School of Endocrinology:

Discrepant GH and IGF-I values in the evaluation of treated acromegalic patients; an ongoing challenge. A meta-analysis.

G. Kanakis[1], A. Chrisoulidou[2], A. Bargiota[3], Z.A. Efstathiadou[4], L. Papanastasiou[5], A. Theodoropoulou[6], S.K.Tigas[7], D.A. Vassiliadi[8], S. Tsagarakis[9] and M. Alevizaki[10]

[1]Department of Endocrinology, Athens Naval & VA Hospital, Athens, Greece; [2]Department of Endocrinology, Theagenio Cancer Hospital, Thessaloniki, Greece; [3]Department of Endocrinology and Metabolic Diseases, University of Thessaly, Larissa, Greece; [4]Department of Endocrinology, "Hippokration" General Hospital of Thessaloniki, Thessaloniki, Greece; [5]Department of Endocrinology and Diabetes Center, Athens General Hospital "G. Gennimatas", Athens, Greece; [6]Department of Internal Medicine, Division of Endocrinology, University Hospital of Patras, Rio, Greece; [7]Department of Endocrinology, University of Ioannina, Ioannina, Greece; [8]Endocrine Unit, 2nd Dept. of Internal Medicine – Propaedeutic, Research Institute and Diabetes Center, Attikon University Hospital, Medical School, National and Kapodistrian University of Athens, Greece; [9]Department of Endocrinology, Evangelismos Hospital Athens, Greece; [10]Endocrine Unit, Department of Medical Therapeutics, University of Athens, Medical School, Athens, Greece

INTRODUCTION - OBJECTIVES

Growth Hormone (GH) and Insulin-like Growth Factor 1 (IGF-I) are currently the principal biomarkers used to assess disease activity in acromegaly and any discrepancy between them, renders interpretation of results inconclusive. The purpose of this study was to assess the frequency of discrepant results and identify parameters that might affect the emergence of this phenomenon.

METHODS

A systematic review of Medline and Scopus was performed (1987 – 2013) followed by a meta-analysis to address the frequency of discrepant results between GH and IGF-I levels. Meta-regression was performed using the year of publication as a continuous variable and subgroup analyses included the different types of GH testing and GH assays used, as well as the results of patients treated with Somatostatin Analogues (SSAs) compared to those treated with non-pharmaceutical modalities.

RESULTS

The analysis retrieved 39 eligible studies totaling 7071 patients. The pooled discordance rate between GH and IGF-I was 25.7% (95% CI: 22.3-29.4) and the predominant format was that of elevated IGF-I with normal GH levels [15.3% (95% CI: 12.5-18.7)].

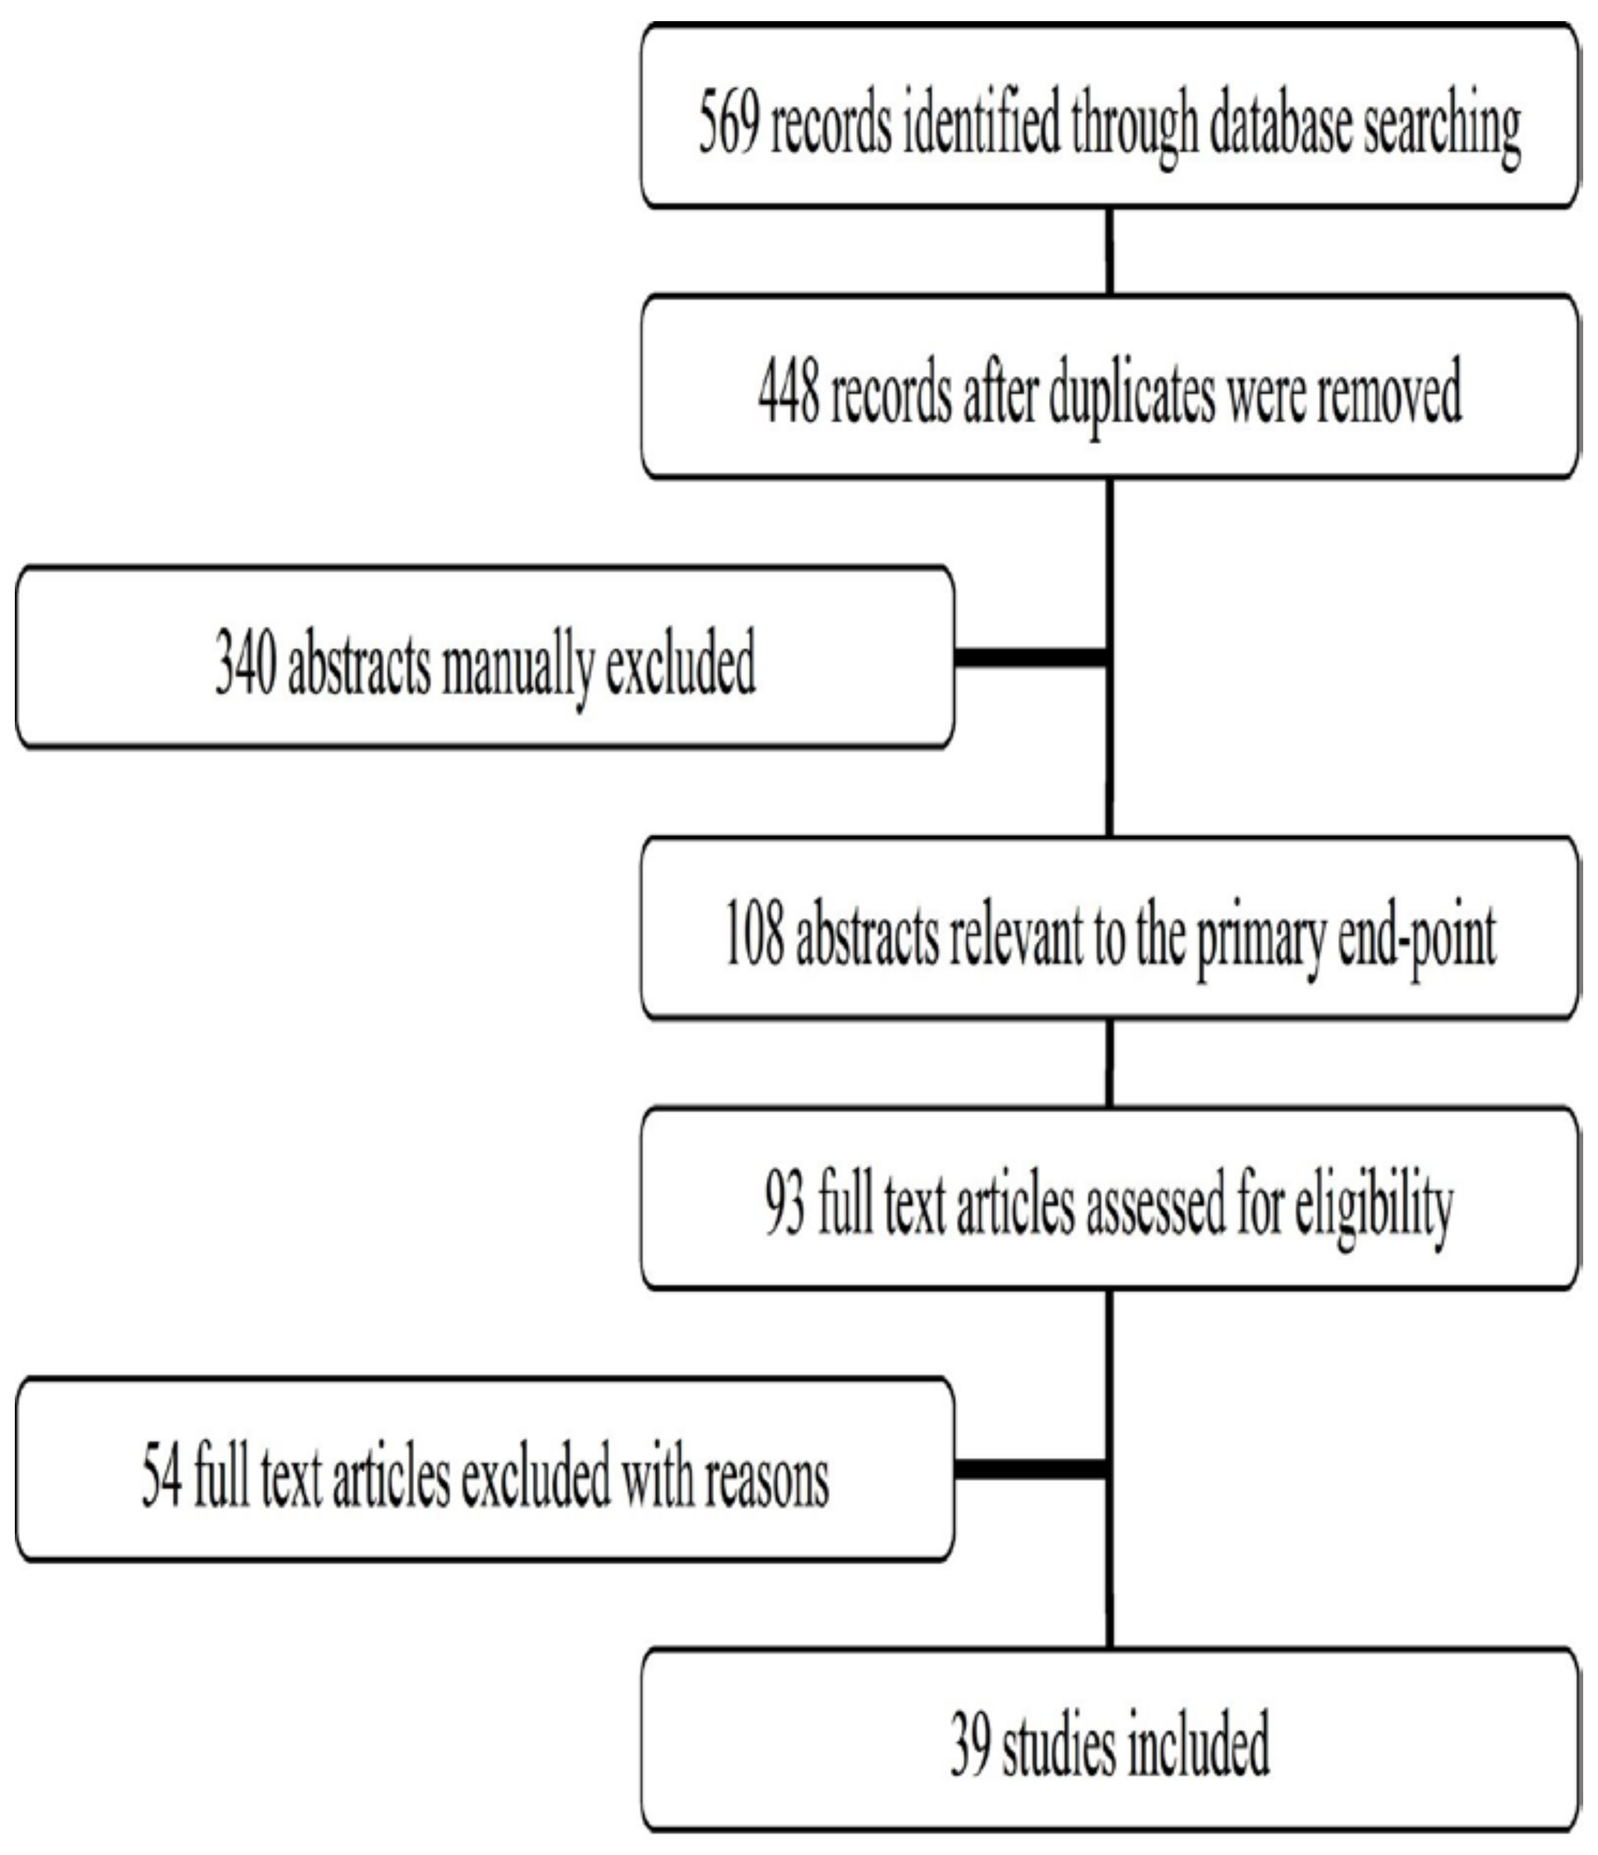
569 records identified through database searching
448 records after duplicates were removed
340 abstracts manually excluded
108 abstracts relevant to the primary end-point
93 full text articles assessed for eligibility
54 full text articles excluded with reasons
39 studies included

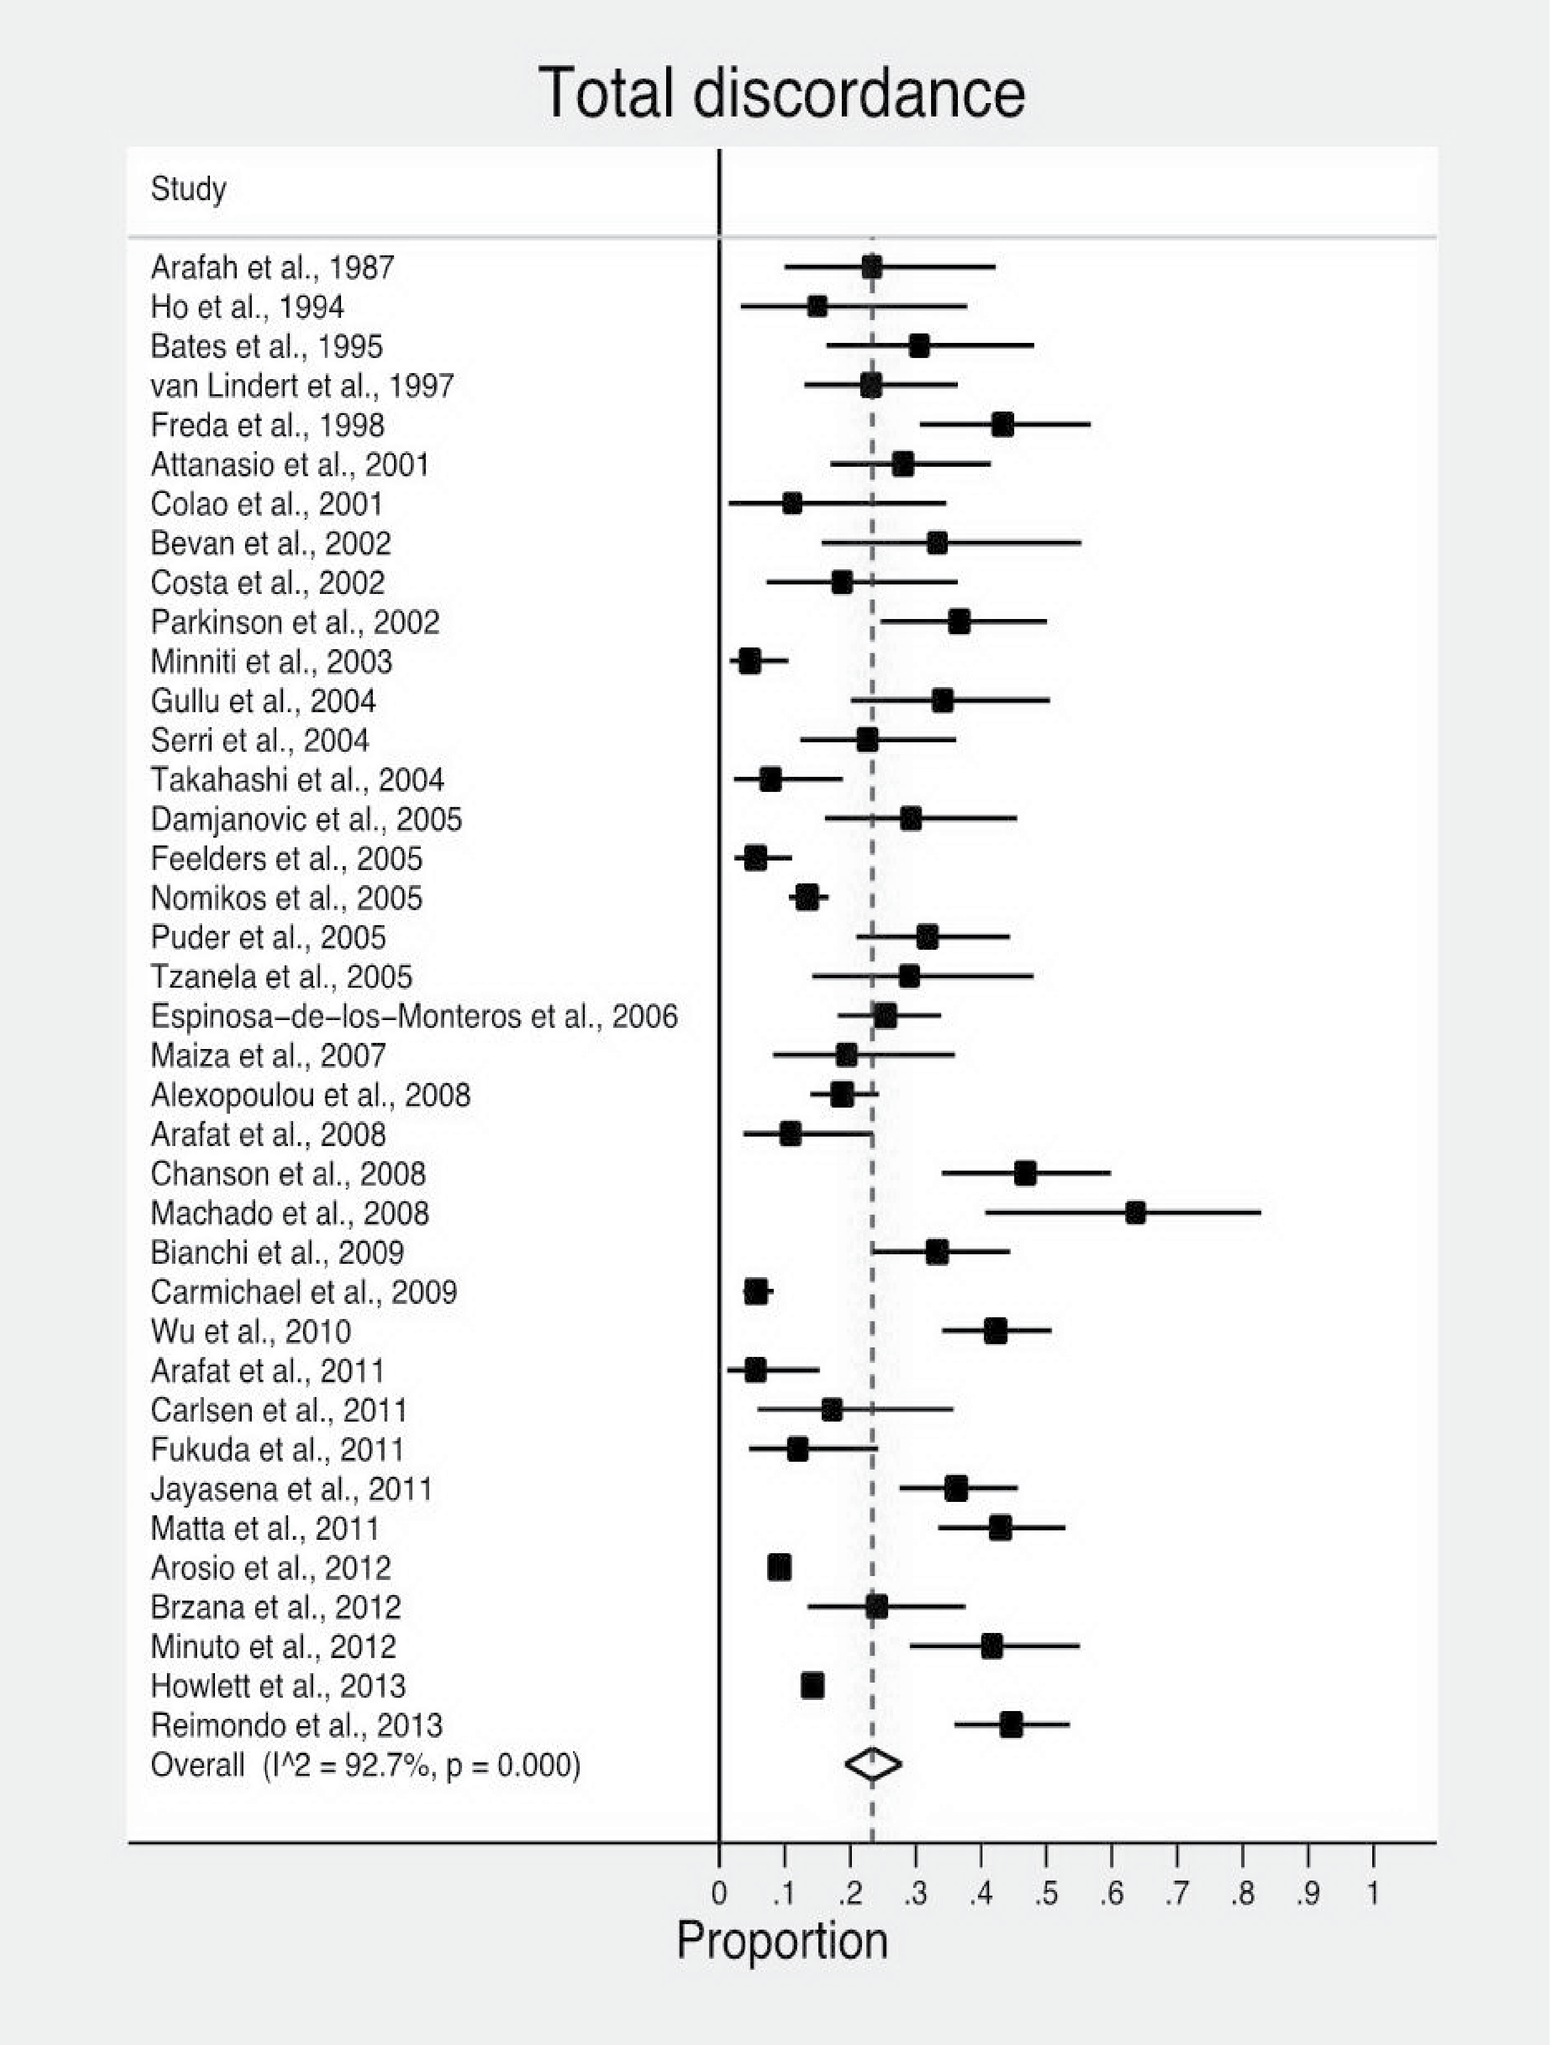
Total discordance
Study
Arafah et al., 1987
Ho et al., 1994
Bates et al., 1995
van Lindert et al., 1997
Freda et al., 1998
Attanasio et al., 2001
Colao et al., 2001
Bevan et al., 2002
Costa et al., 2002
Parkinson et al., 2002
Minniti et al., 2003
Gullu et al., 2004
Serri et al., 2004
Takahashi et al., 2004
Damjanovic et al., 2005
Feelders et al., 2005
Nomikos et al., 2005
Puder et al., 2005
Tzanela et al., 2005
Espinosa–de–los–Monteros et al., 2006
Maiza et al., 2007
Alexopoulou et al., 2008
Arafat et al., 2008
Chanson et al., 2008
Machado et al., 2008
Bianchi et al., 2009
Carmichael et al., 2009
Wu et al., 2010
Arafat et al., 2011
Carlsen et al., 2011
Fukuda et al., 2011
Jayasena et al., 2011
Matta et al., 2011
Arosio et al., 2012
Brzana et al., 2012
Minuto et al., 2012
Howlett et al., 2013
Reimondo et al., 2013
Overall (I^2 = 92.7%, p = 0.000)
0 .1 .2 .3 .4 .5 .6 .7 .8 .9 1
Proportion

No significant correlation between the discordance rate and the year of publication was shown Coeff: +0.003 95% CI: -0.008 to +0.134 (p=0.589)

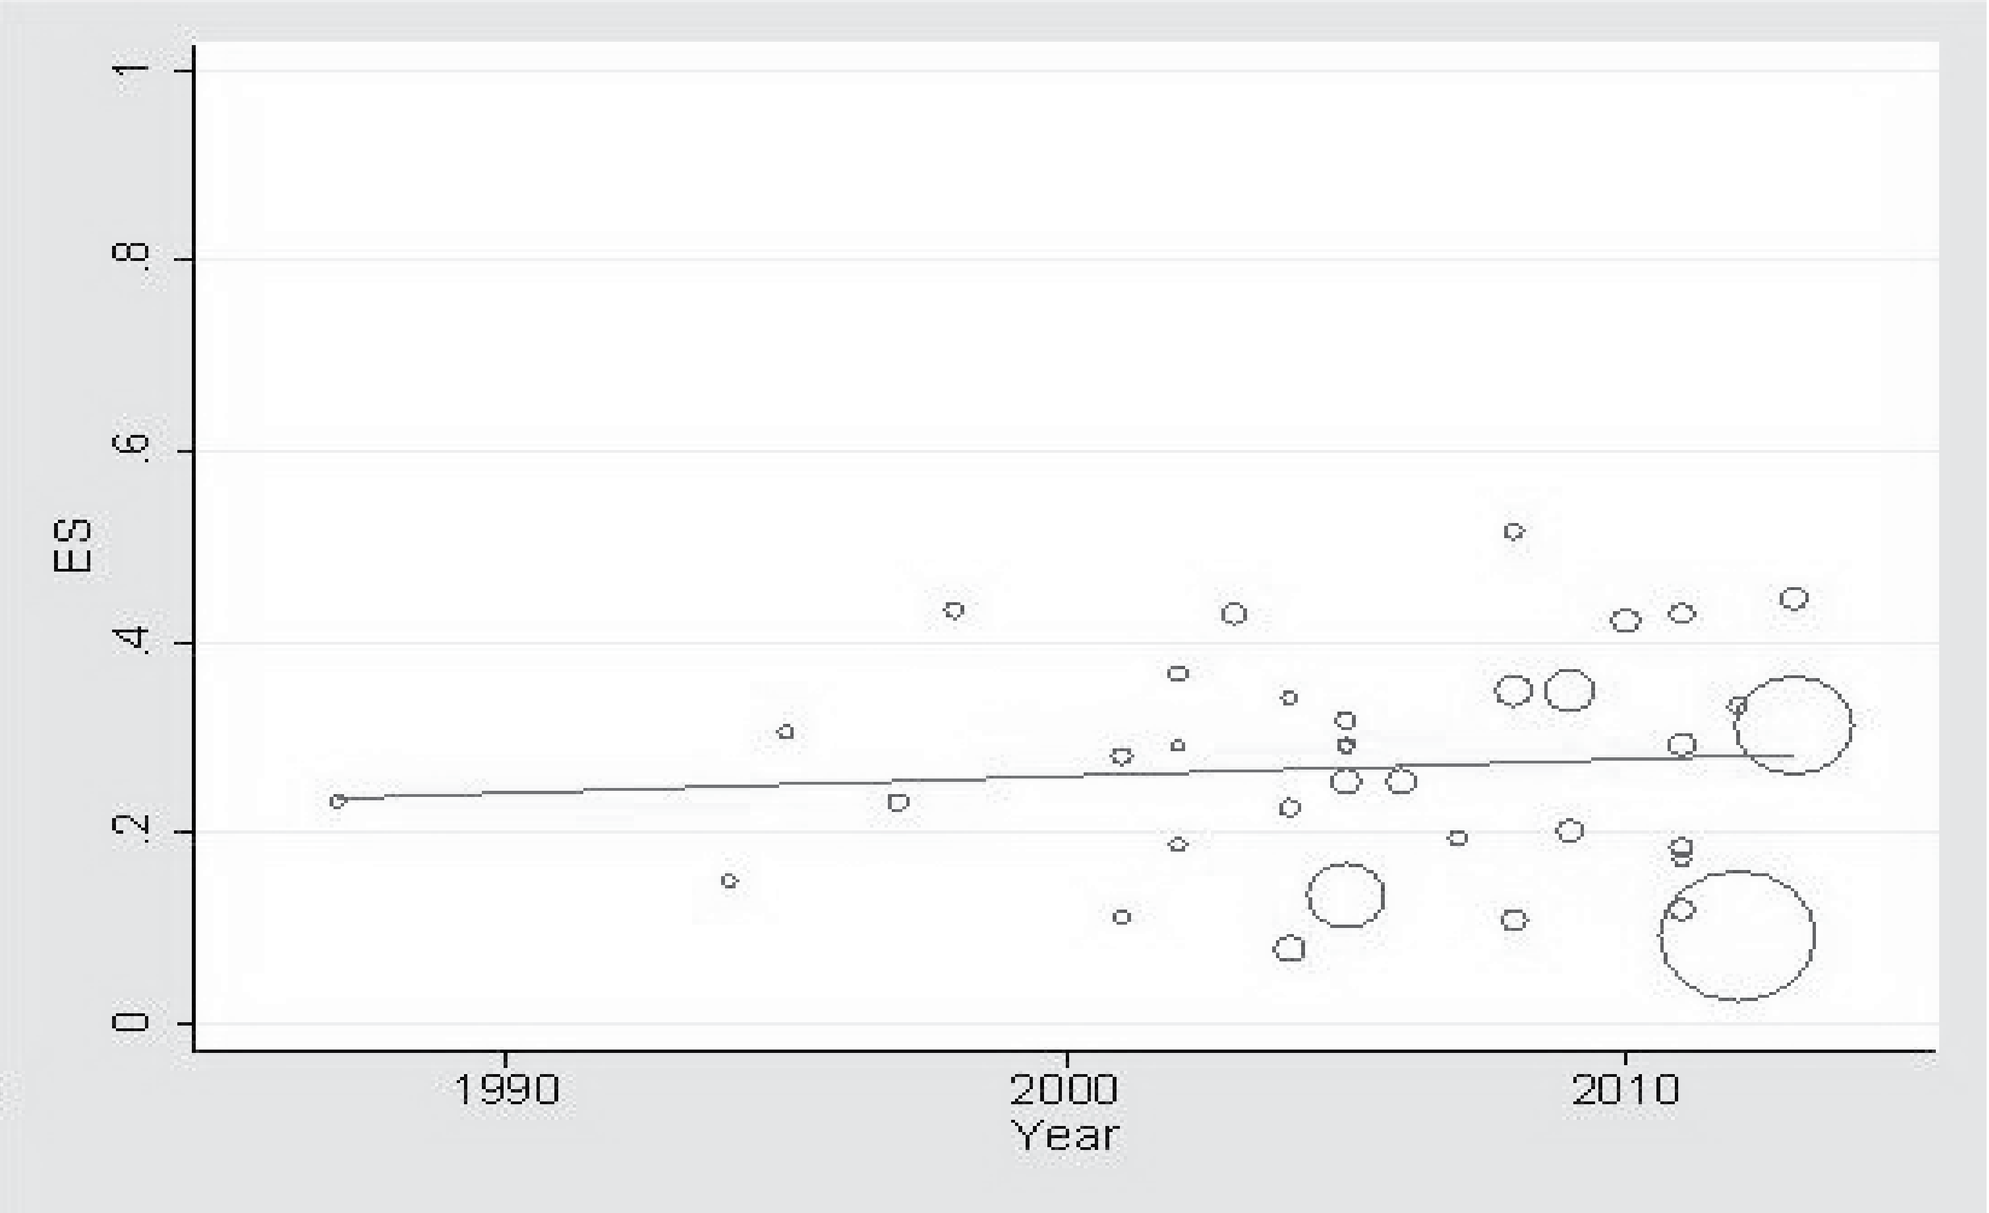
ES
0 .2 .4 .6 .8 1
1990 2000 2010
Year

The use of ultrasensitive GH assays resulted in higher discordance rates (30.7%, 95% CI: 25.9-35.9) vs. (19.8%, 95% CI: 14.1-27.2, p=0.04). On the contrary, GH nadir values obtained during an OGTT (GHn) yielded the lowest discordance rates over a random GH value (GHr) and over the mean value (GHm) of a day curve (22,0%, vs. 29,1 vs. 33,5%, p=0,041).

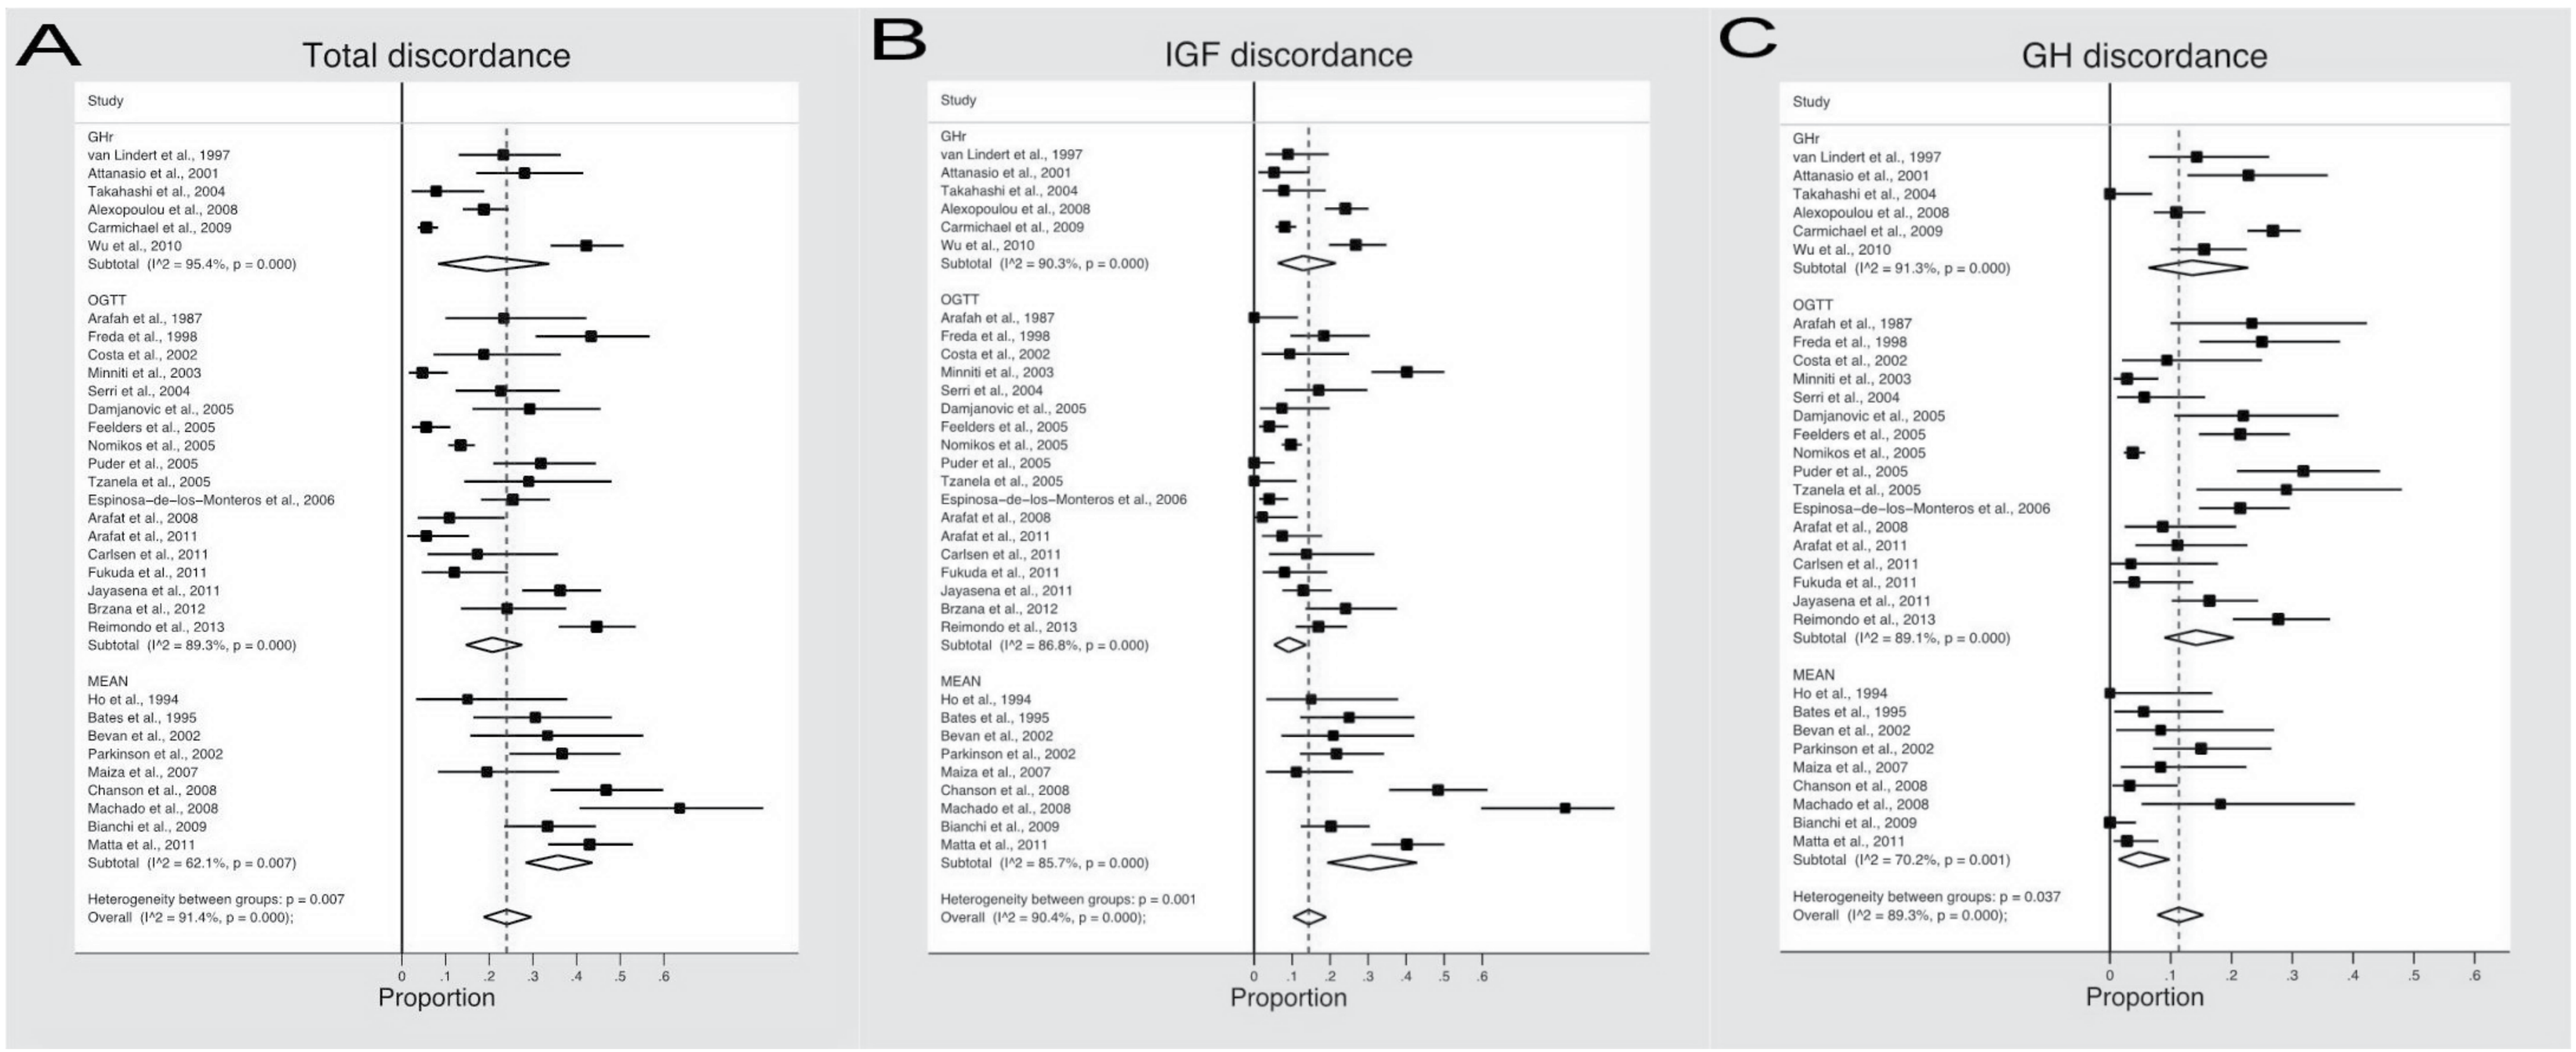
A
Total discordance
B
IGF discordance
C
GH discordance
Proportion

Patients receiving SSAs presented a discordance rate significantly higher than those not on pharmaceutical therapy (32.5%, 95% CI: 27.8-37.4) vs. (21.6%, 95% CI: 17.8-25.6, p=0.001)

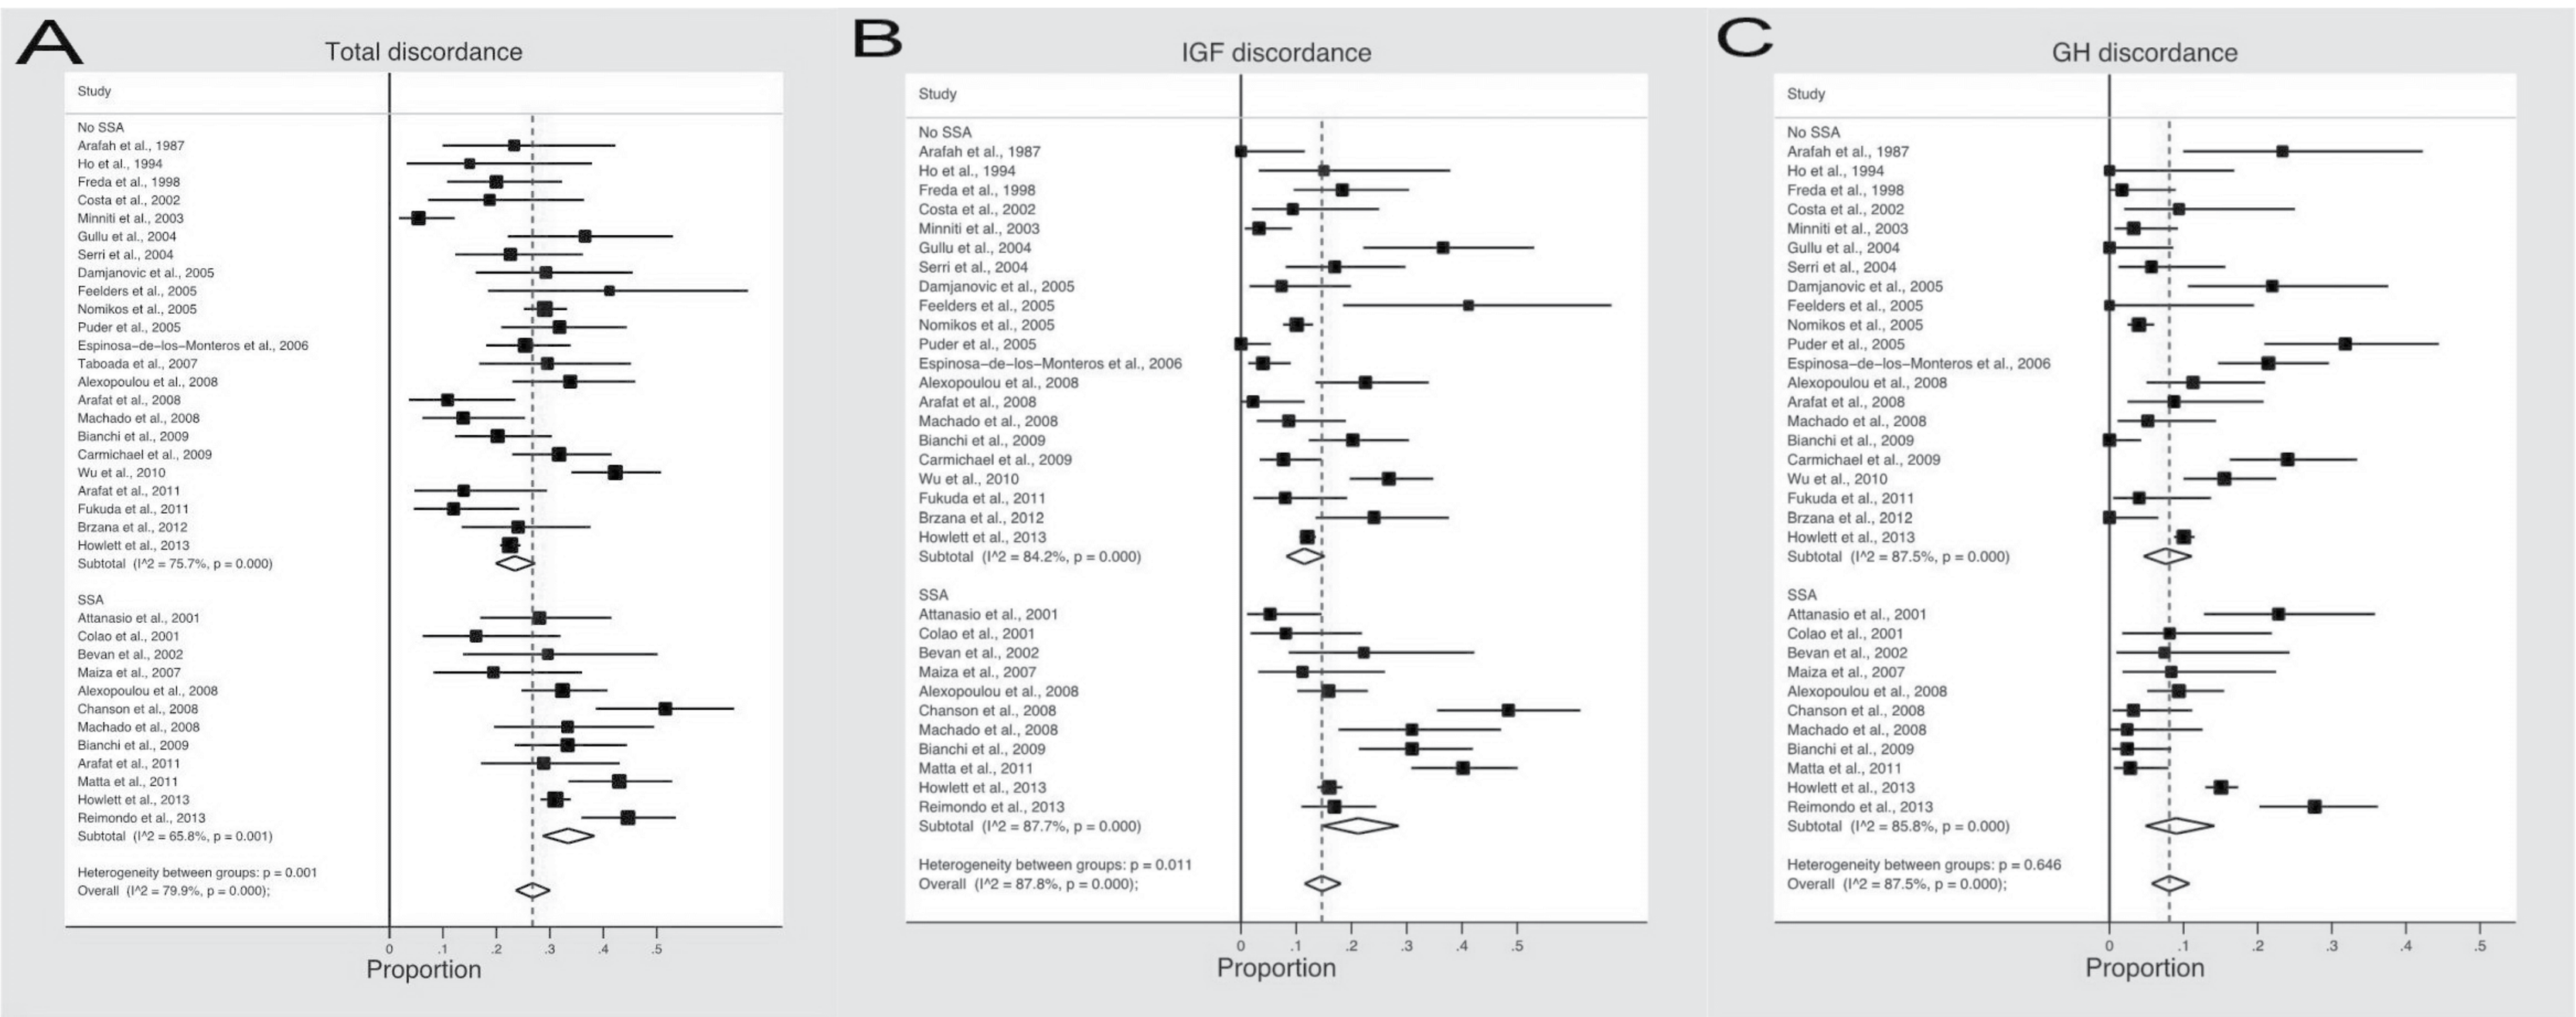
A
Total discordance
B
IGF discordance
C
GH discordance
Proportion

CONCLUSIONS

Discrepancy between GH and IGF-I results is encountered in almost a quarter of treated patients with acromegaly, especially when using ultrasensitive GH assays or in patients receiving SSAs, a fact that the clinician should take into consideration when making clinical decisions.

REFERENCES

1. Giustina, A., Chanson, P., Bronstein, M. D., et al. (2010). A consensus on criteria for cure of acromegaly. *J Clin Endocrinol Metab*, **95** (7), 3141-8.

2. Alexopoulou, O., Bex, M., Abs, R., et al. (2008). Divergence between growth hormone and insulin-like growth factor-i concentrations in the follow-up of acromegaly. *J Clin Endocrinol Metab*, **93** (4), 1324-30.

3. Freda, P. U. (2009). Monitoring of acromegaly: what should be performed when GH and IGF-1 levels are discrepant? Clin Endocrinol (Oxf), 71 (2), 166-70.

4. Zeinalizadeh, M., Habibi, Z., Fernandez-Miranda, J. C., et al. (2015). Discordance between growth hormone and insulin-like growth factor-1 after pituitary surgery for acromegaly: a stepwise approach and management. Pituitary, 18 (1), 48-59.

The Greek School of Endocrinology Initiative was organized and funded by Ipsen. Statistical support and publication costs associated with this work were also funded by Ipsen. The development at all stages of the concept and content was the sole responsibility of the authors.

Lower Ribbon Requested by the Organizers

ECE 2016 874-EP

Pituitary - Clinical

George Kanakis

Poster presented at: ECE2016 28 – 31 May 2016 MUNICH, GERMANY 18th EUROPEAN CONGRESS OF ENDOCRINOLOGY

PosterSessionOnline.com