Restoring the circadian cortisol rhythm with metyrapone in patients with adrenal incidentalomas and subclinical hypercortisolism reduces IL-6 levels

Miguel Debono, Robert F Harrison, Rita Chadarevian, Carole Gueroult, Jean-Louis Abitbol, John Newell-Price

University of Sheffield, Sheffield, UK (M.D., R.F.H, J.N-P.), HRA Pharma, Paris, France (R.C., C.G., J-L.A.)

Introduction

- Adrenal incidentalomas are present in 10% of the elderly population
- Depending on criteria used, between 4 to 47% of these adrenocortical adenomas secrete excess cortisol
- Adrenal incidentalomas with subclinical hypercortisolism are associated with increased cardiovascular risk factors, cardiovascular events, vertebral fractures and an elevated mortality rate [1, 2]
- A disturbed cortisol rhythm is associated with the development of Type 2 diabetes, obesity and cardiovascular mortality
- Il-6 is high in patients with Cushing's syndrome and the presence of this cytokine is associated with endothelial dysfunction and hence implicated in the pathogenesis of atherosclerosis

Hypothesis

- Patients with adrenal incidentalomas and subclinical hypercortisolism have an abnormal cortisol rhythm resulting in excess nocturnal cortisol exposure and this is associated with elevated IL-6 levels.
- By using metyrapone, a short acting 11β-hydroxylase inhibitor, we aimed to reset the cortisol rhythm to normal and test whether this leads to a change in IL-6 concentrations.

Methods

- Study group (dexamethasone cortisol > 50nmol/L & ACTH <10pg/mL or dexamethasone cortisol > 80nmol/L)
 - 6 patients with adrenal incidentalomas (AI) and subclinical hypercortisolism (SCH)
- Control groups (dexamethasone cortisol <50nmol/L)
 - i) 6 patients with adrenal incidentalomas
 - ii) 6 healthy, normal volunteers with no adrenal incidentaloma by MRI
- 24 hour sampling for serum cortisol, IL-6 and day time salivary cortisol starting in the afternoon on a Clinical Research Facility
- Part A: We initially analysed baseline cortisol exposure and IL-6 concentrations for each group using area under the curve and studied the differences in rhythms between groups
- Part B: By using metyrapone we intervened at specific times to alter the differences in rhythm between the groups and leaving the rest of the rhythms unaltered

Serum cortisol rhythm in patients with adrenal incidentalomas and SCH and control groups

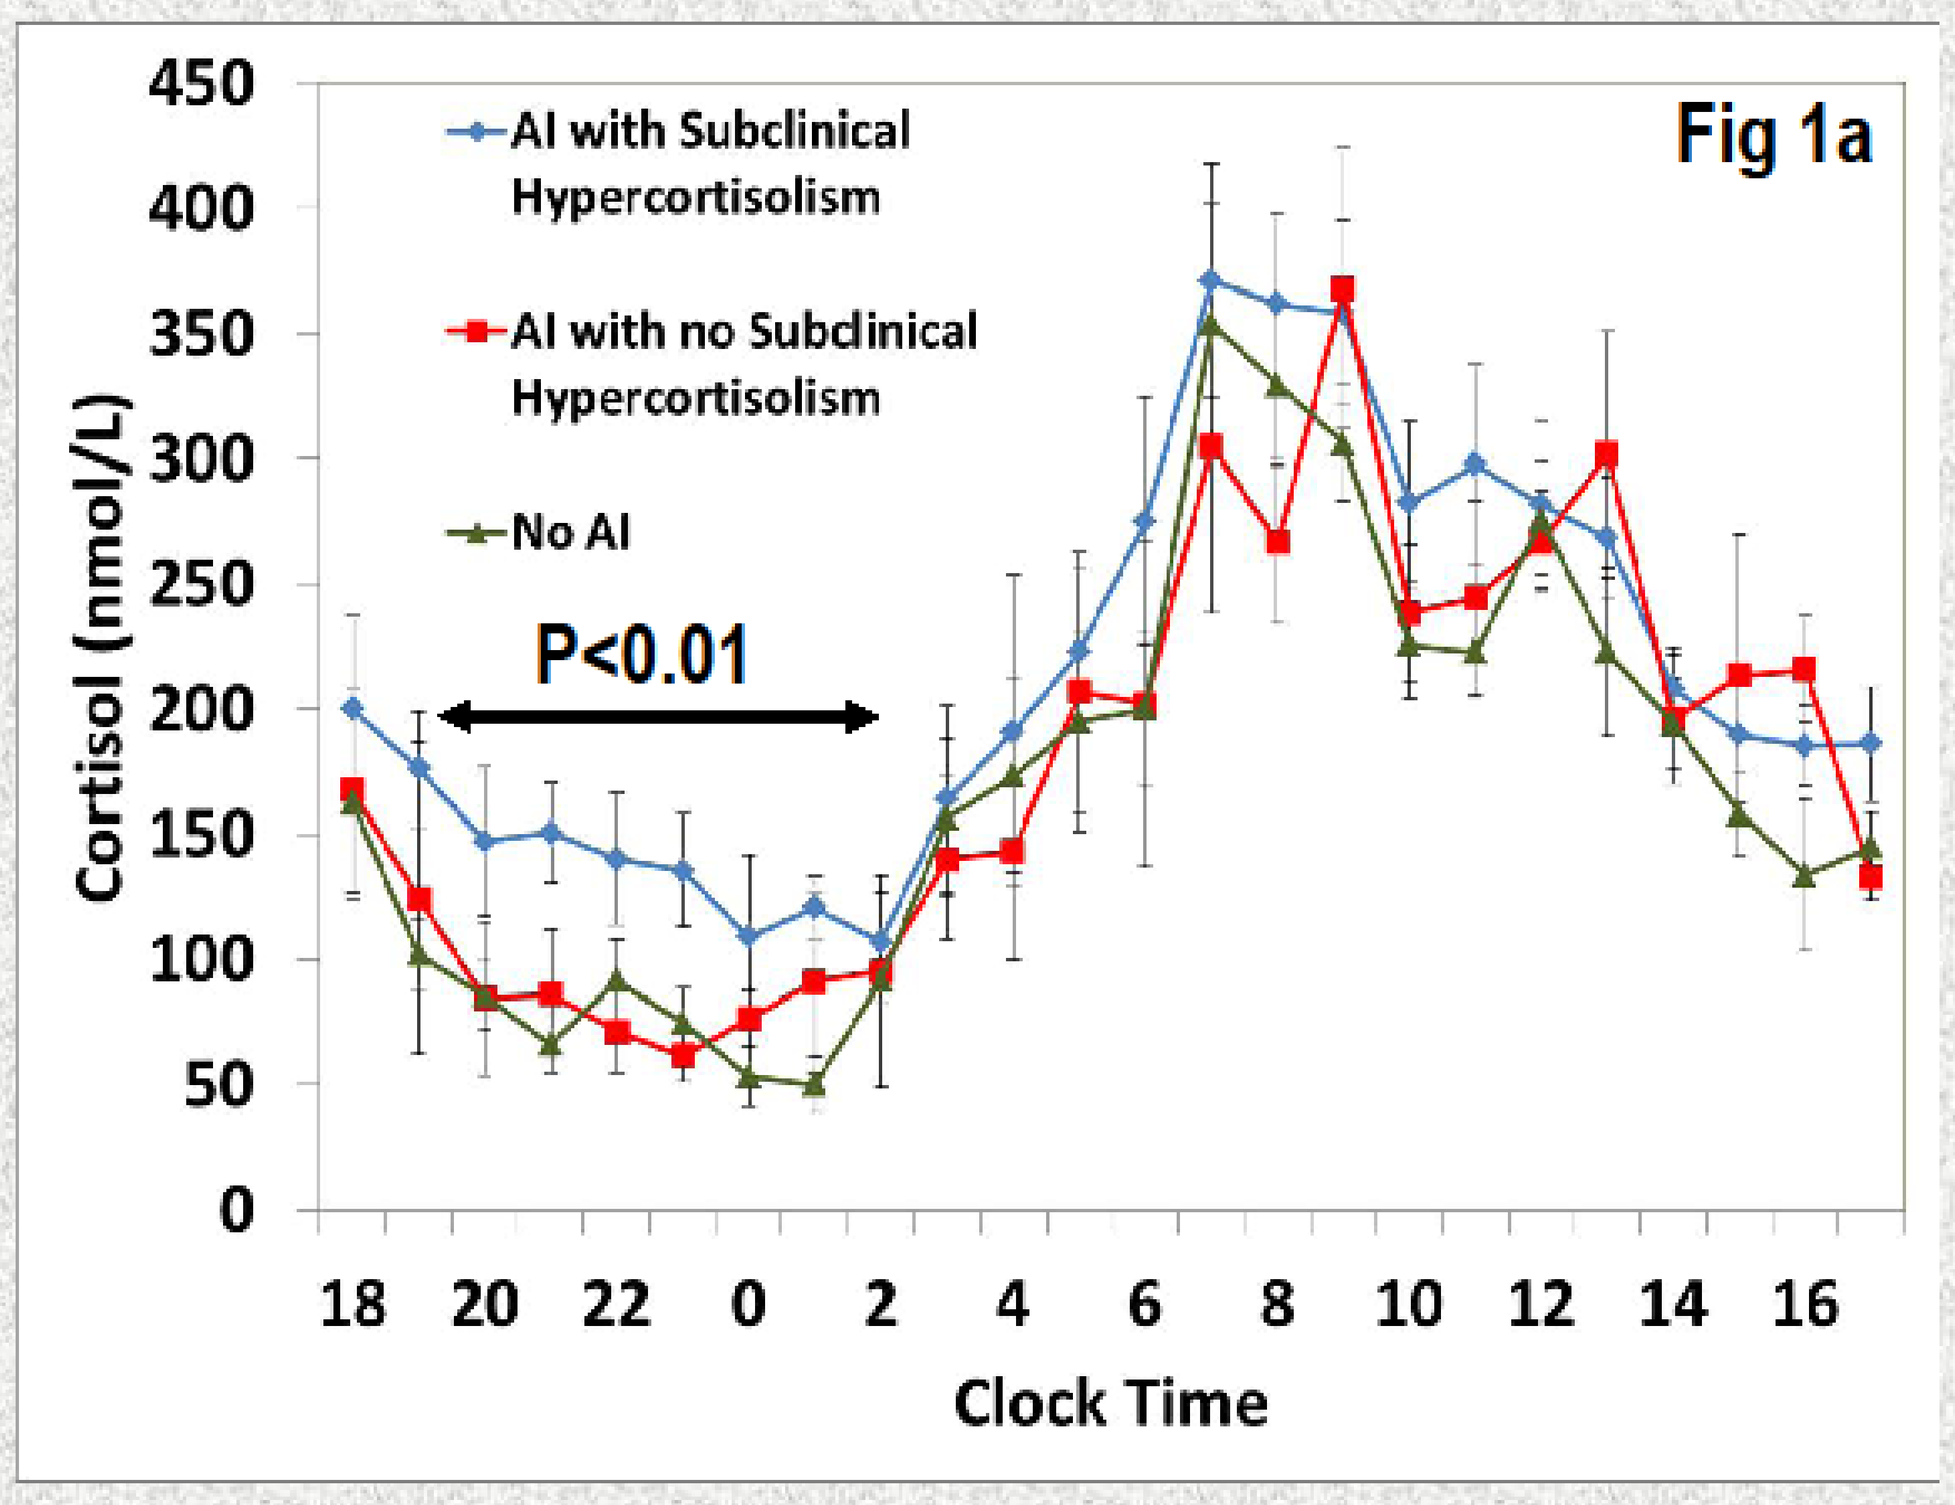
Fig 1a
AI with Subclinical Hypercortisolism
AI with no Subclinical Hypercortisolism
No AI
P<0.01
Cortisol (nmol/L)
Clock Time

This graph demonstrates the differences in cortisol rhythm between patients with SCH and controls with no SCH. Patients with SCH have higher nocturnal cortisol exposure when compared to other groups. AI: adrenal incidentalomas

Re-setting the serum cortisol rhythm in patients with adrenal incidentalomas and SCH using metyrapone

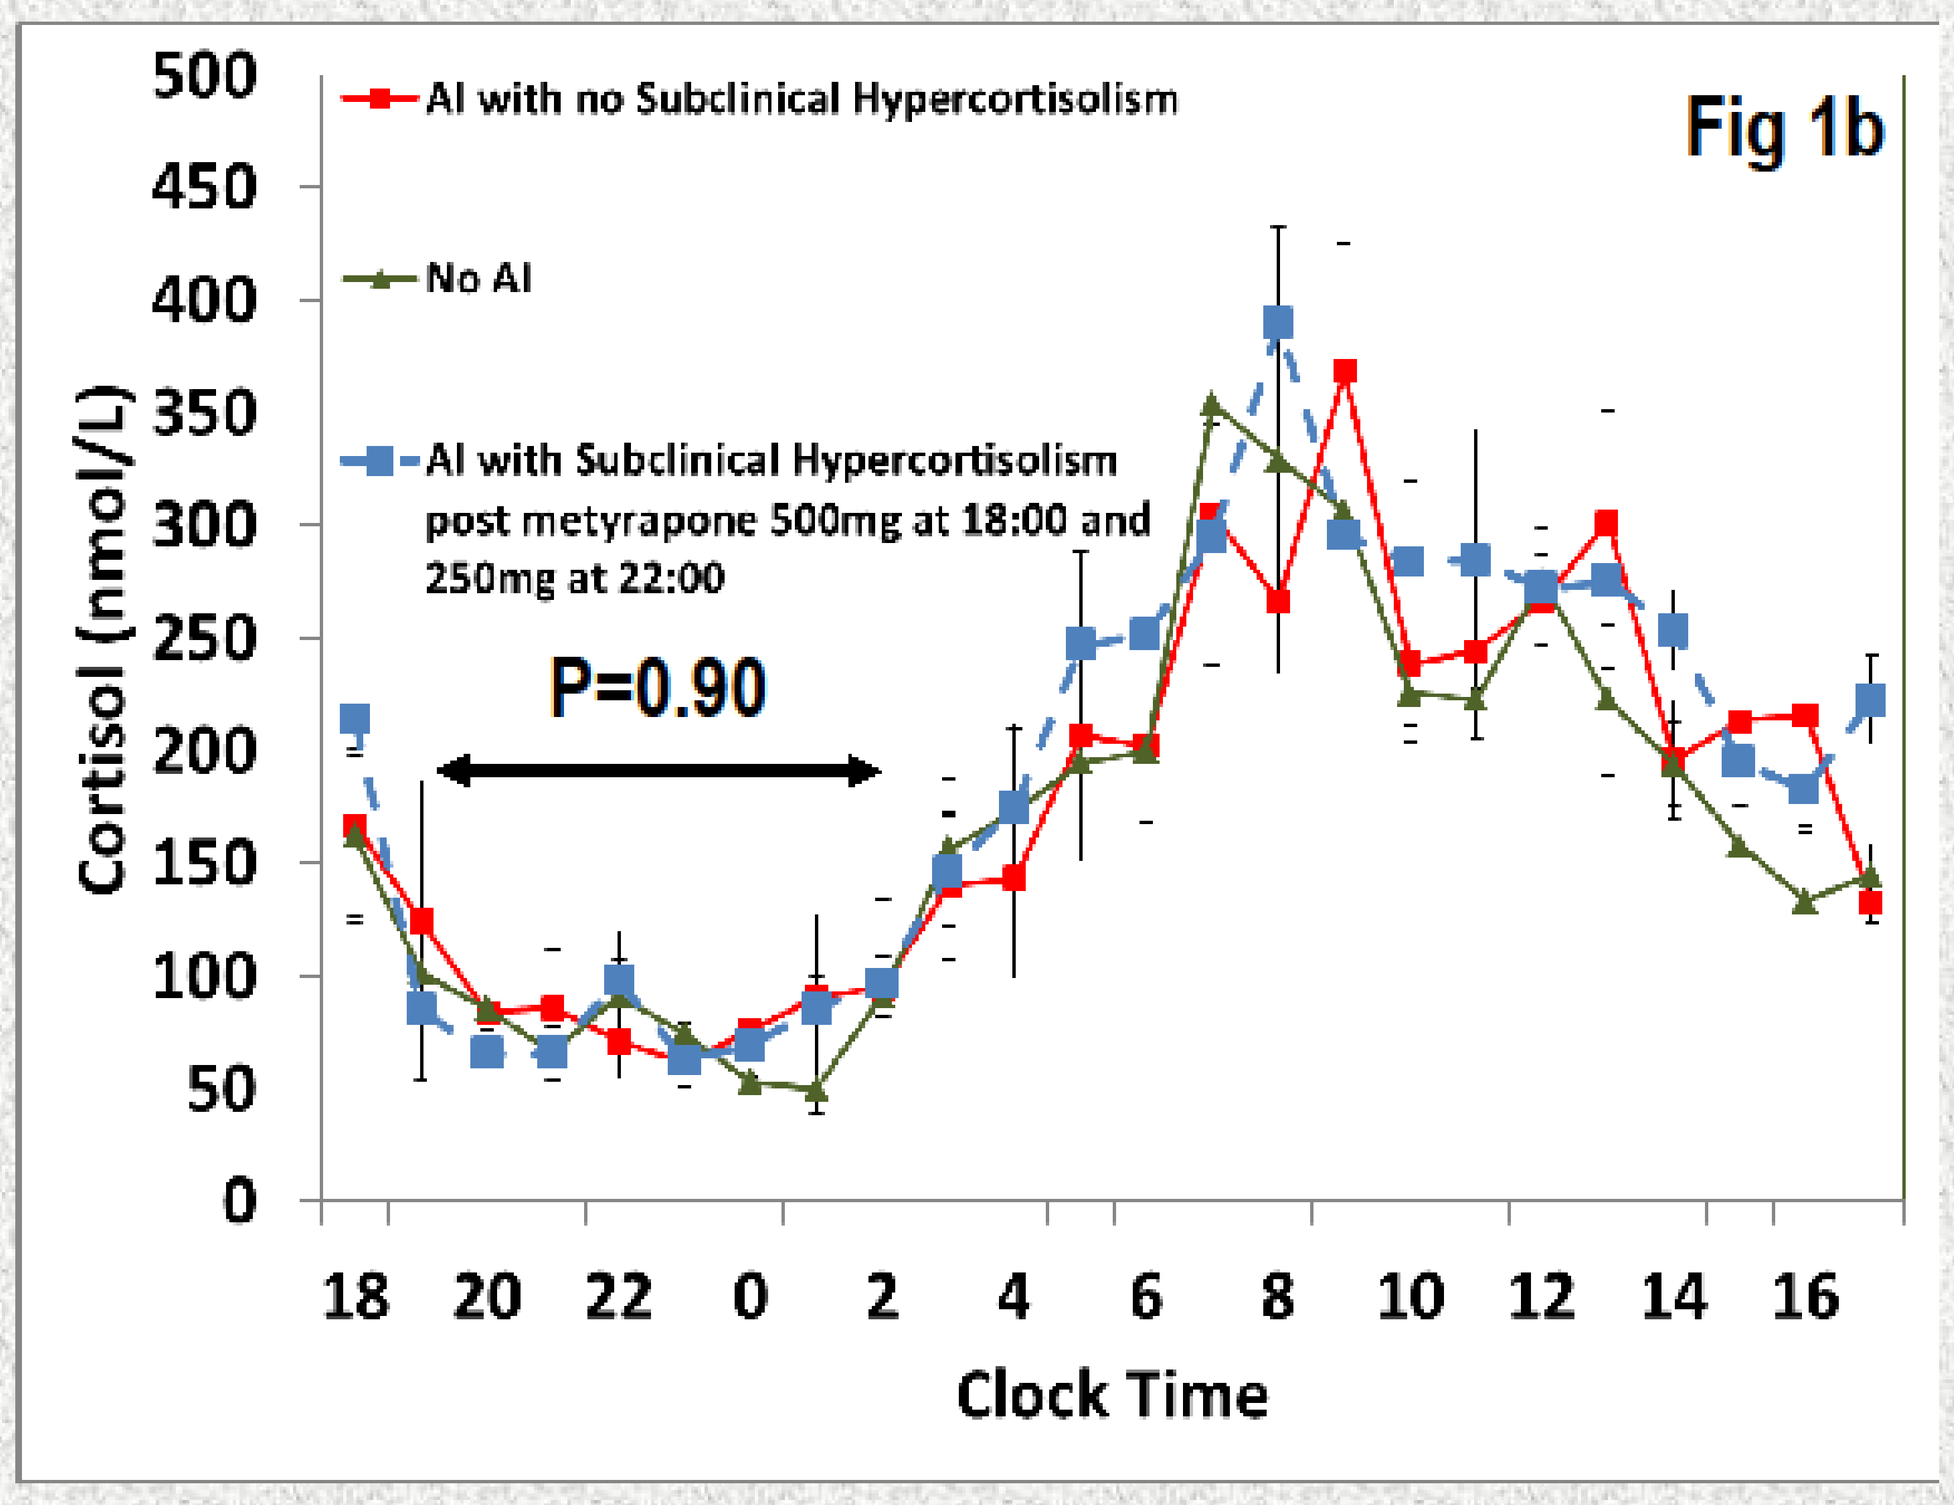
Fig 1b
AI with no Subclinical Hypercortisolism
No AI
AI with Subclinical Hypercortisolism post metyrapone 500mg at 18:00 and 250mg at 22:00
P=0.90
Cortisol (nmol/L)
Clock Time

This graph shows the re-setting of cortisol rhythm in patients with SCH after administration of metyrapone 500mg at 18:00 and 250mg at 22:00. After metyrapone there were no significant differences in cortisol exposure at any time. 08:00 cortisol was not altered, that is was not too low, indicating drug safety.

Salivary Cortisone: A surrogate marker for free cortisol to detect hypercortisolism in patients with adrenal incidentalomas

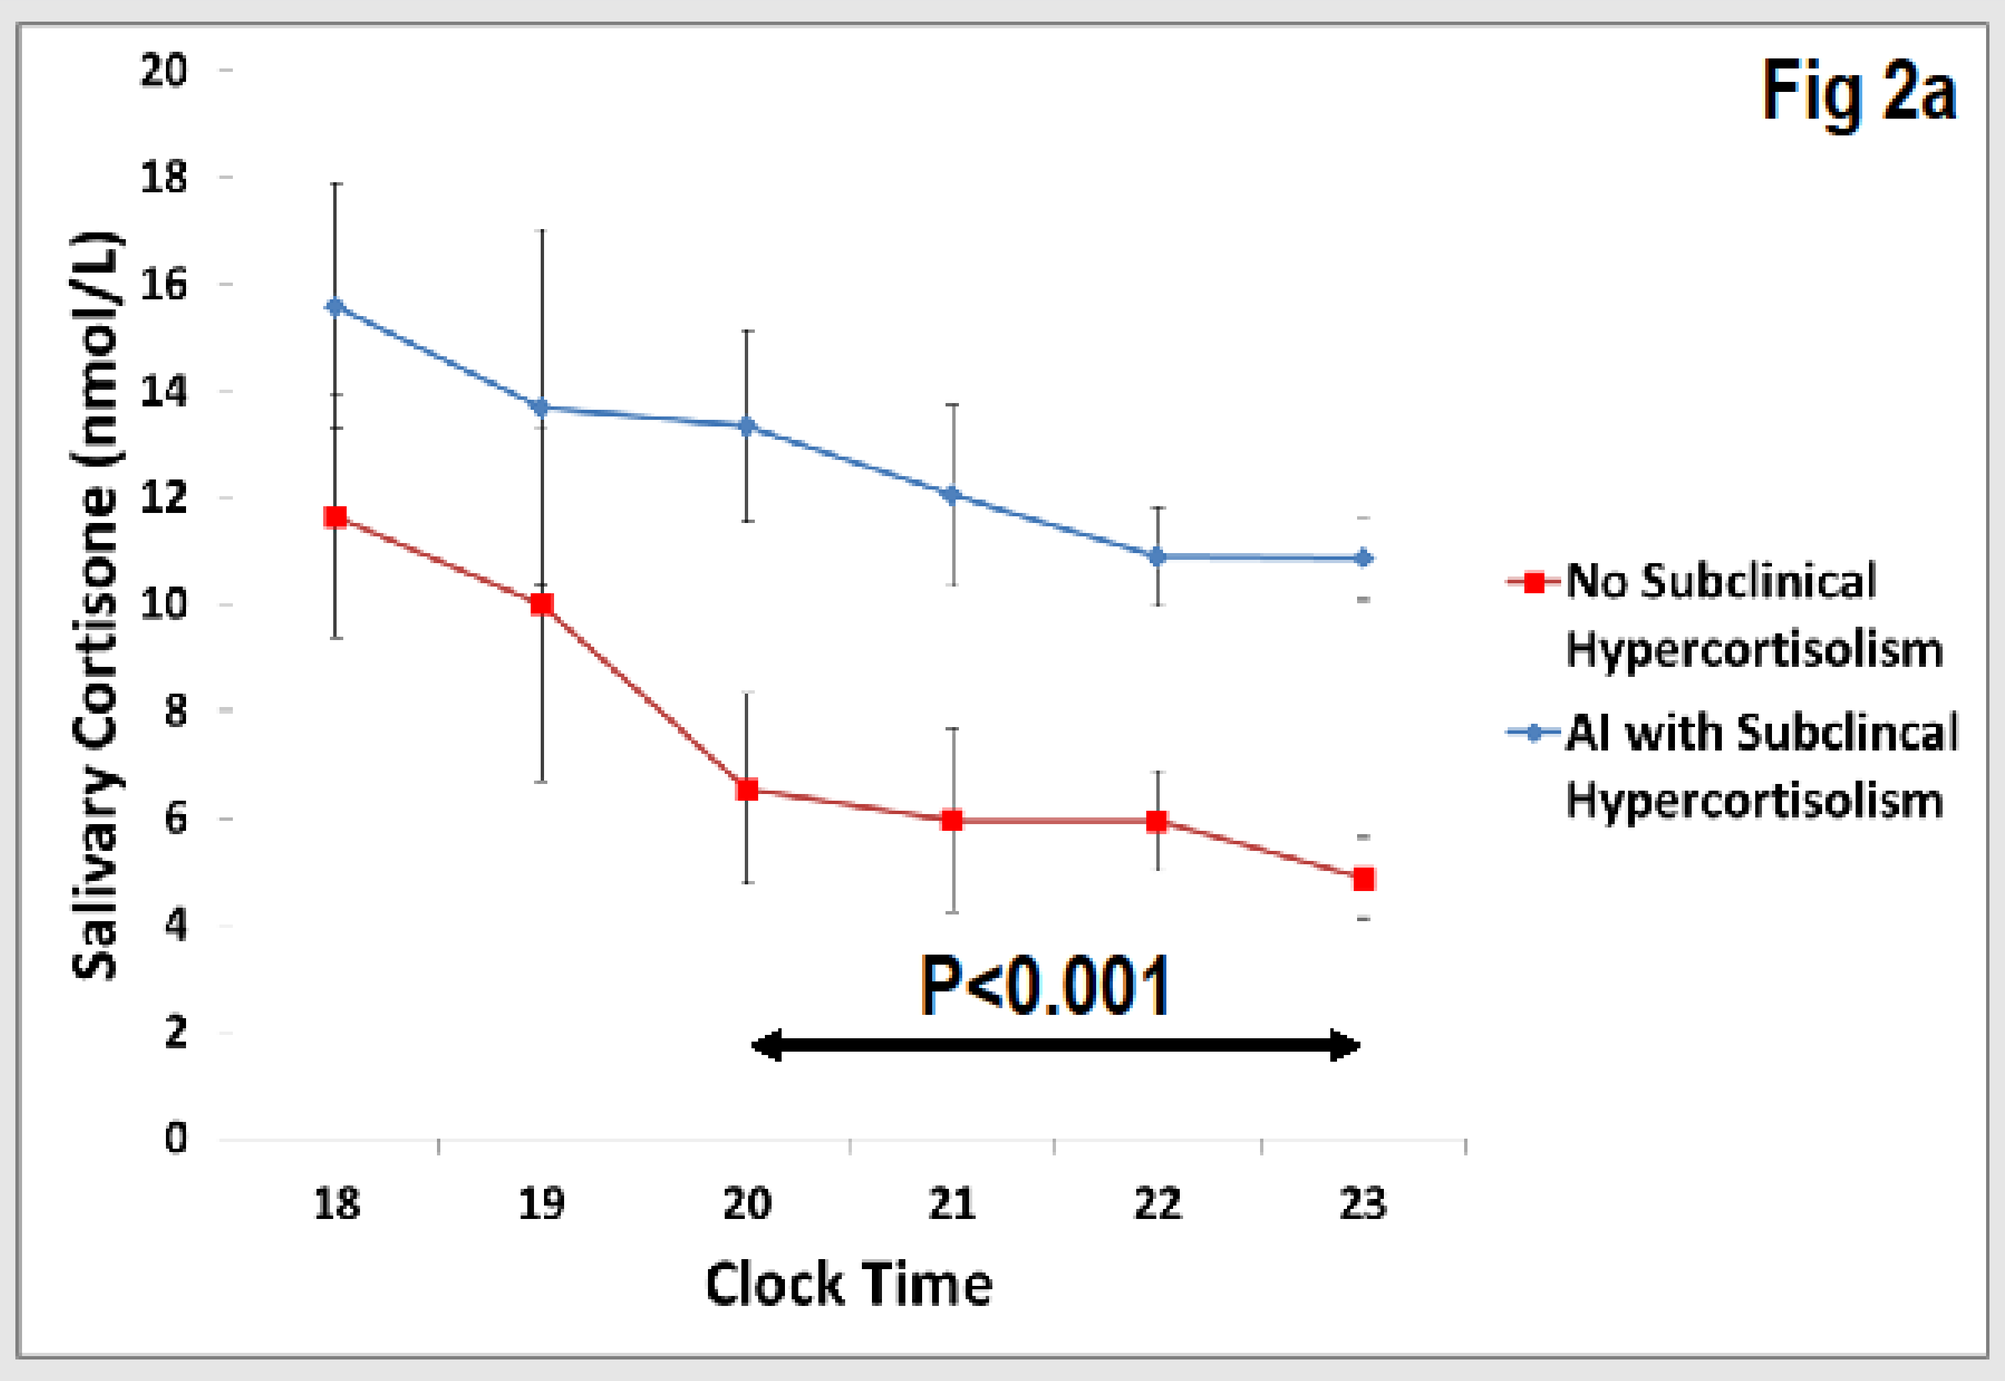
Fig 2a
No Subclinical Hypercortisolism
AI with Subclincal Hypercortisolism
P<0.001
Salivary Cortisone (nmol/L)
Clock Time

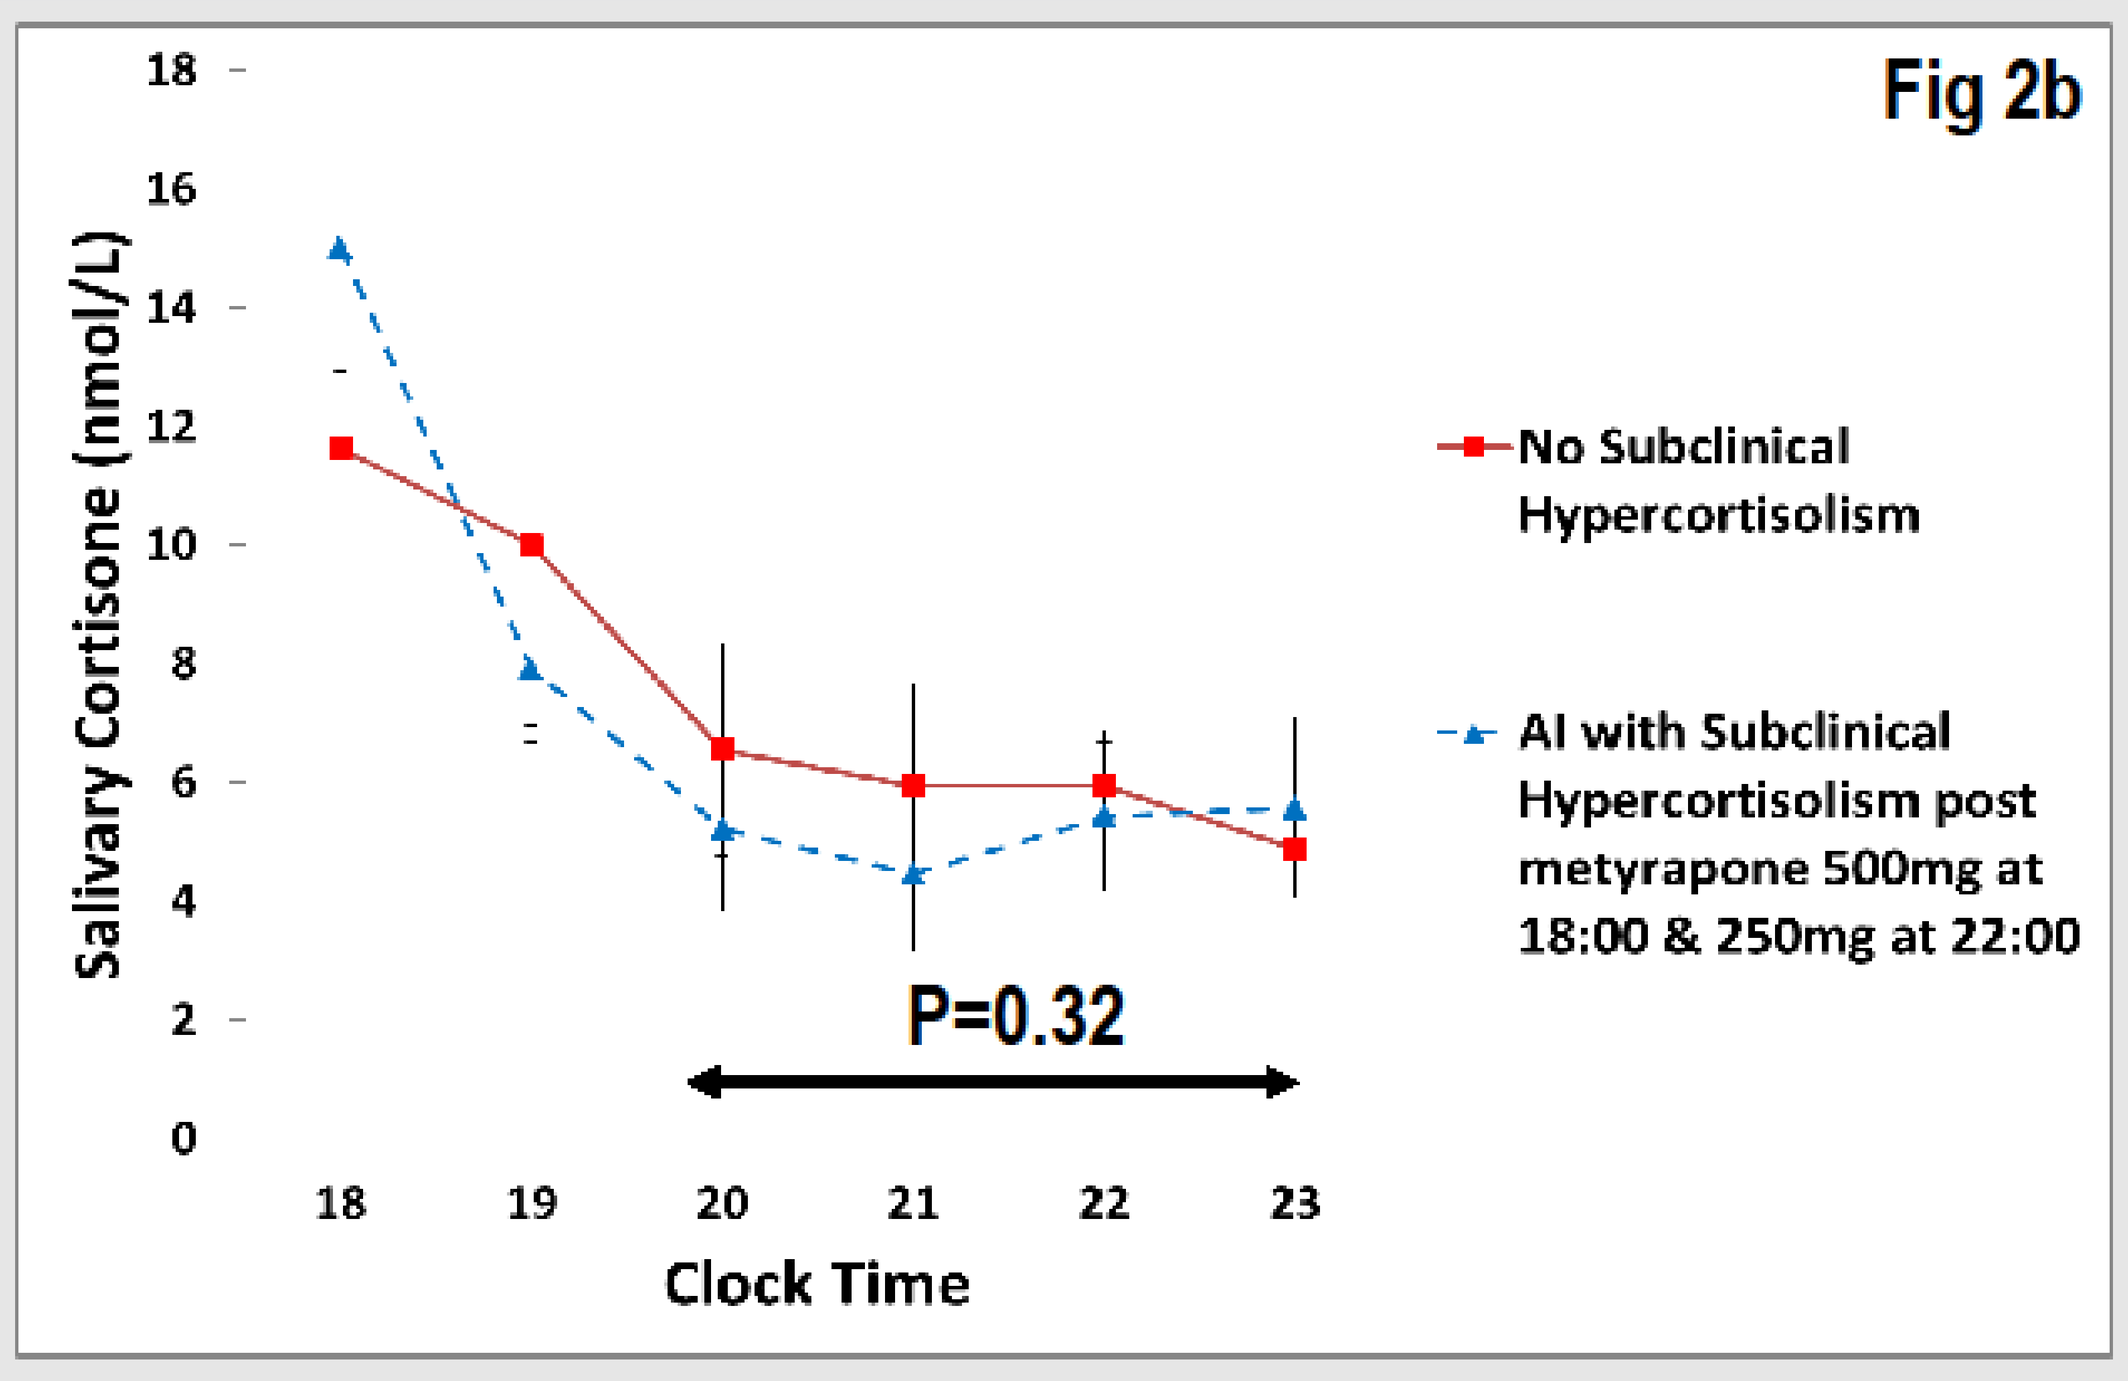
Fig 2b
No Subclinical Hypercortisolism
AI with Subclinical Hypercortisolism post metyrapone 500mg at 18:00 & 250mg at 22:00
P=0.32
Salivary Cortisone (nmol/L)
Clock Time

Salivary cortisone[3] was shown to detect the differences in cortisol exposure at baseline (Fig 2a). After metyrapone there was no difference between patients with SCH and the other control groups combined together (No Subclinical Hypercortisolism) (Fig 2b).

IL-6 concentrations in patients with adrenal incidentalomas

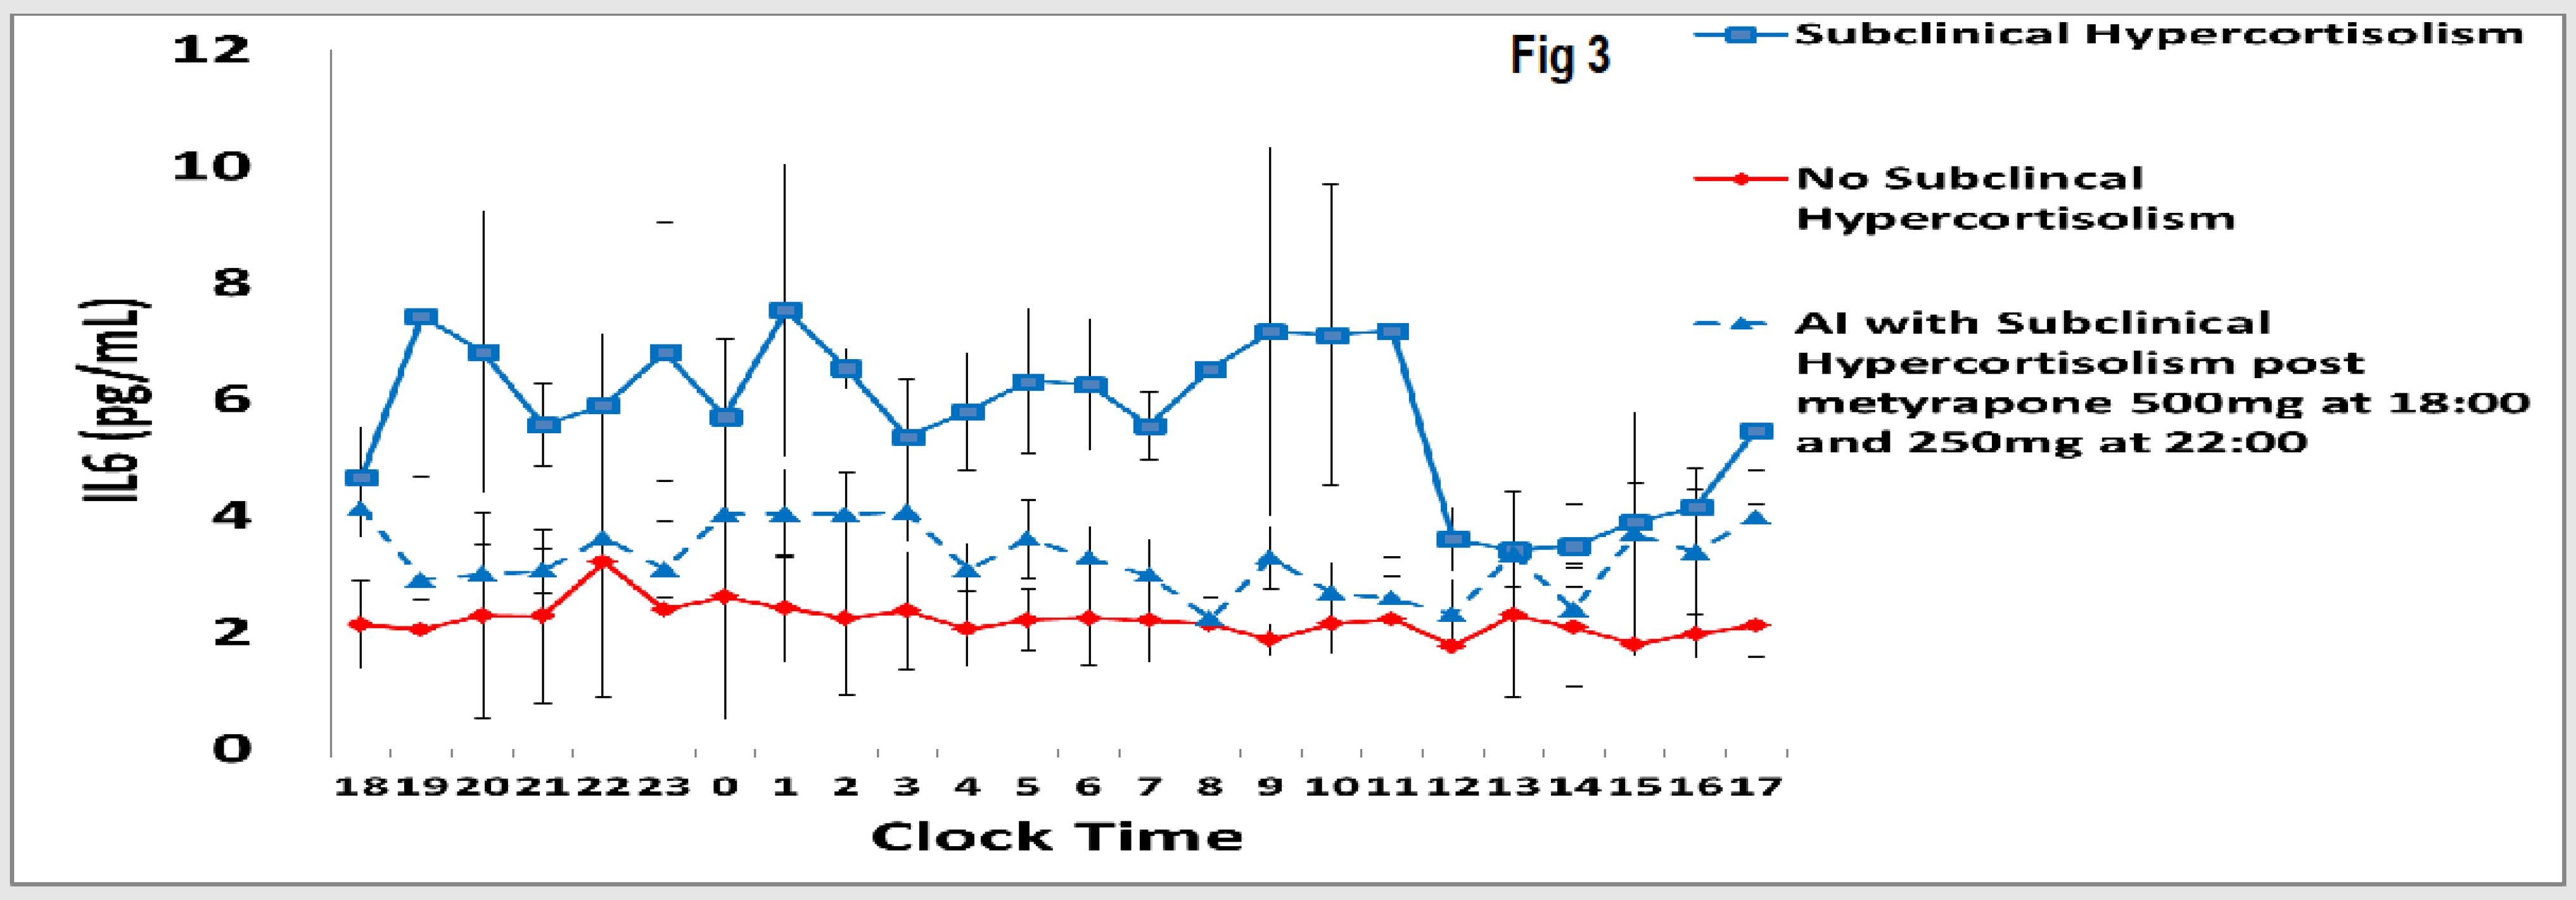
Fig 3
Subclinical Hypercortisolism
No Subclincal Hypercortisolism
AI with Subclinical Hypercortisolism post metyrapone 500mg at 18:00 and 250mg at 22:00
IL6 (pg/mL)
Clock Time

Graph highlights elevated IL-6 levels, a cortisol dependent marker, in patients with adrenal incidentalomas and SCH compared to the combined control groups with no SCH at baseline (continuous lines). Levels are rapidly restored to normal in patients with SCH after the administration of metyrapone potentially resulting in a decrease in cardiovascular risk (dotted line).

Conclusions

- Adrenal incidentalomas with SCH have a disturbed cortisol rhythm with increased nocturnal cortisol exposure
- The cortisol rhythm may be restored by administering metyrapone at specific times of the day
- Salivary cortisone may be used to detect higher nocturnal cortisol exposure in patients with SCH
- IL-6, a marker of cardiovascular risk, is elevated in patients with SCH and concentrations are immediately lowered after metyrapone administration
- A longer intervention study is needed to assess the impact of longer term treatment

References
[1]Debono et al JCEM 2014 99: 4462 – 4470
[2]Di Dalmazi et al Lancet Diabetes and Endocrinology 2014 2: 396 – 405
[3]Debono et al JCEM 2016 101: 1469 - 1477

This study was funded by HRA Pharma and the National Institute for Health Research

ECE 2016 28--GP
Adrenal 3
Miguel Debono
Poster presented at: ECE2016 28 – 31 May 2016 MUNICH, GERMANY 18th EUROPEAN CONGRESS OF ENDOCRINOLOGY
PosterSessionOnline.com