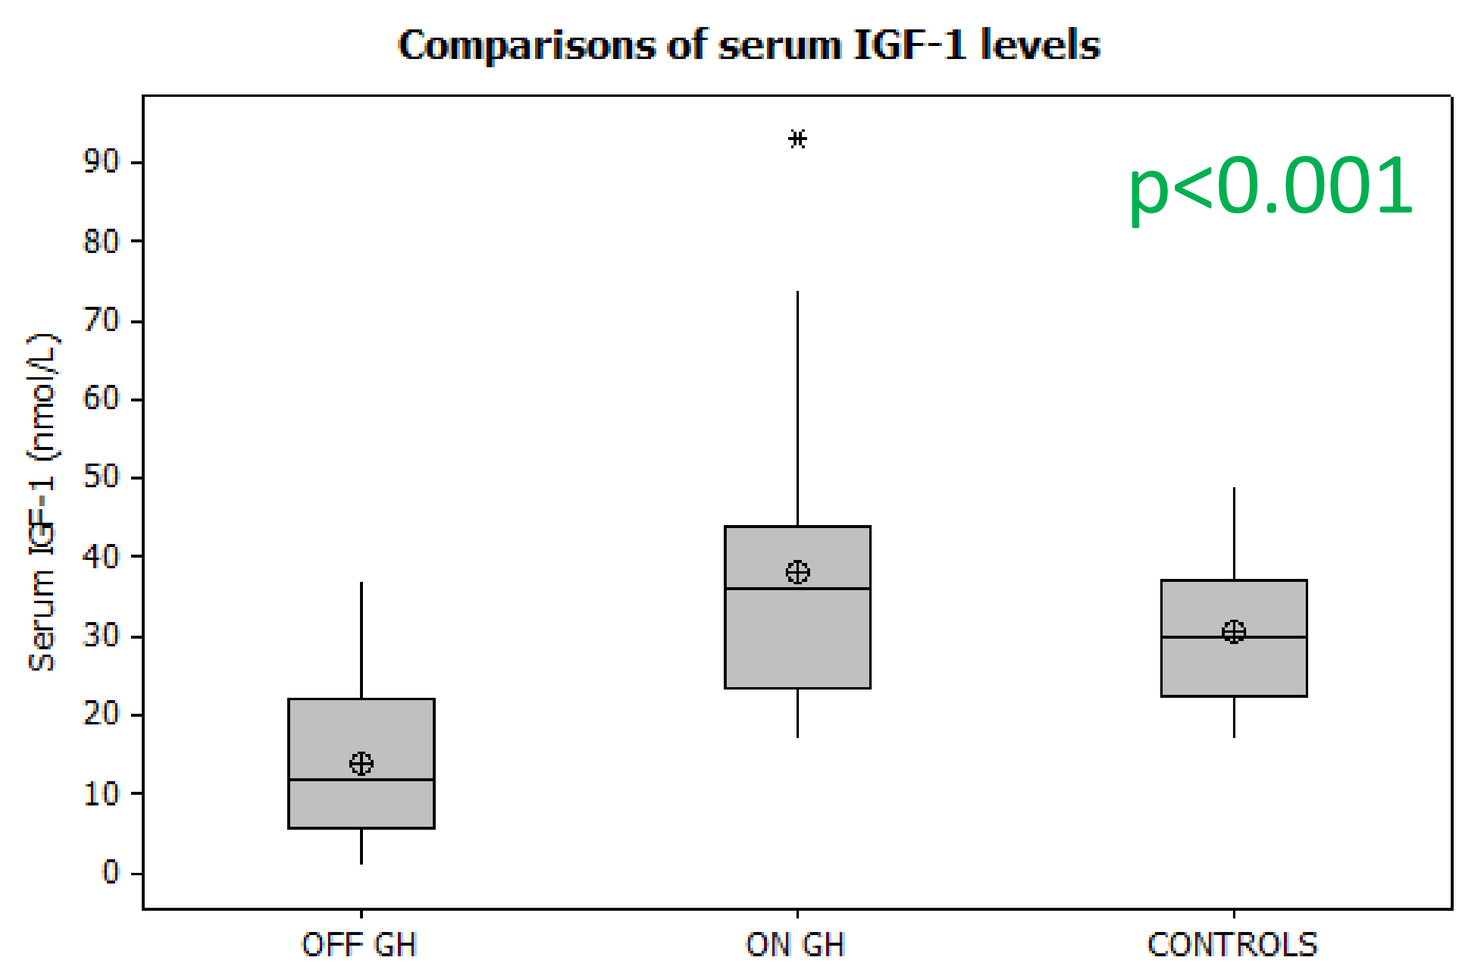
Comparisons of serum IGF-1 levels
Serum IGF-1 (nmol/L)
p<0.001
OFF GH
ON GH
CONTROLS

Fig 3: Comparison of serum IGF-1 levels across the three groups showing lower levels in the untreated GHD group (ANOVA)

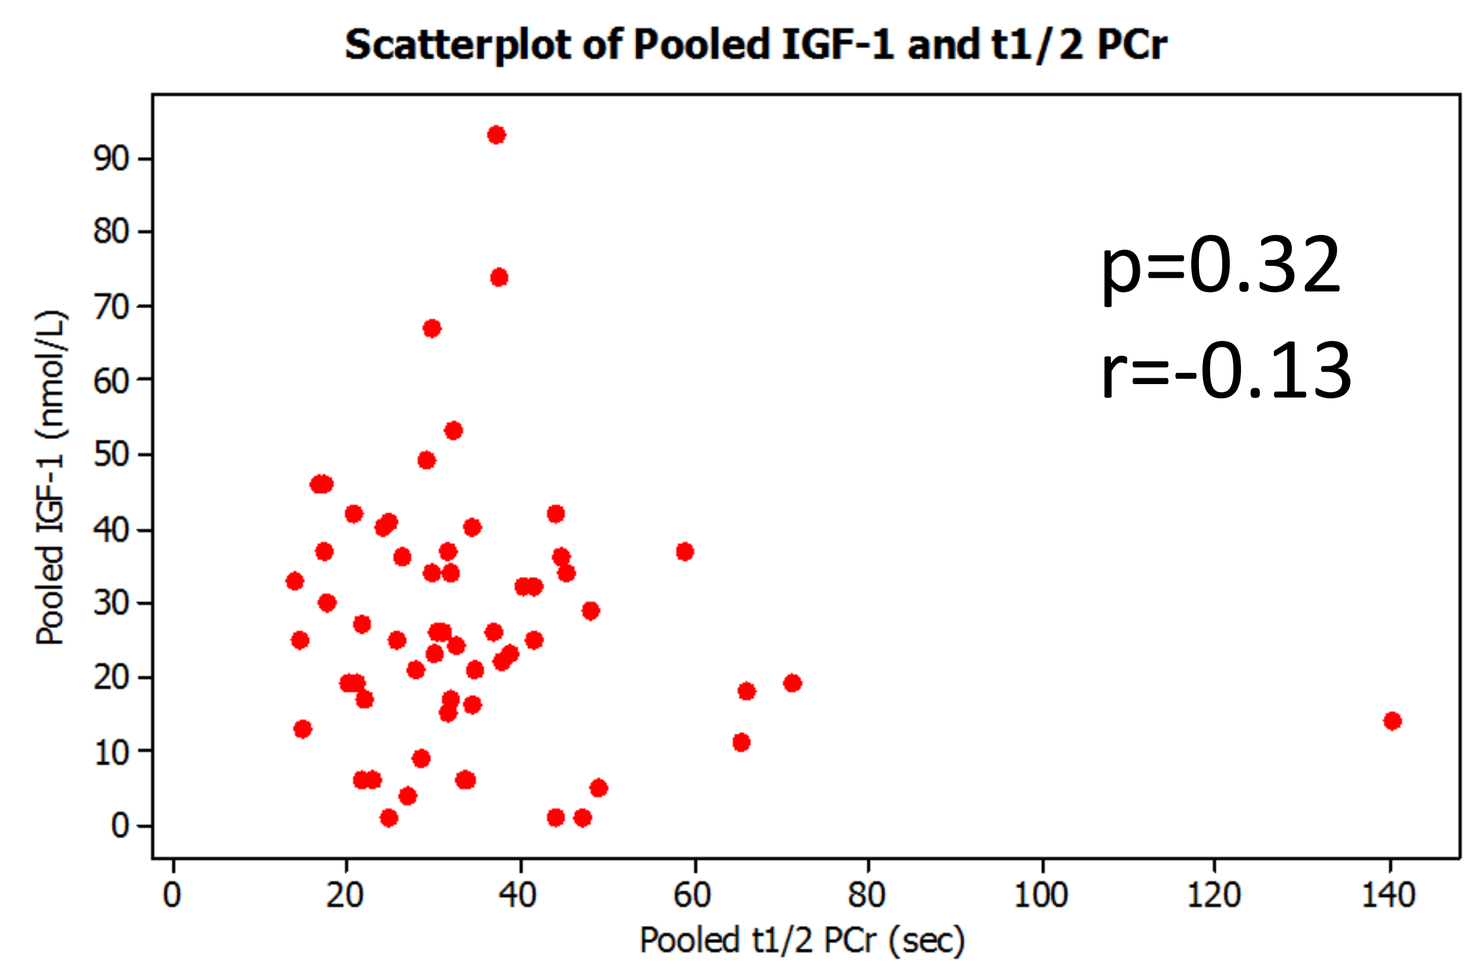
Scatterplot of Pooled IGF-1 and t1/2 PCr
Pooled IGF-1 (nmol/L)
p=0.32
r=-0.13
Pooled t1/2 PCr (sec)

Fig 4: Regression for pooled IGF-1 and pooled $\tau^{1/2}$ PCr showing no correlation between IGF-1 and mitochondrial function (Linear regression)

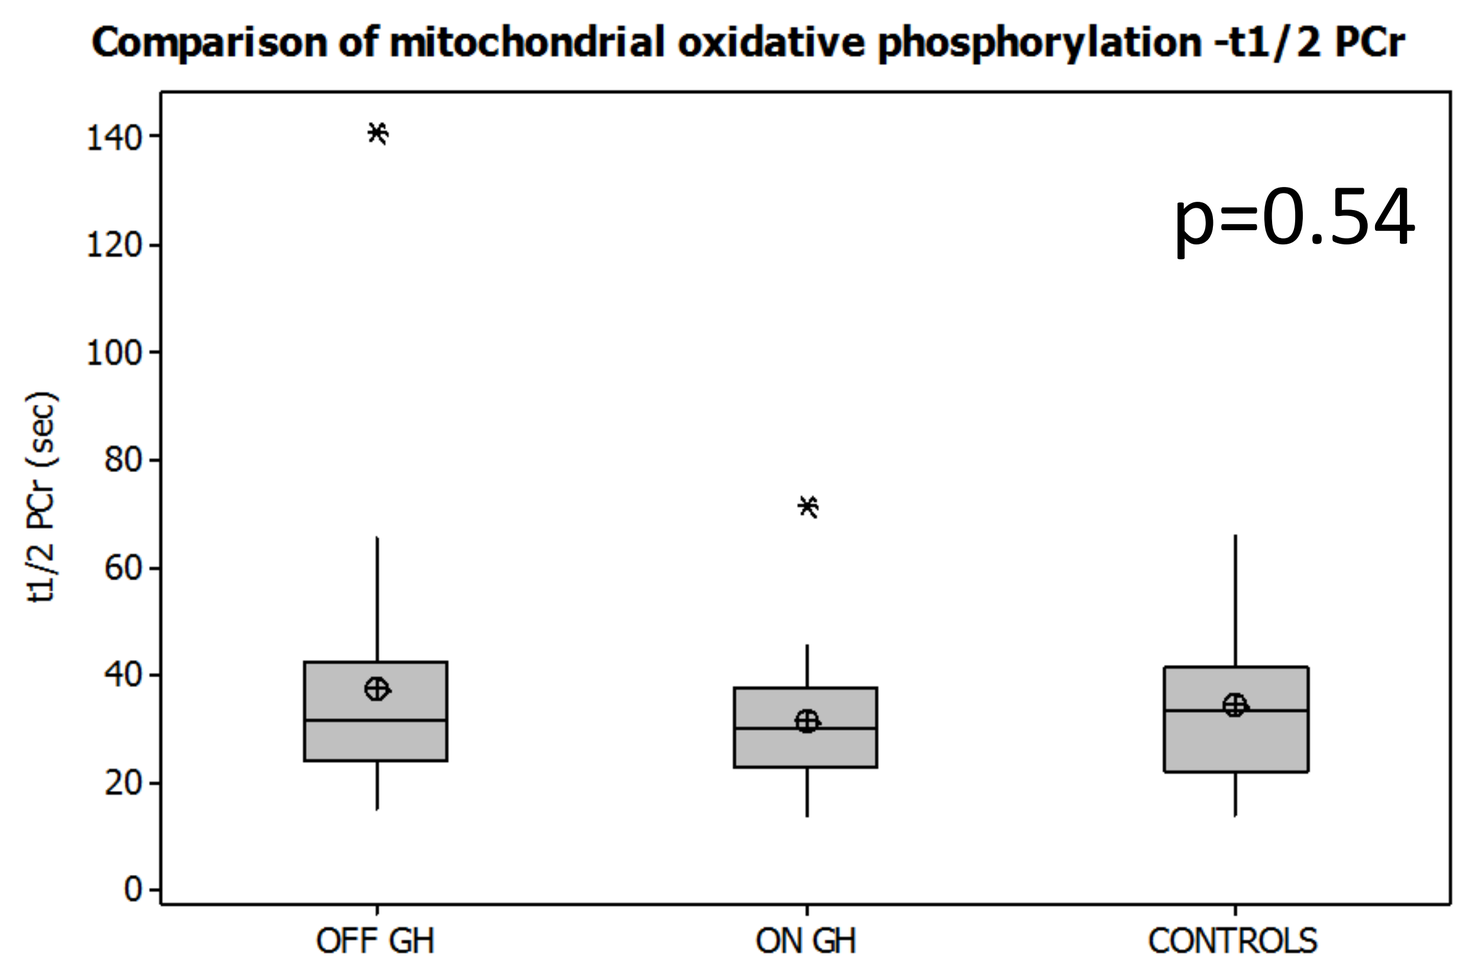
Comparison of mitochondrial oxidative phosphorylation -t1/2 PCr
t1/2 PCr (sec)
p=0.54
OFF GH
ON GH
CONTROLS

Fig 5: The plot above shows no difference in mitochondrial oxidative function between the 3 groups (ANOVA)

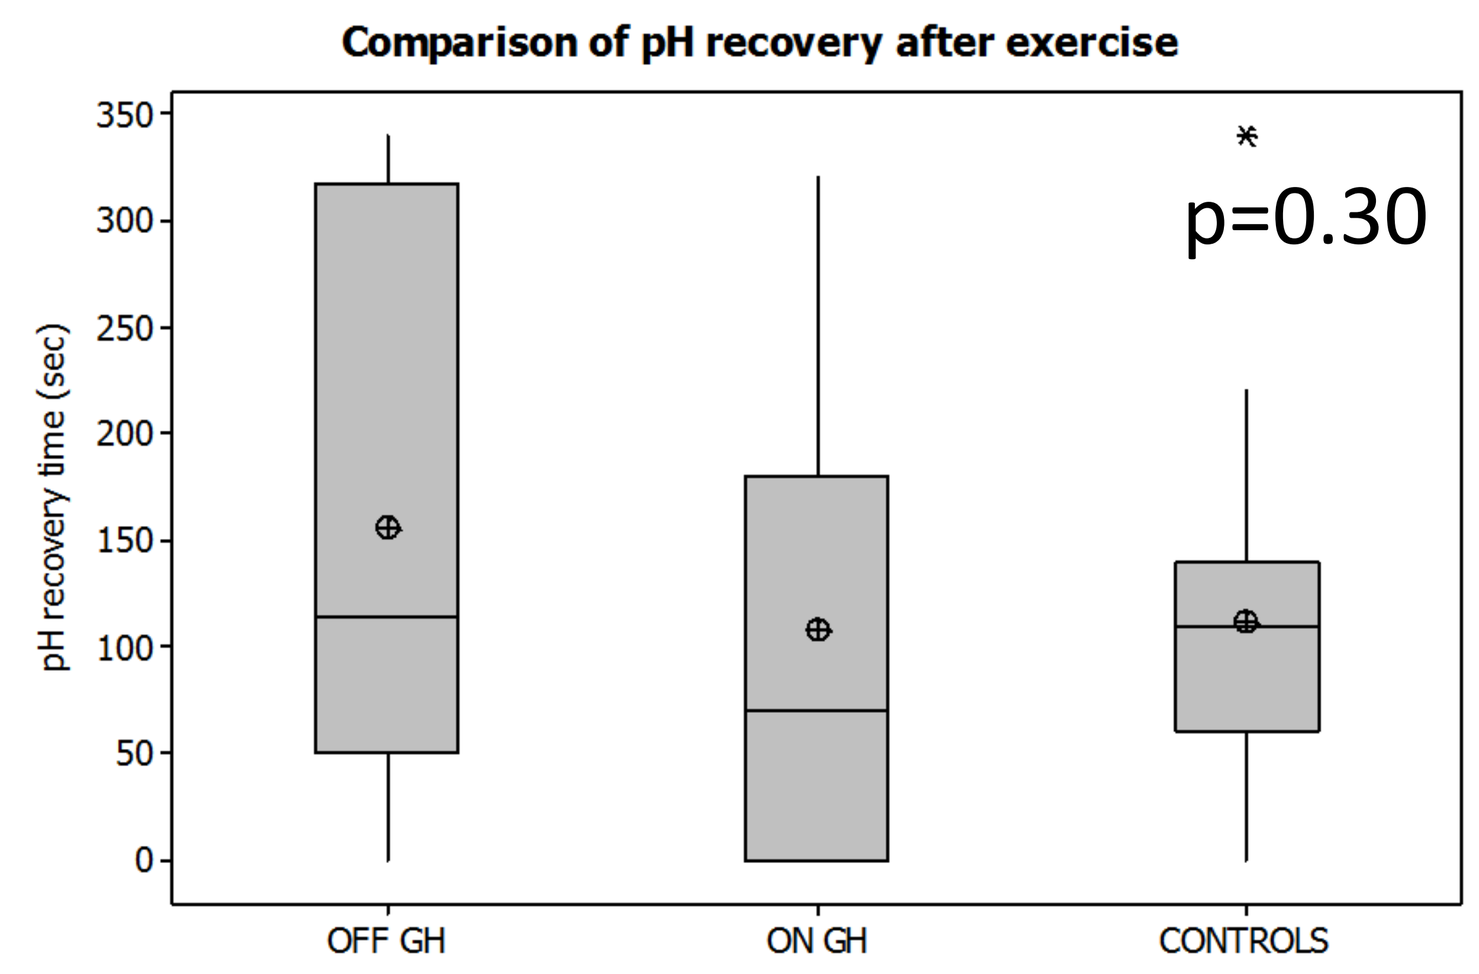
Comparison of pH recovery after exercise
pH recovery time (sec)
p=0.30
OFF GH
ON GH
CONTROLS

Fig 6: The plot above shows no difference in pH recovery times between the 3 groups (ANOVA)

Conclusions

- We found no evidence to suggest that GH status or IGF-1 modulates skeletal muscle mitochondrial oxidative capacity.
- Although untreated GHD adults experience problematic fatigue compared with treated GHD adults and volunteers, they do not demonstrate any persistent abnormalities in skeletal muscle metabolism (mitochondrial oxidative function nor pH recovery), thereby indicating a likely central component in the pathophysiology of fatigue in GHD.

Corresponding author: Akash.sinha@ncl.ac.uk

References:
1. Shalet et al, The effect of long-term untreated GHD and 9 years of GH replacement on the QoL of GHD adults. Clin Endo,2002. **57**(3): p.363-70
2. Makimura et al, The association of growth hormone parameters with skeletal muscle phosphocreatine recovery in adult men. J Clin Endocrinol Metab, 2011. **96**(3): p. 817-23

Disclosure & Acknowledgements:
1. Pfizer Inc and Merc Serono Inc for funding the 1st and 2nd year of this study respectively
2. Staff at the MR centre, Newcastle University and Endocrine Sp nurses, NUTH