EP 429

An acute oral fructose bolus affects circulating FGF21 in rats, but not in mice

Maximilian Bielohuby, Mark Sommerfeld, Jennifer Grund, Silke Loy, Vanessa Pruss, Uwe Schwahn, Angelika Winkel, Andreas Herling & Norbert Tennagels

Sanofi-Aventis Deutschland GmbH, Department of Insulin Biology, TA Diabetes,
Industriepark Höchst, Frankfurt am Main, Germany. Contact: Maximilian.Bielohuby@sanofi.com

ECE 2016 meeting, Munich, Germany, 28 – 31 May 2016

Introduction

Impaired glucose tolerance and insulin sensitivity are established hallmarks of diabetes and it was speculated that also fructose tolerance could be impaired. However, investigation of fructose tolerance has been hampered by the lack of easy accessible pharmacodynamic markers. In humans, Dushay et al. have recently demonstrated that the hormone FGF21 is a sensitive pharmacodynamic marker responding to an oral fructose bolus and exhibiting a divergent secretion pattern in obese diabetic and healthy lean subjects[1]. Here we investigated whether FGF21 excursion after an oral fructose challenge follows the same rules in obese mice and in healthy rats. If so, this novel test could be used to address specific research questions. Furthermore, obese mice and healthy rats were pre-treated with the DPP4 inhibitor Sitagliptin to investigate if a potential FGF21 response to oral fructose is affected by this commonly used oral anti-diabetic drug.

[1] Reference: Fructose ingestion acutely stimulates circulating FGF21 levels in humans. Dushay JR, Toschi E, Mitten EK, Fisher FM, Herman MA, Maratos-Flier E. Mol Metab. 2014 Oct 8;4(1):51-7. doi: 10.1016/j.molmet.2014.09.008.

Materials and Methods

Adult lean ob/- and obese ob/ob as well as adult male Wistar WU rats were obtained from Charles-River, Germany. Two weeks prior to fructose treatment, mice were subjected to a standard oral glucose tolerance test (oGTT) to confirm the expected impairment of oral glucose tolerance (Fig. 1). After recovery from the oGTT, fasted (4h) lean ob/- and obese ob/ob mice received a fructose bolus by oral gavage (1g/kg, n=10/group). Another cohort of ob/ob mice received a single pre-treatment with Sitagliptin (at t=-30min, 40µg/mouse, MSD Sharp and Dohme) before exposure to oral fructose (“ob/ob-S”, n=10). Healthy Wistar rats were divided into 3 experimental groups: a) no pre-treatment and tap water (n=6); b) vehicle pre-treatment and oral fructose (1g/kg, n=5) and c) single pre-treatment with Sitagliptin (40µg/rat, t=-30min) and oral fructose (1g/kg, n=5). Blood was collected at timepoints -30, 0, 30, 60, 120 and 180min after fructose treatment for analysis of glucose and plasma FGF21 (mice). In rats, higher blood volume allowed higher multiple sampling volumes for analysis of glucose, serum fructose, plasma insulin and FGF21. Data are presented as means ± SEM.

Results

Mice

Fig. 1: oGTT in mice before oral fructose load: as expected, ob/ob mice showed an impaired oral glucose tolerance in comparison to lean ob/- mice.

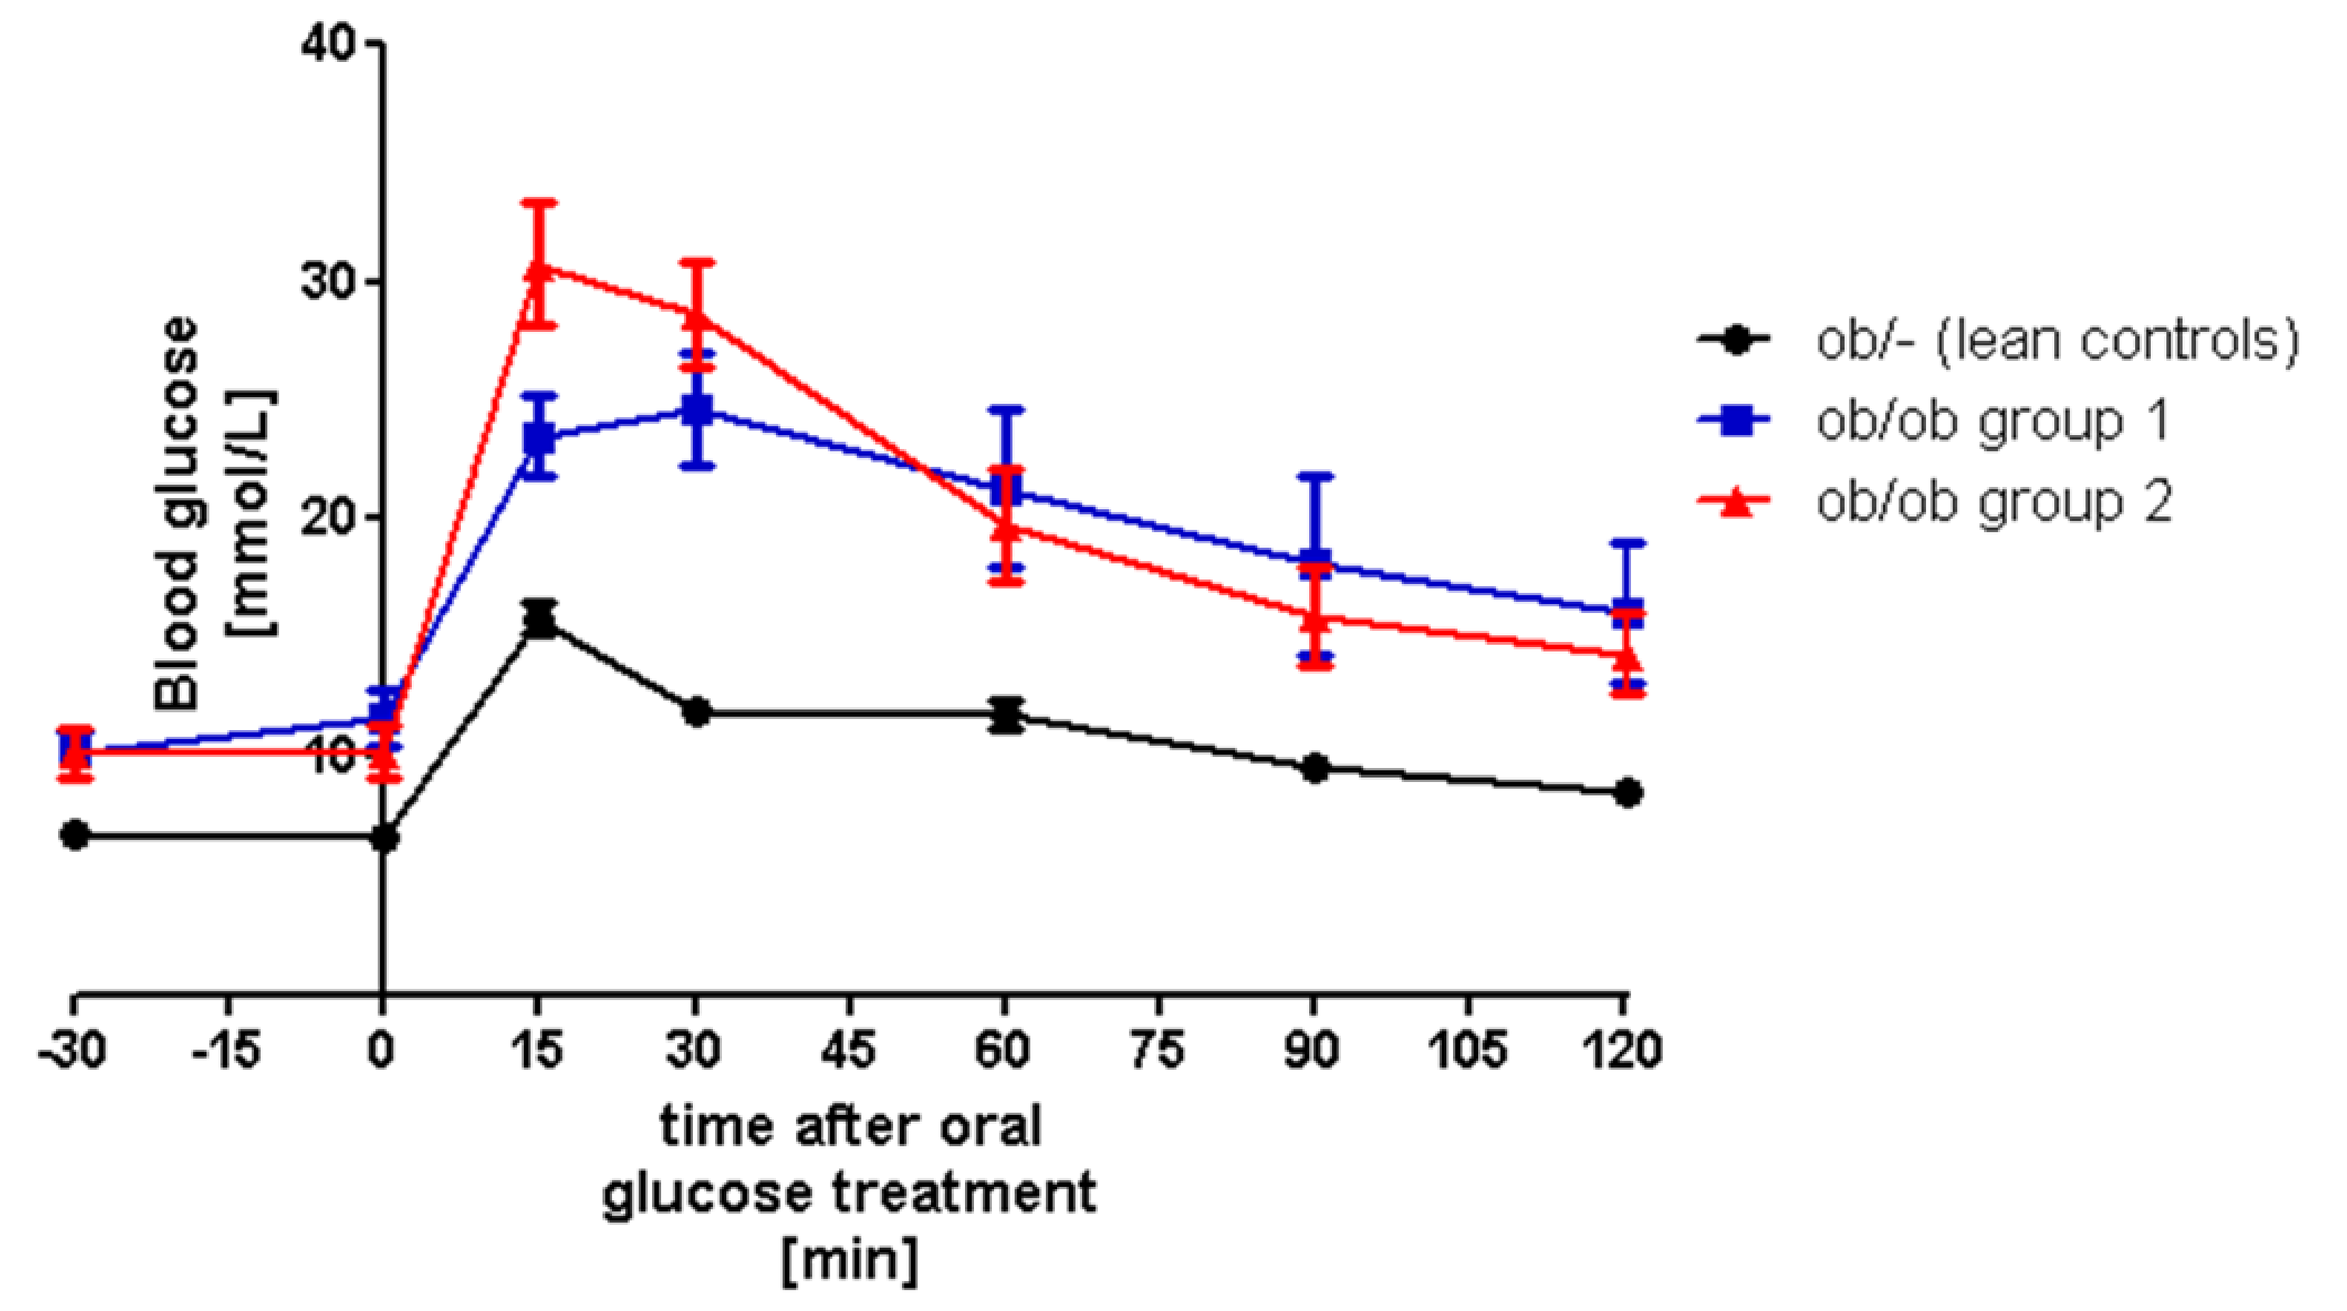
Blood glucose [mmol/L]
40
30
20
10
-30 -15 0 15 30 45 60 75 90 105 120
time after oral glucose treatment [min]
ob/- (lean controls)
ob/ob group 1
ob/ob group 2

Fig. 2: Blood glucose after an oral fructose bolus (1g/kg): ob/ob mice displayed higher baseline glucose concentrations. Oral gavage with fructose did not significantly affect blood glucose concentrations.

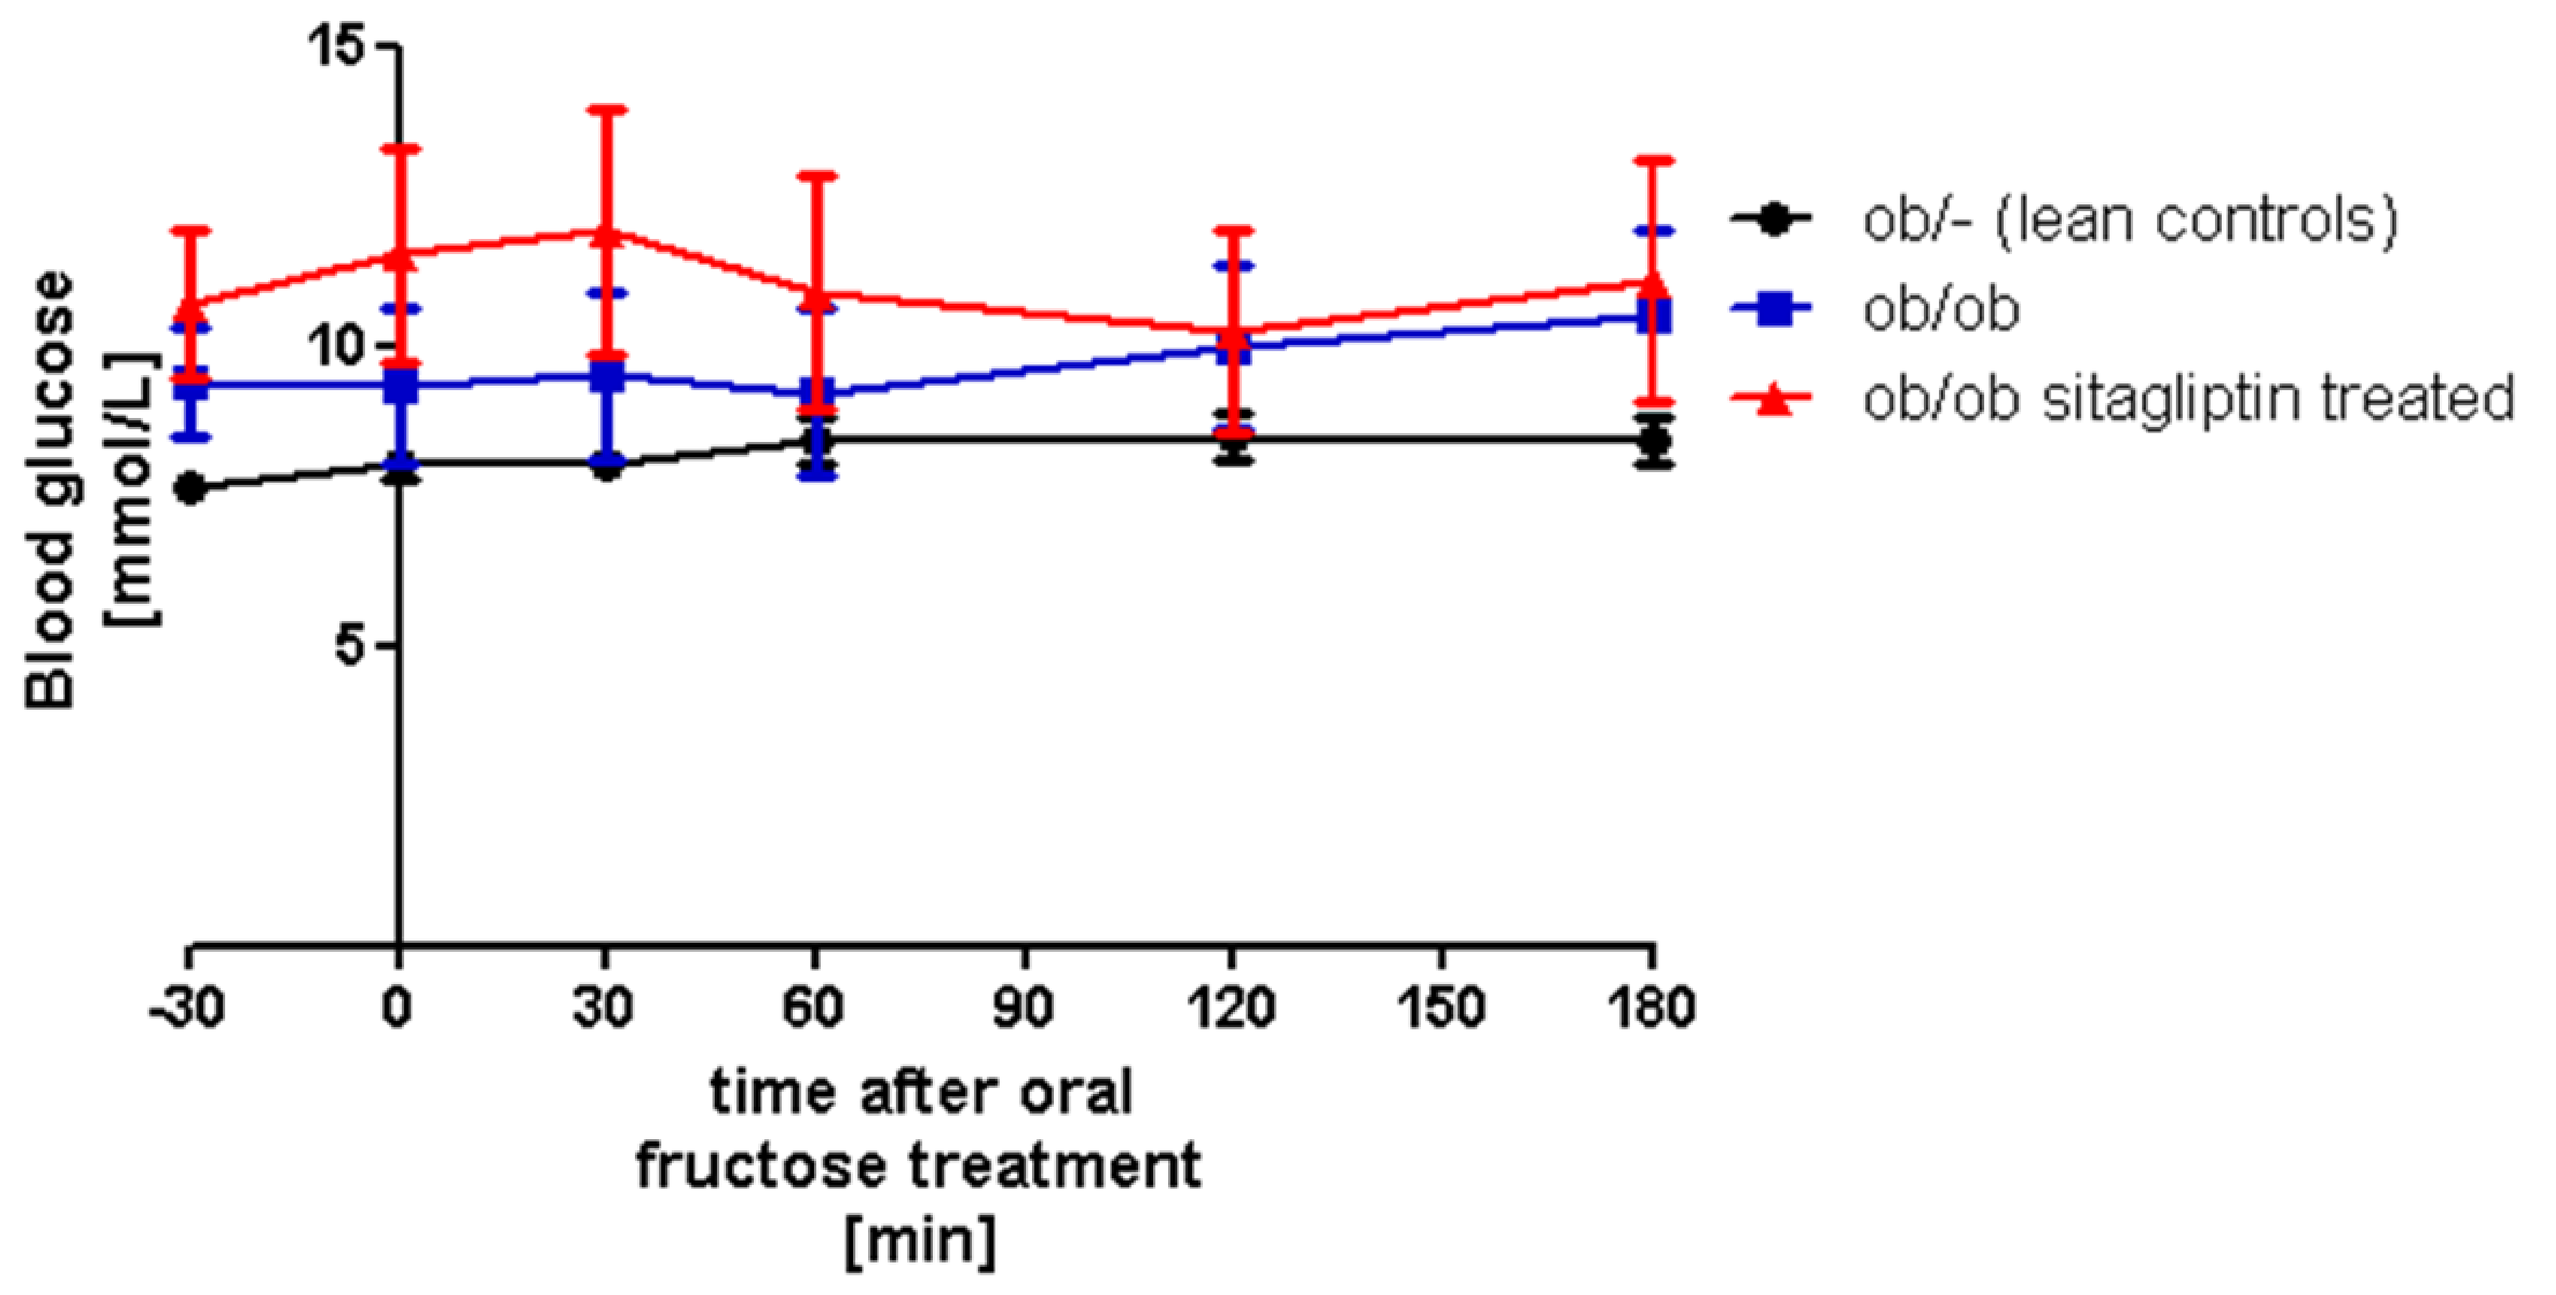
Blood glucose [mmol/L]
15
10
5
-30 0 30 60 90 120 150 180
time after oral fructose treatment [min]
ob/- (lean controls)
ob/ob
ob/ob sitagliptin treated

Fig. 3: Plasma FGF21 after an oral fructose bolus (1g/kg): plasma FGF21 was significantly higher in ob/ob mice compared to lean ob/- mice. However, plasma FGF21 was not responsive to the oral fructose load.

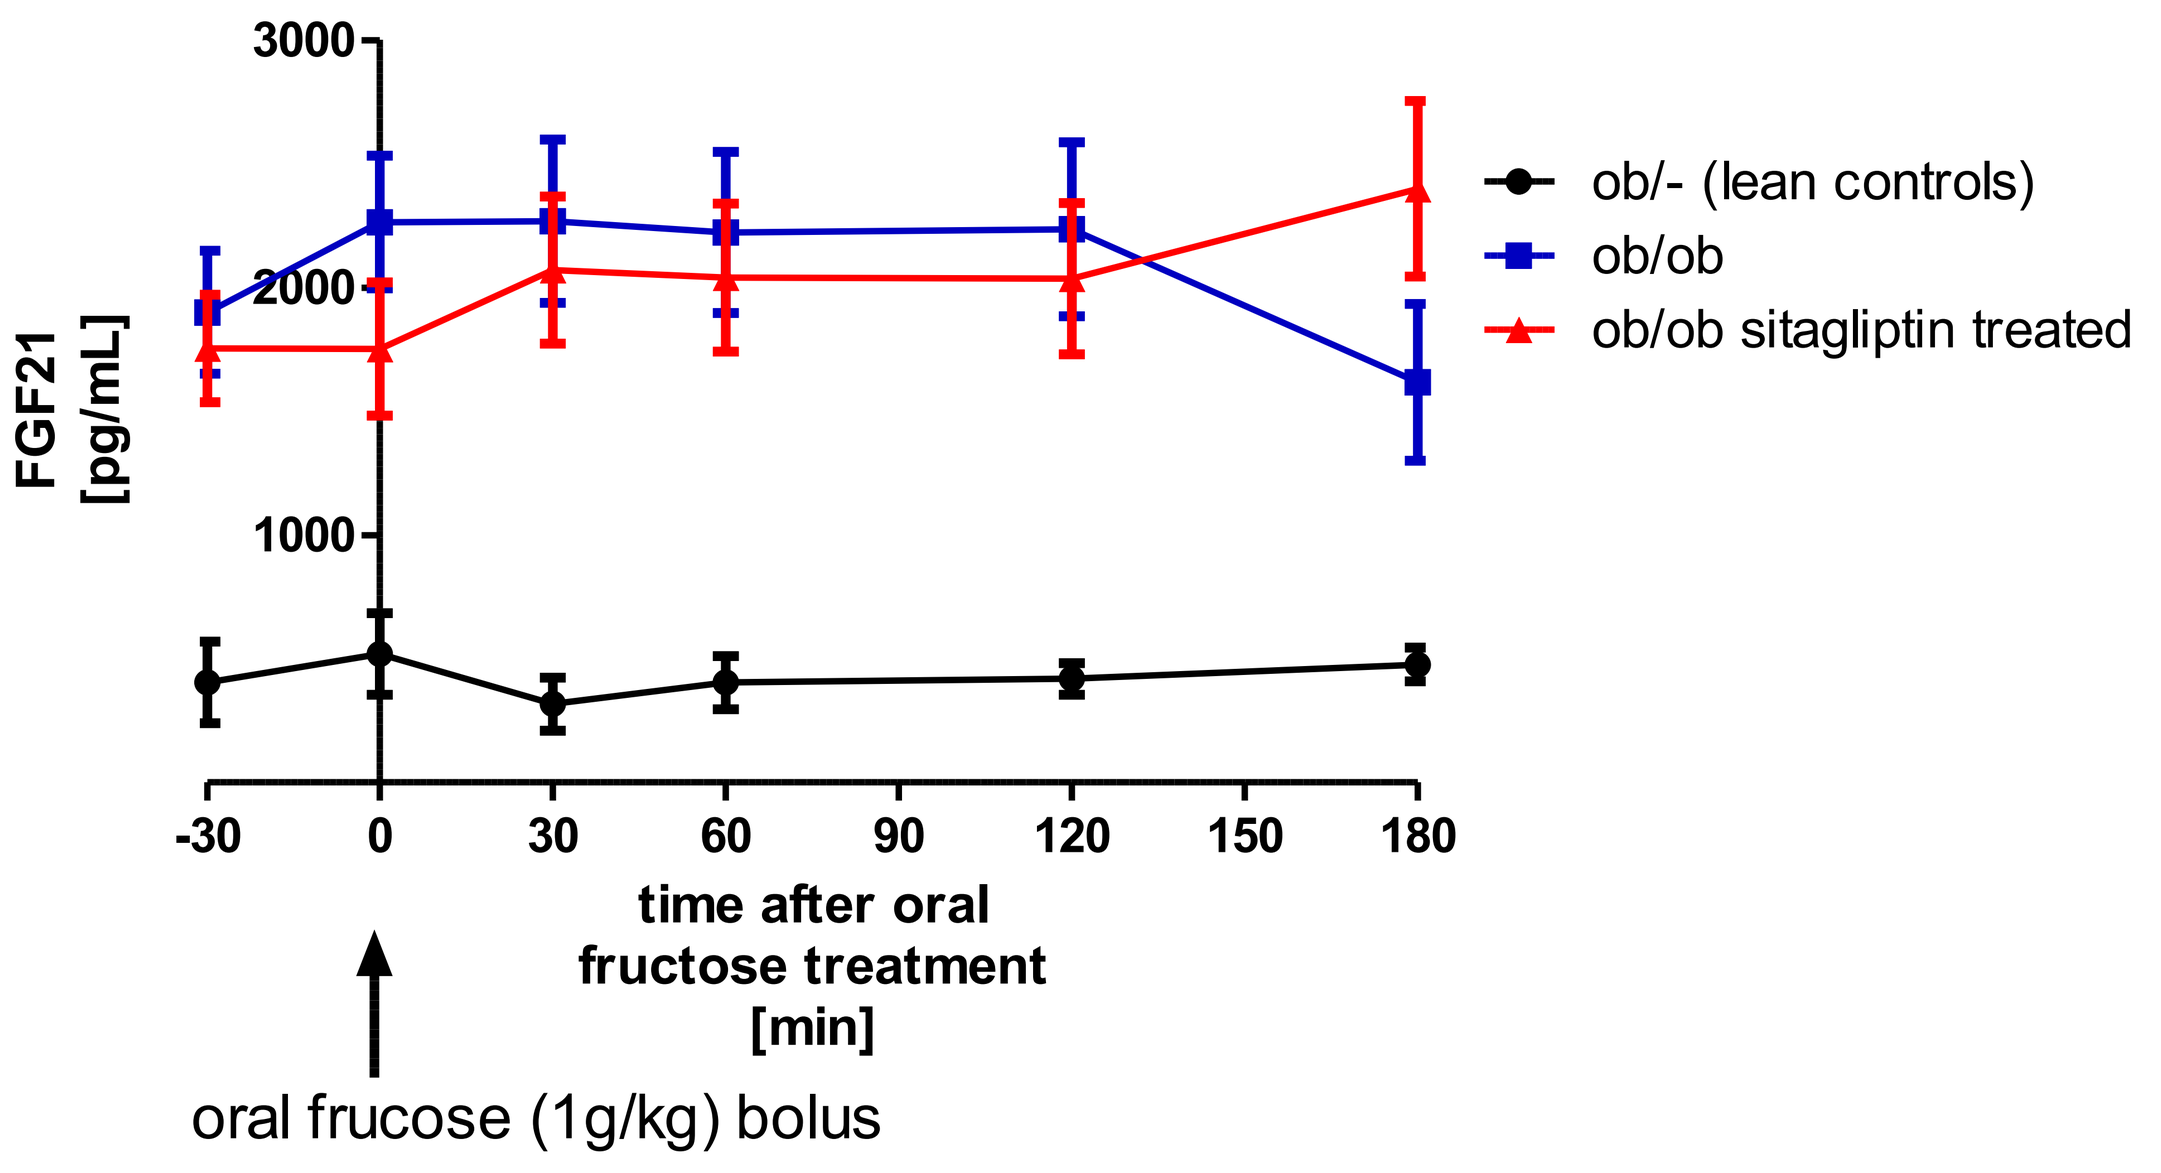
FGF21 [pg/mL]
3000
2000
1000
-30 0 30 60 90 120 150 180
time after oral fructose treatment [min]
oral frucose (1g/kg) bolus
ob/- (lean controls)
ob/ob
ob/ob sitagliptin treated

Rats

Fig. 4: Serum fructose after an oral fructose bolus (1g/kg) in rats: as expected, the oral fructose load led to significant increases in serum fructose concentrations with maximum concentrations approx. 30min after the fructose bolus.

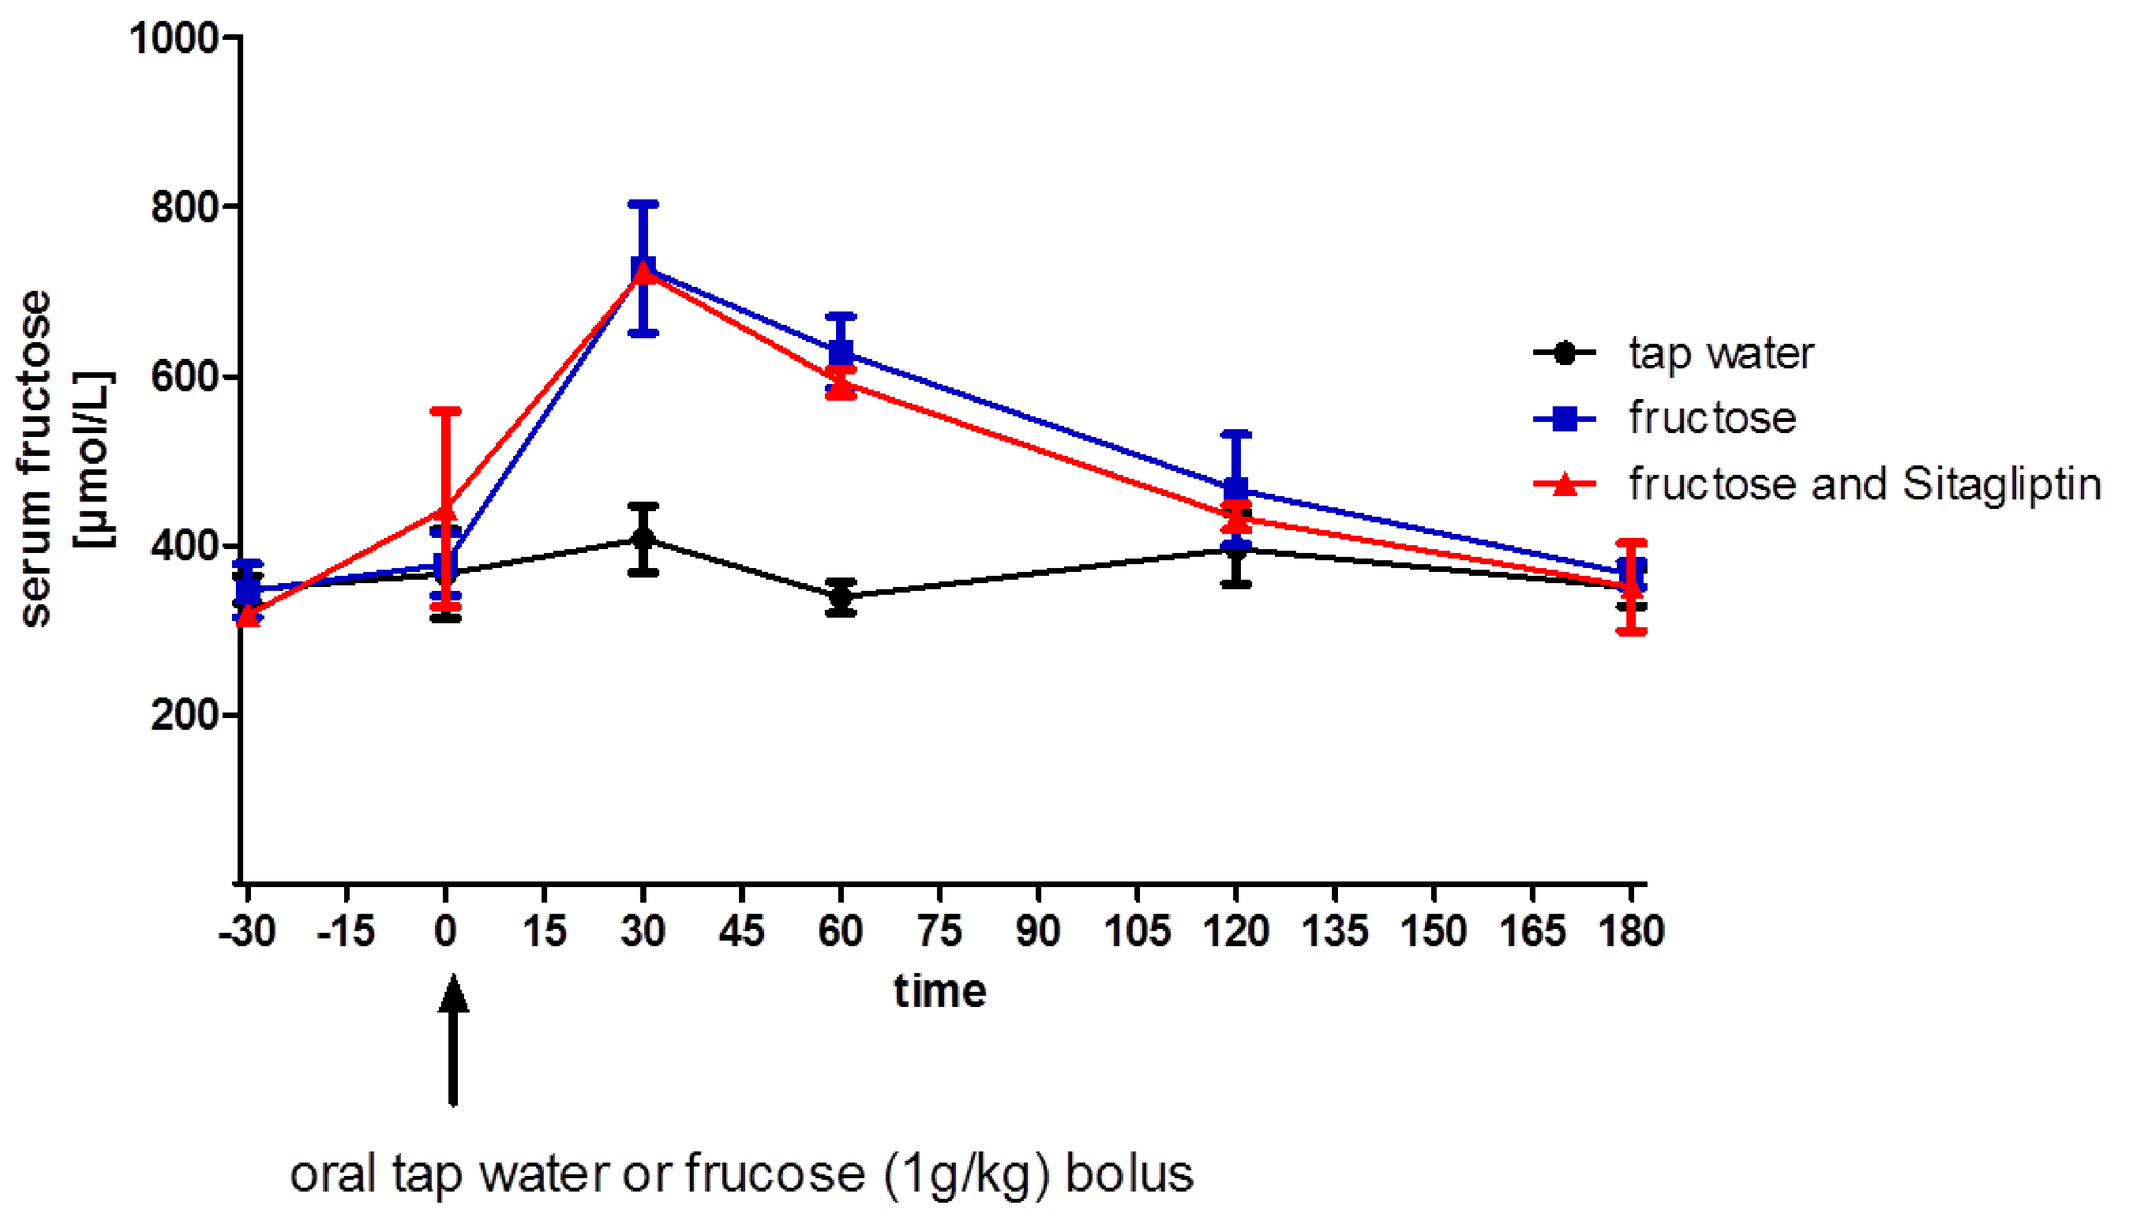
serum fructose [µmol/L]
1000
800
600
400
200
-30 -15 0 15 30 45 60 75 90 105 120 135 150 165 180
time
oral tap water or frucose (1g/kg) bolus
tap water
fructose
fructose and Sitagliptin

Fig. 5: Blood glucose (left graph) and plasma insulin (right graph) after an oral fructose bolus (1g/kg): oral frucose did not significantly affect circulating glucose and insulin.

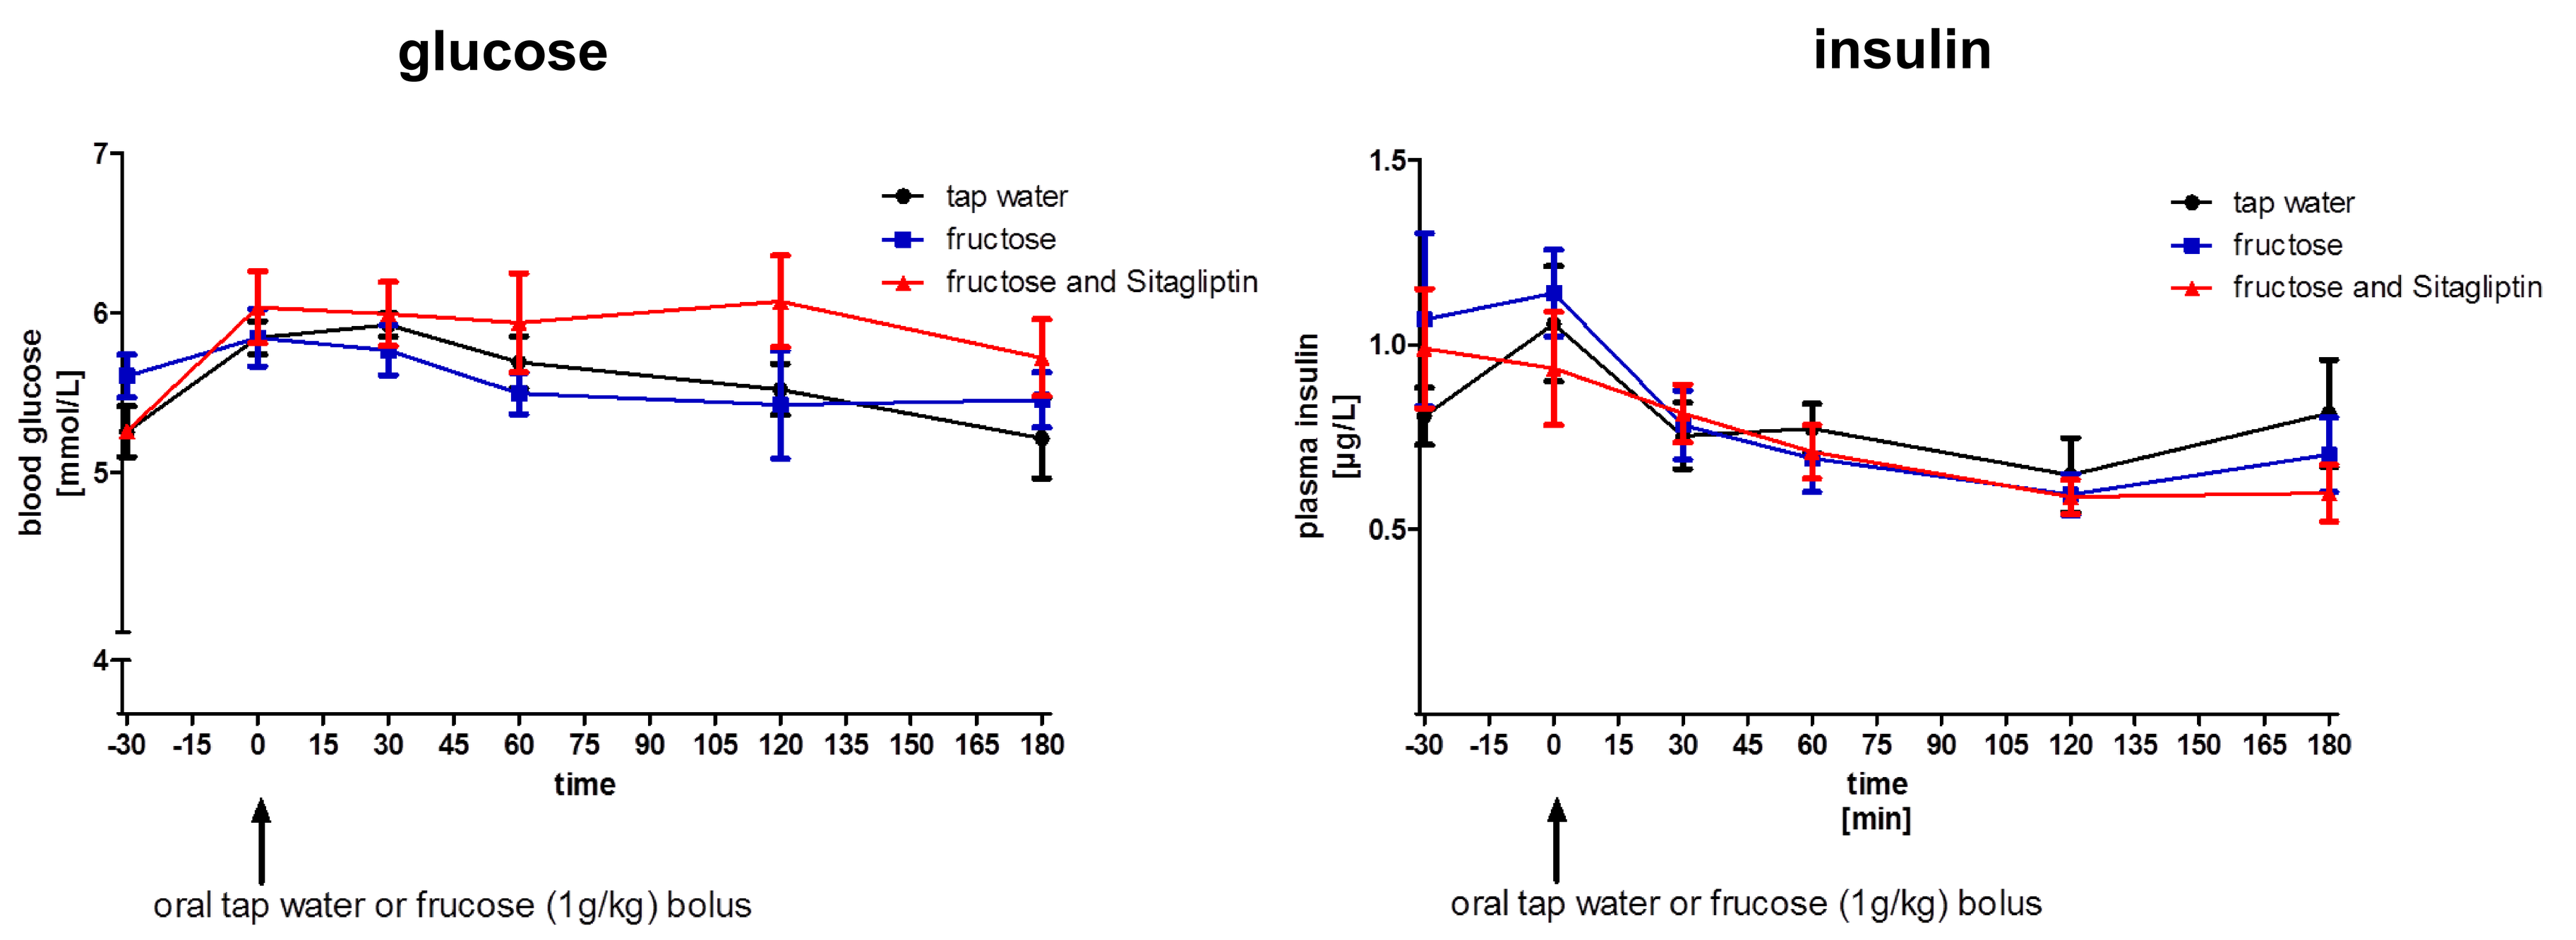
glucose
insulin
blood glucose [mmol/L]
7
6
5
4
-30 -15 0 15 30 45 60 75 90 105 120 135 150 165 180
time
oral tap water or frucose (1g/kg) bolus
plasma insulin [µg/L]
1.5
1.0
0.5
time [min]
tap water
fructose
fructose and Sitagliptin

Fig. 6: Plasma FGF21 after an oral fructose bolus (1g/kg) in rats: in rats, the oral fructose bolus led to a significant increase in plasma FGF21 compared to rats which received tap water, only. FGF21 peak concentrations were detected approx. 60 min after the oral fructose bolus. Sitagliptin pre-treatment had no significant effect on fructose-stimulated plasma FGF21 in healthy rats.

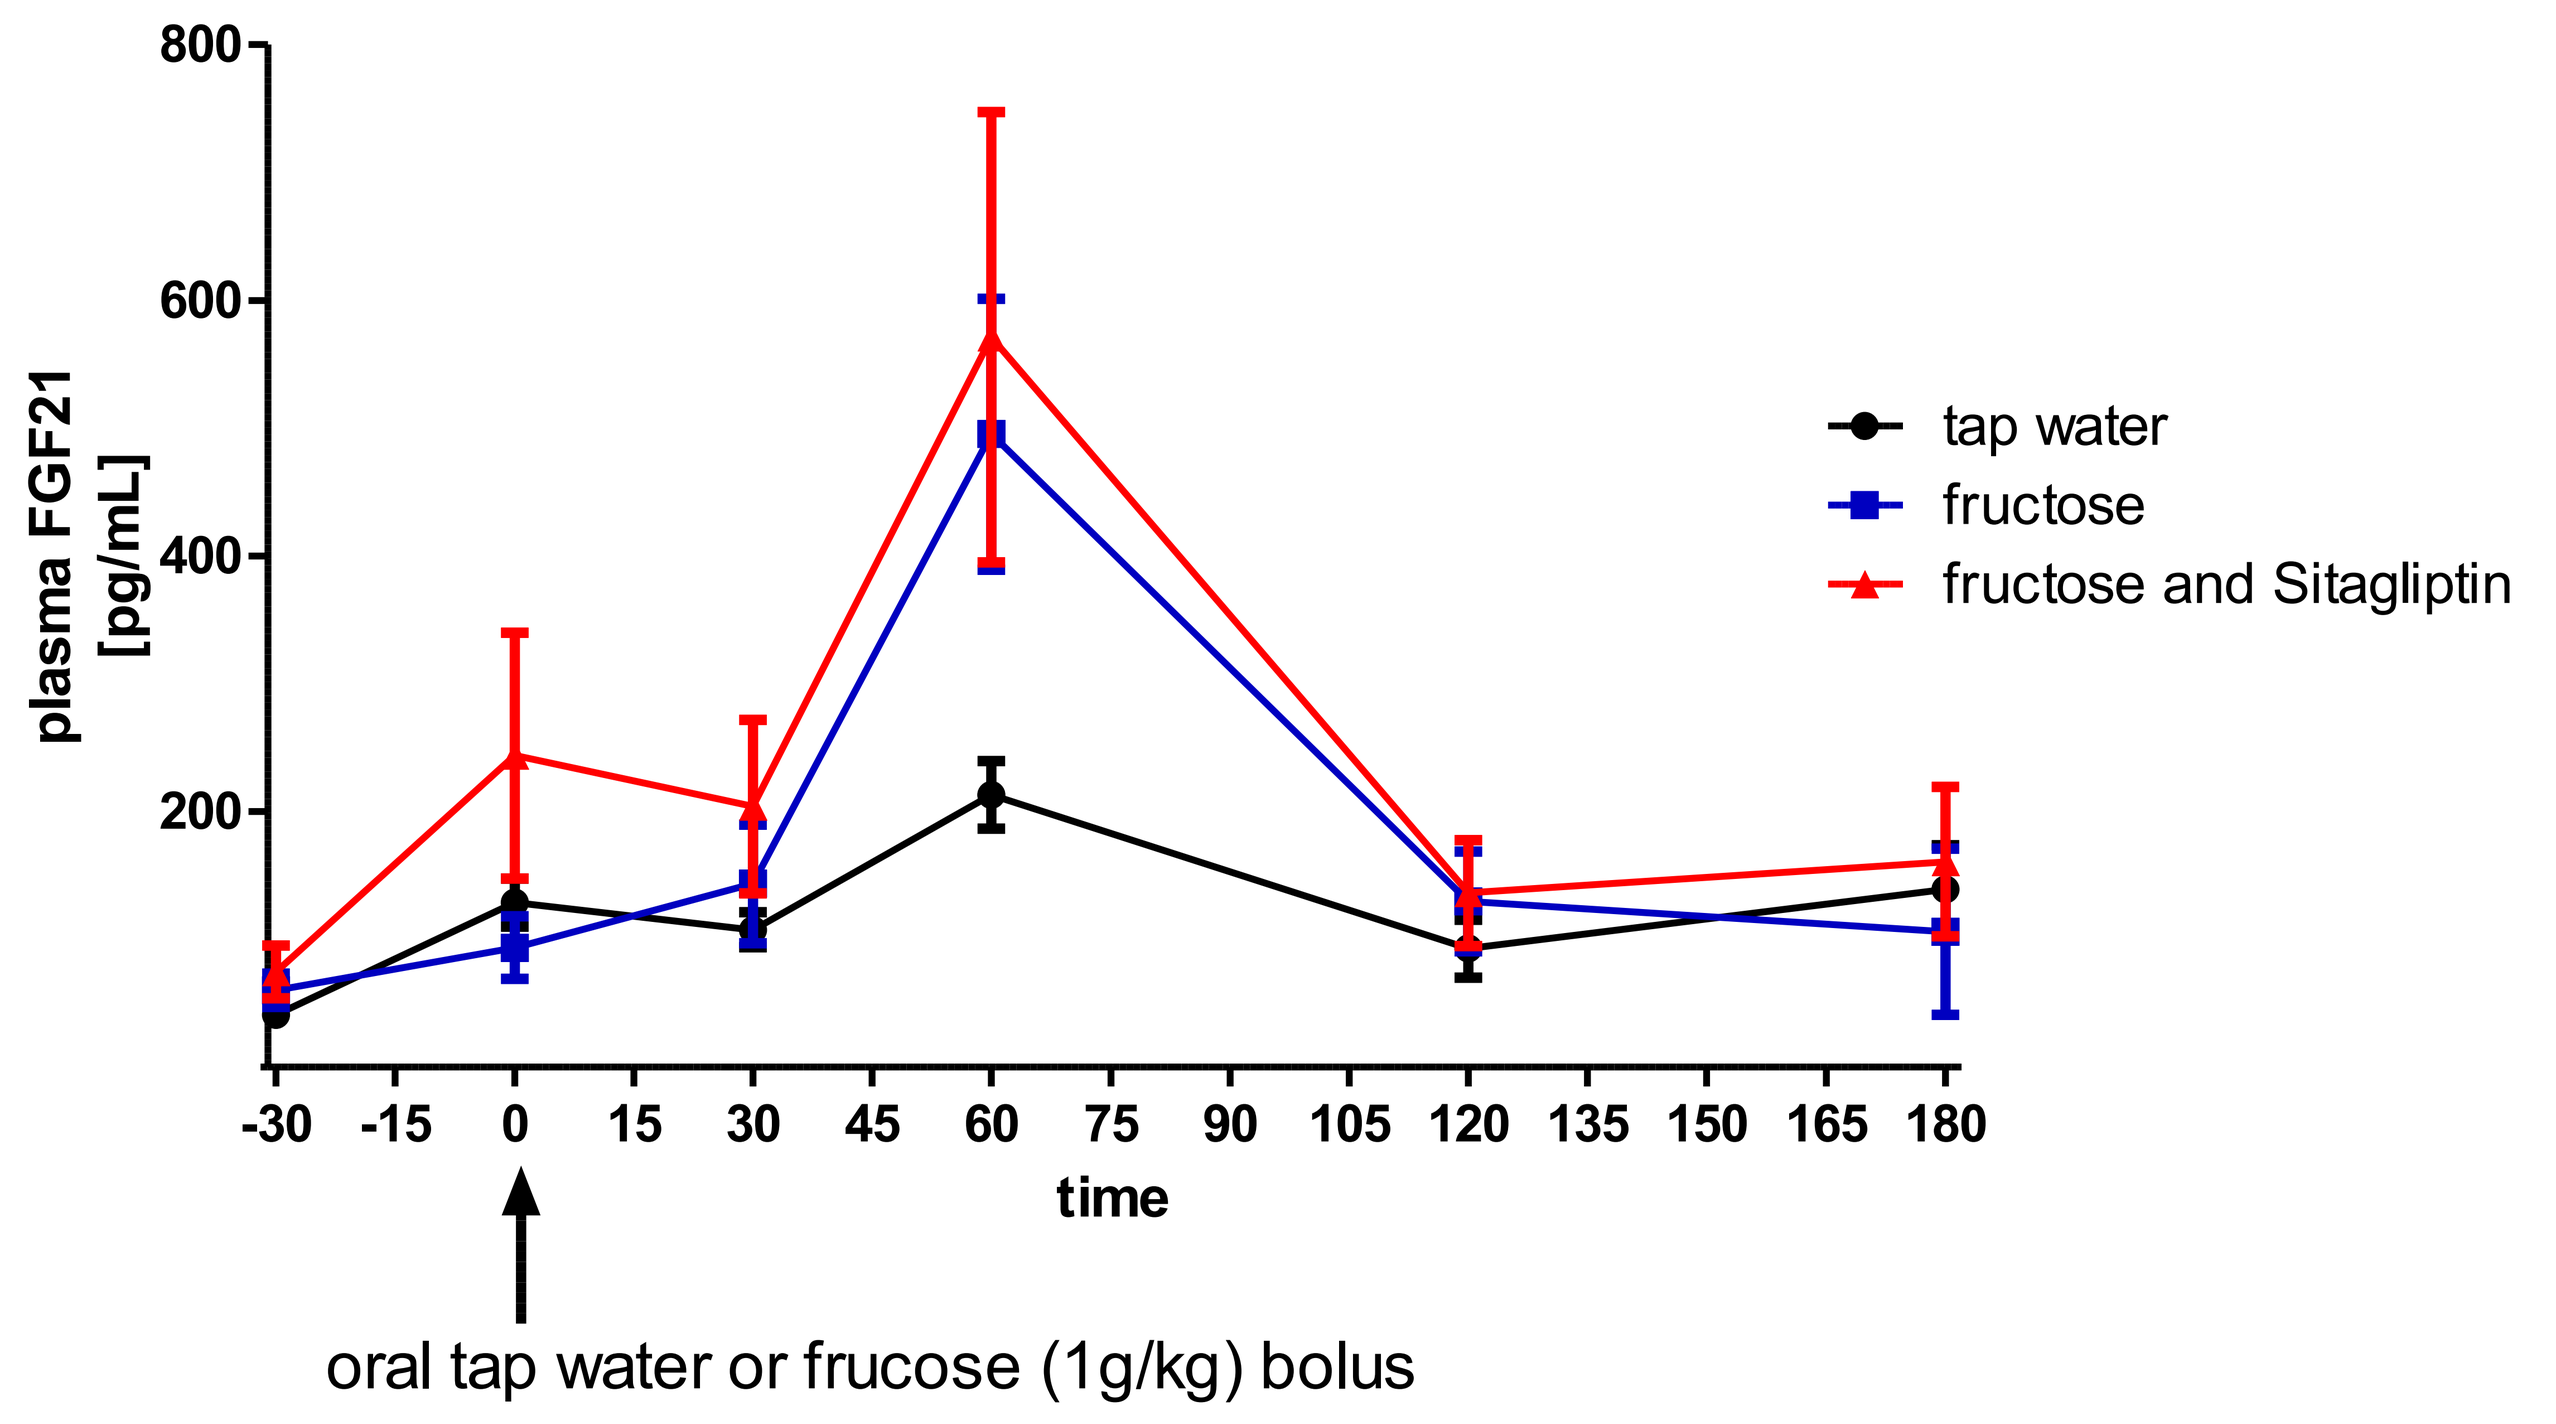
plasma FGF21 [pg/mL]
800
600
400
200
-30 -15 0 15 30 45 60 75 90 105 120 135 150 165 180
time
oral tap water or frucose (1g/kg) bolus
tap water
fructose
fructose and Sitagliptin

Discussion & Conclusion

In humans, Dushay et al. have recently shown that plasma FGF21 is responsive to oral fructose thus paving the way for a new diagnostic tool to investigate oral fructose tolerance. In mice, circulating FGF21 was not responsive to the oral fructose load, at least under the conditions studied. Interestingly, obese ob/ob mice showed significantly higher plasma FGF21 concentrations than lean ob/- mice (irrespective of the oral fructose bolus). In contrast to mice, rats responded to the oral fructose bolus with sharply increased plasma FGF21, while glucose and insulin concentrations were not affected. A single treatment with a DPP4 inhibitor did not affect the FGF21 response in healthy rats. Future studies need to show if the FGF21 response if divergently affected in obese and diabetic vs. healthy rats. It currently remains unclear why FGF21 concentrations show a species specific response to oral fructose. However, one might speculate that rats display significantly lower basal FGF21 concentrations compared to mice which might be easier to stimulate with fructose.

Conclusion: Our data showed that an oral fructose tolerance test with the pharmacodynamic readout FGF21 cannot be easily transferred from humans to laboratory rodents. While mice appear to be not responsive, rats might be used in future studies to investigate oral fructose tolerance.

ECE 2016 429-EP

Diabetes (to include epidemiology, pathophysiology)

Maximilian Bielohuby

Poster presented at: ECE2016 28 – 31 May 2016 MUNICH, GERMANY 18th EUROPEAN CONGRESS OF ENDOCRINOLOGY

PosterSessionOnline.com