A systematic review and meta-analysis of interventions for the treatment of chronic non-hypovolaemic hypotonic hyponatraemia

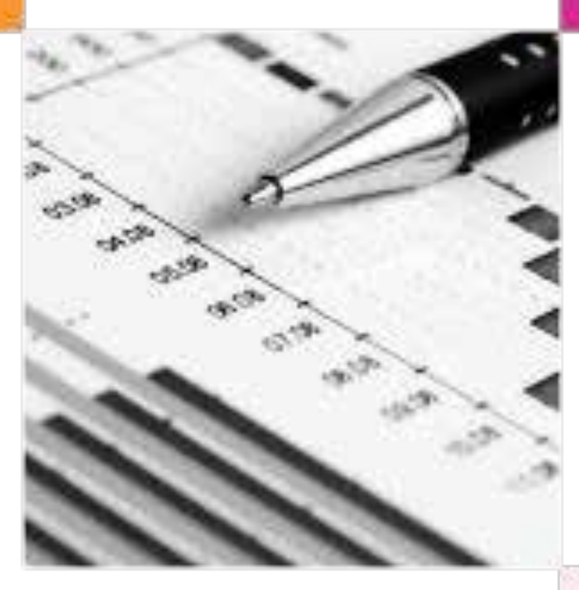

McCool R[1], Glanville J[1], Quinn J[2], O'Reilly K[2], Petrakova L[2], Jamookeeah C[2]

[1]York Health Economics Consortium, York, UK. [2]Otsuka Pharmaceutical Europe Ltd., Wexham, UK.

BACKGROUND AND OBJECTIVES

International and national guidelines on the treatment of chronic non-hypovolaemic hypotonic hyponatraemia differ widely in their recommendations. We used Cochrane methods to conduct a systematic review and meta-analysis (PROSPERO registration number: CRD42015016670) of the effectiveness and safety of interventions for this condition.

Figure 1: Study selection (PRISMA Diagram)

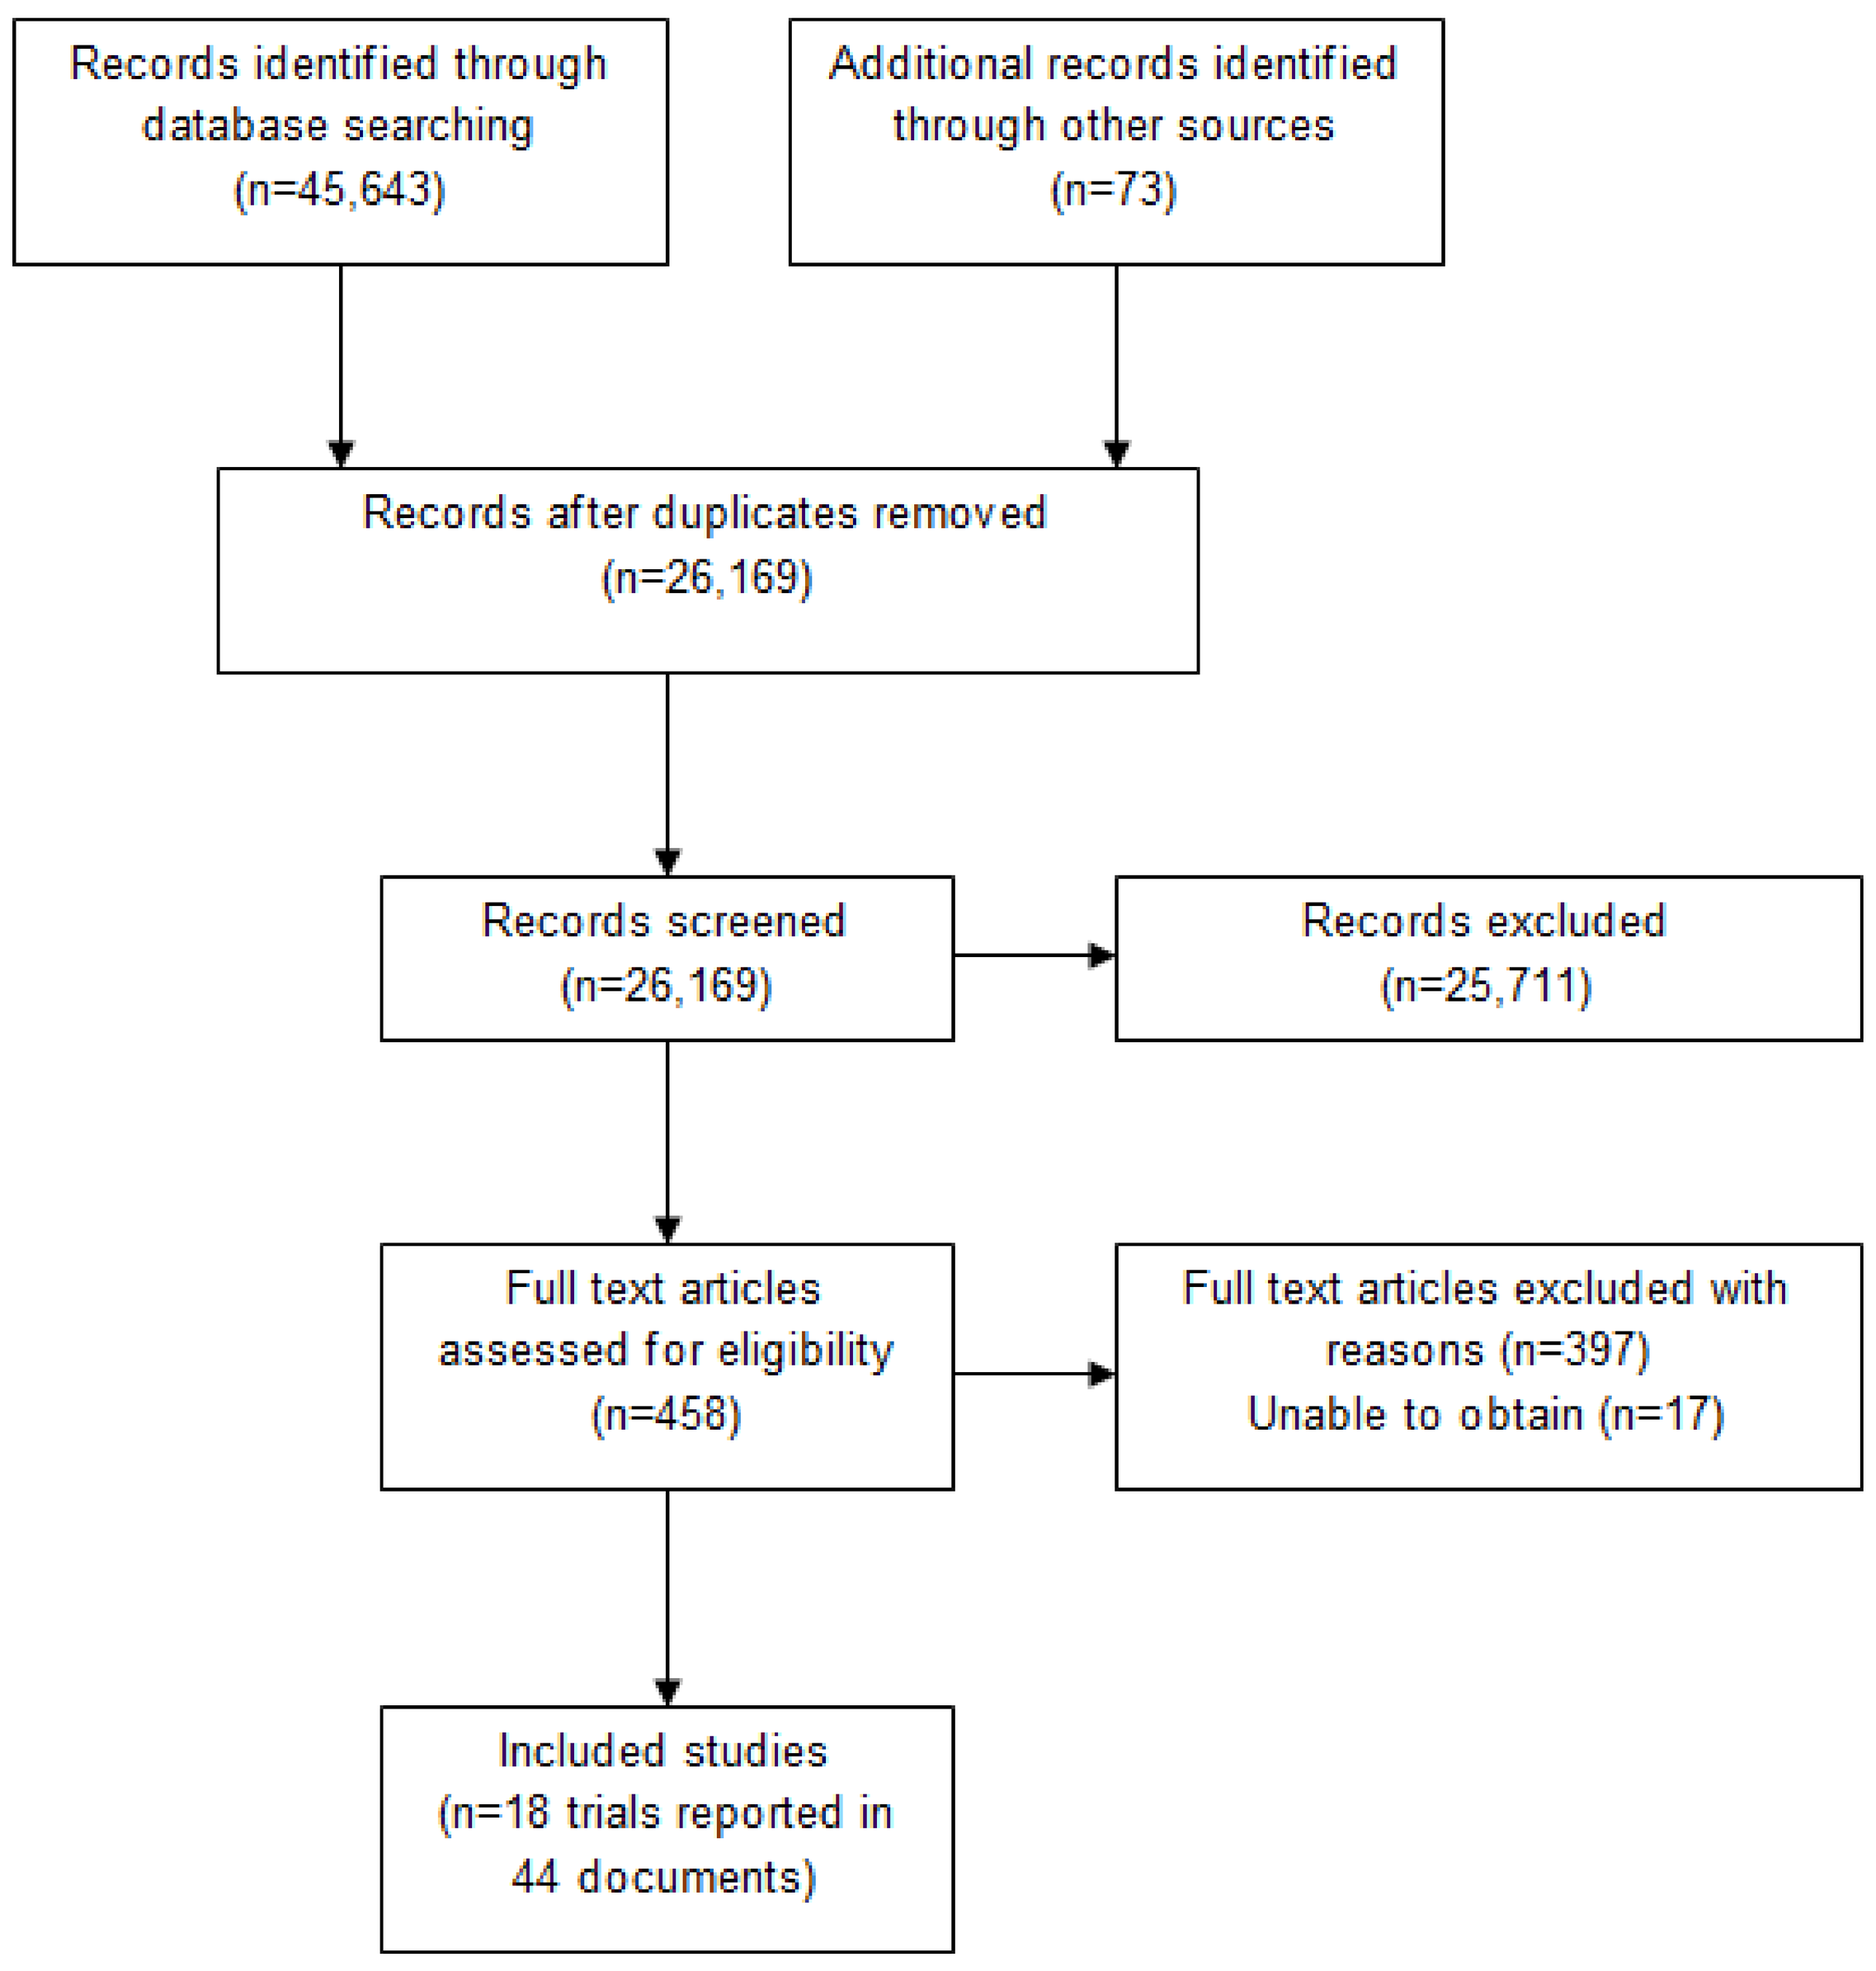
Records identified through database searching (n=45,643)
Additional records identified through other sources (n=73)
Records after duplicates removed (n=26,169)
Records screened (n=26,169)
Records excluded (n=25,711)
Full text articles assessed for eligibility (n=458)
Full text articles excluded with reasons (n=397)
Unable to obtain (n=17)
Included studies (n=18 trials reported in 44 documents)

METHODS

Eligible studies were randomised or quasi-randomised controlled trials published in English that compared any degree of fluid restriction or any drug treatment with the aim of increasing serum sodium concentration in patients with chronic non-hypovolaemic hypotonic hyponatraemia. A range of outcomes were specified including: change in serum sodium concentration (mmol/L), response (defined as an increase in serum sodium of ≥ 5 mmol/L or normalisation of serum sodium concentration (≥135 to 145 mmol/L), mortality and discontinuation, which are reported here. Outcomes relating to health related quality of life, length of hospital stay, outcomes related to over-correction of serum sodium concentration and adverse events were also investigated but are not reported here. The review protocol was registered with PROSPERO.

Systematic literature searches were conducted in 7 databases (including Medline and Embase) as well as the websites of relevant conference proceedings up to February 2015.

Studies were screened by two independent reviewers against the eligibility criteria. Any disagreements were discussed with a third reviewer. Data extraction and quality assessment was carried out by two independent reviewers. Key data were collected on the trial design and methodology, patient population, details of interventions and relevant outcomes. Risk of bias was assessed using the Cochrane Risk of Bias tool (1). Where appropriate, outcome data were combined using both fixed- and random-effects models in a meta-analysis using RevMan (version 5.3) (2). Results of the random effects model were deemed most appropriate and are reported here. A sensitivity analysis was conducted excluding studies with a high risk of bias.

RESULTS

The searches retrieved 45,716 records and 18 trials met the eligibility criteria or reported data for an eligible subgroup of patients; 2 trials assessed conivaptan, 4 trials assessed lixivaptan, 3 trials assessed satavaptan, and 9 trials assessed tolvaptan*. No RCT evidence was identified in relation to the alternative strategies commonly employed such as oral urea, salt tablets, mannitol, loop diuretics, demeclocycline and lithium.

Following an assessment of the risk of bias of the included trials, 5 trials were judged to be of a high risk, 12 were deemed to have an unclear risk and only one trial had a low risk of bias.

In both the individual trials and in the pooled analyses, all four "vaptans" were found to significantly improve serum sodium concentration and were associated with greater rates of response compared to placebo, with the exception of tolvaptan at day 14. There was no evidence of a difference between any of the vaptans compared with placebo for mortality, discontinuation and rates of over-correction in the individual trials or pooled analyses.

The sensitivity analyses excluding studies with a high risk of bias did not significantly alter the results of the meta-analyses.

*Lixivaptan, satavaptan and conivaptan do not have marketing authorisations in Europe. Tolvaptan (Samsca®) is licenced in Europe for the treatment of adult patients with hyponatraemia secondary to syndrome of inappropriate antidiuretic hormone secretion (SIADH).

Table 1: Results of the meta-analyses

Intervention	Time point (days)	Change in serum sodium concentration		Response	
		Number of studies	Mean Difference [95%CI]	Number of studies	Relative Risk [95%CI]
Conivaptan	5	2	5.34 [2.82, 7.86]	-	-
Lixivaptan	7	4	2.82 [1.70, 3.95]	3	2.81 [1.34, 5.91]
	56	-	-	1	1.39 [1.03, 1.88]
Tolvaptan	4	5	3.98 [2.31, 5.65]	4	3.30 [1.97, 5.54]
	7	-	-	4	2.22 [1.25, 3.93]
	14	4	4.22 [2.09, 6.34]	2	4.24 [0.55, 32.40]
	25-30	3	3.54 [2.15, 4.94]	3	2.25 [1.74, 2.89]
Satavaptan	4-5	3	4.95 [2.81, 7.10]	3	2.57 [1.66, 4.00]

CONCLUSIONS

The RCT evidence indicates that each of the vaptans significantly improves serum sodium concentration compared with placebo and is associated with greater rates of response than placebo. Few studies measured health-related quality of life (QoL) using a range of different instruments and showed no evidence that vaptans negatively impact QoL. For all vaptans, cases of overcorrection were rare. There was no evidence of a difference between any of the vaptans compared with placebo for mortality, discontinuation or rates of hypernatremia. With the exception of vaptans, no RCT evidence was identified in relation to the alternative strategies commonly employed including urea, mannitol, loop diuretics, corticosteroids, demeclocycline, lithium and phenytoin. Further head to head RCTs are required to determine the comparative efficacy of other treatments currently used in clinical practice or recommended in guidelines (hypertonic saline, oral urea, salt tablets, loop diuretics, mannitol, demeclocycline, lithium) for chronic hypotonic non-hypovolaemic hyponatraemia.

REFERENCES

1. Higgins JP, Green S. Cochrane handbook for systematic reviews of interventions: The Cochrane Collaboration, 2011. **2.** Review Manager (RevMan) [Computer program]. Version 5.3. Copenhagen: The Nordic Cochrane Centre, The Cochrane Collaboration, 2014.

York Health Economics Consortium

ECE 2016 230-EP

Cardiovascular Endocrinology and Lipid Metabolism

Larisa Petrakova

Poster presented at: ECE2016 28 – 31 May 2016 MUNICH, GERMANY 18th EUROPEAN CONGRESS OF ENDOCRINOLOGY

PosterSessionOnline.com