Circadian variation in serum cortisol during hydrocortisone replacement is not attributable to changes in cortisol binding globulin

Gunganah K[1], Chung T[1] ,Monson J.P[1], Drake W.M[1]
1. St Bartholomew's hospital London UK

Introduction

Patients taking hydrocortisone (HC) replacement for primary or secondary adrenal failure require individual adjustment of their dose. Previous observations in our department suggest that total serum cortisol levels achieved following an afternoon or evening hydrocortisone dose of 5mg are almost as high as those that result from a 10mg dose in the early morning; and that the 'area under the cortisol curve' (AUC) generated by an evening 5mg dose is broader than after 10mg taken early in the morning .One potential explanation for this phenomenon might be a circadian variation in serum cortisol binding globulin (CBG) concentration, which may, in turn, impact on the interpretation of cortisol profiles and individual dose selection for patients on hydrocortisone replacement therapy. The purpose of this study was to investigate the hypothesis that there is a circadian variation in CBG levels.

Methods

- Single centred, non-therapeutic, clinical observational study
- 34 patients divided into 3 groups:
 - 10 patients with non-somatotroph structural pituitary disease on HC replacement
 - 11 patients with treated acromegaly on HC replacement
 - 13 patients with treated acromegaly not on HC replacement (control group)
- 10 healthy volunteers (control group)
- Cortisol and CBG levels were measured at 6 timepoints (8am, 11am, 1pm, 3pm, 5pm and 7pm)
- Serum cortisol was measured by electrochemiluminescent immunoassay on Modular Analytics E170 platform (Roche Diagnostics, Burgess Hill, UK).
- All measurements of CBG were performed in a single assay run using a radioimmunoassay using the Transcortin RIA CBG kit from Immunodiagnostics systems Limited (Boldon, UK).
- The protocol received institutional board review and all participants gave informed, written consent

Results

			Hormone Deficiencies				
Diagnosis	Number of patients	Median age (range)	ACTH	GH	TSH	LH/FSH	ADH
NFPA/ cystic lesions on HC replacement	10	50.73 (18 -73)	10	7	10	9	3
Treated acromegaly on HC replacement	11	61 (44-74)	11	2	10	9	2
Treated acromegaly not on HC	13	47 (31 - 71)	0	0	0	1	0
Healthy controls	10	30 (21 -47)	0	0	0	0	0

Table 1: Demographics of subjects

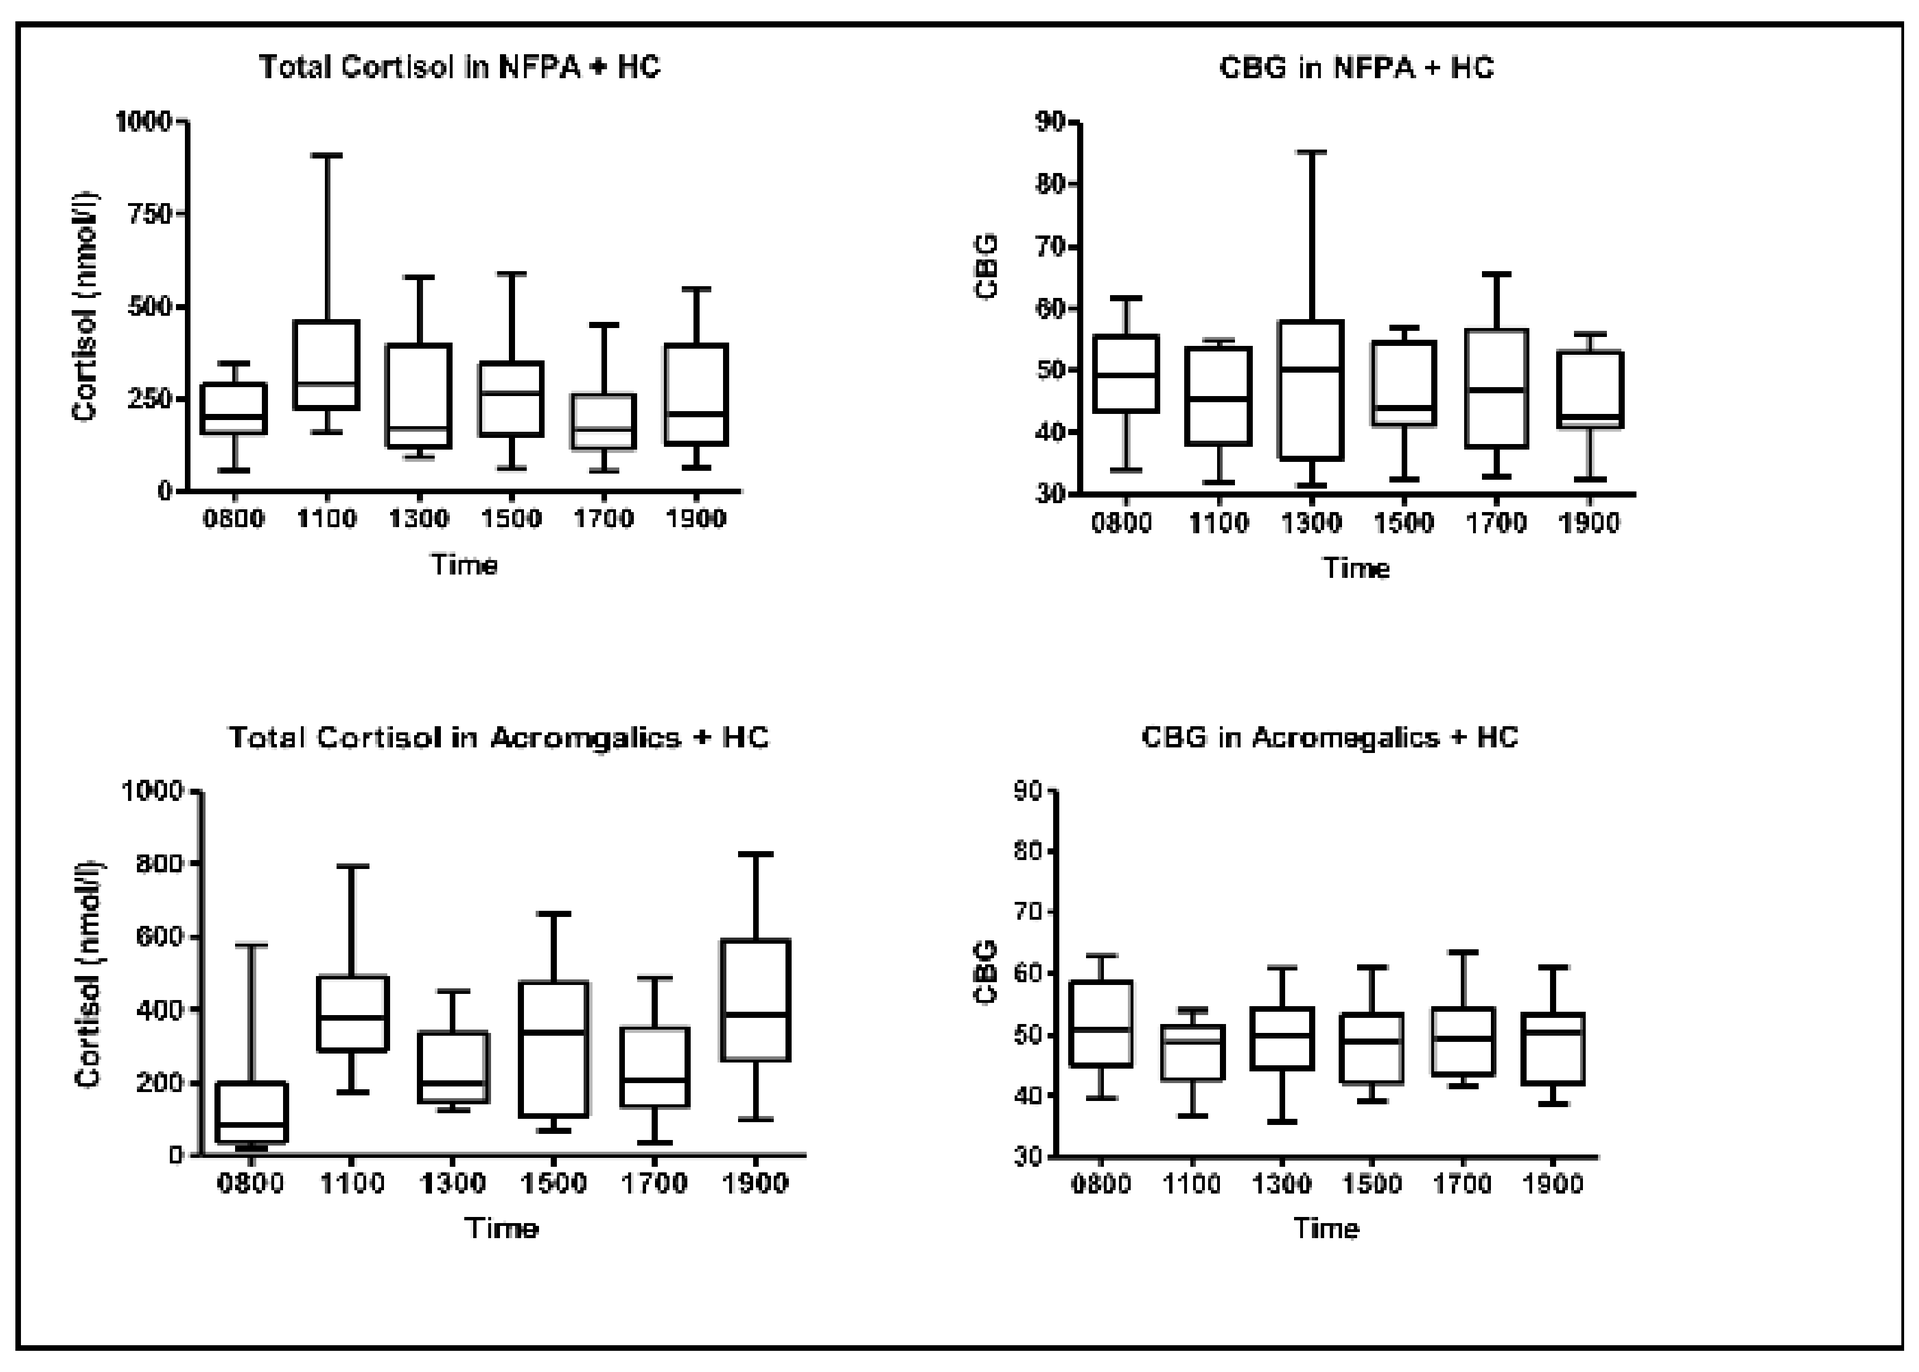
Total Cortisol in NFPA + HC
Cortisol (nmol/l)
CBG in NFPA + HC
CBG
Total Cortisol in Acromgalics + HC
CBG in Acromegalics + HC
0800 1100 1300 1500 1700 1900
Time

Figure 2a. Results for the 2 groups on HC replacement

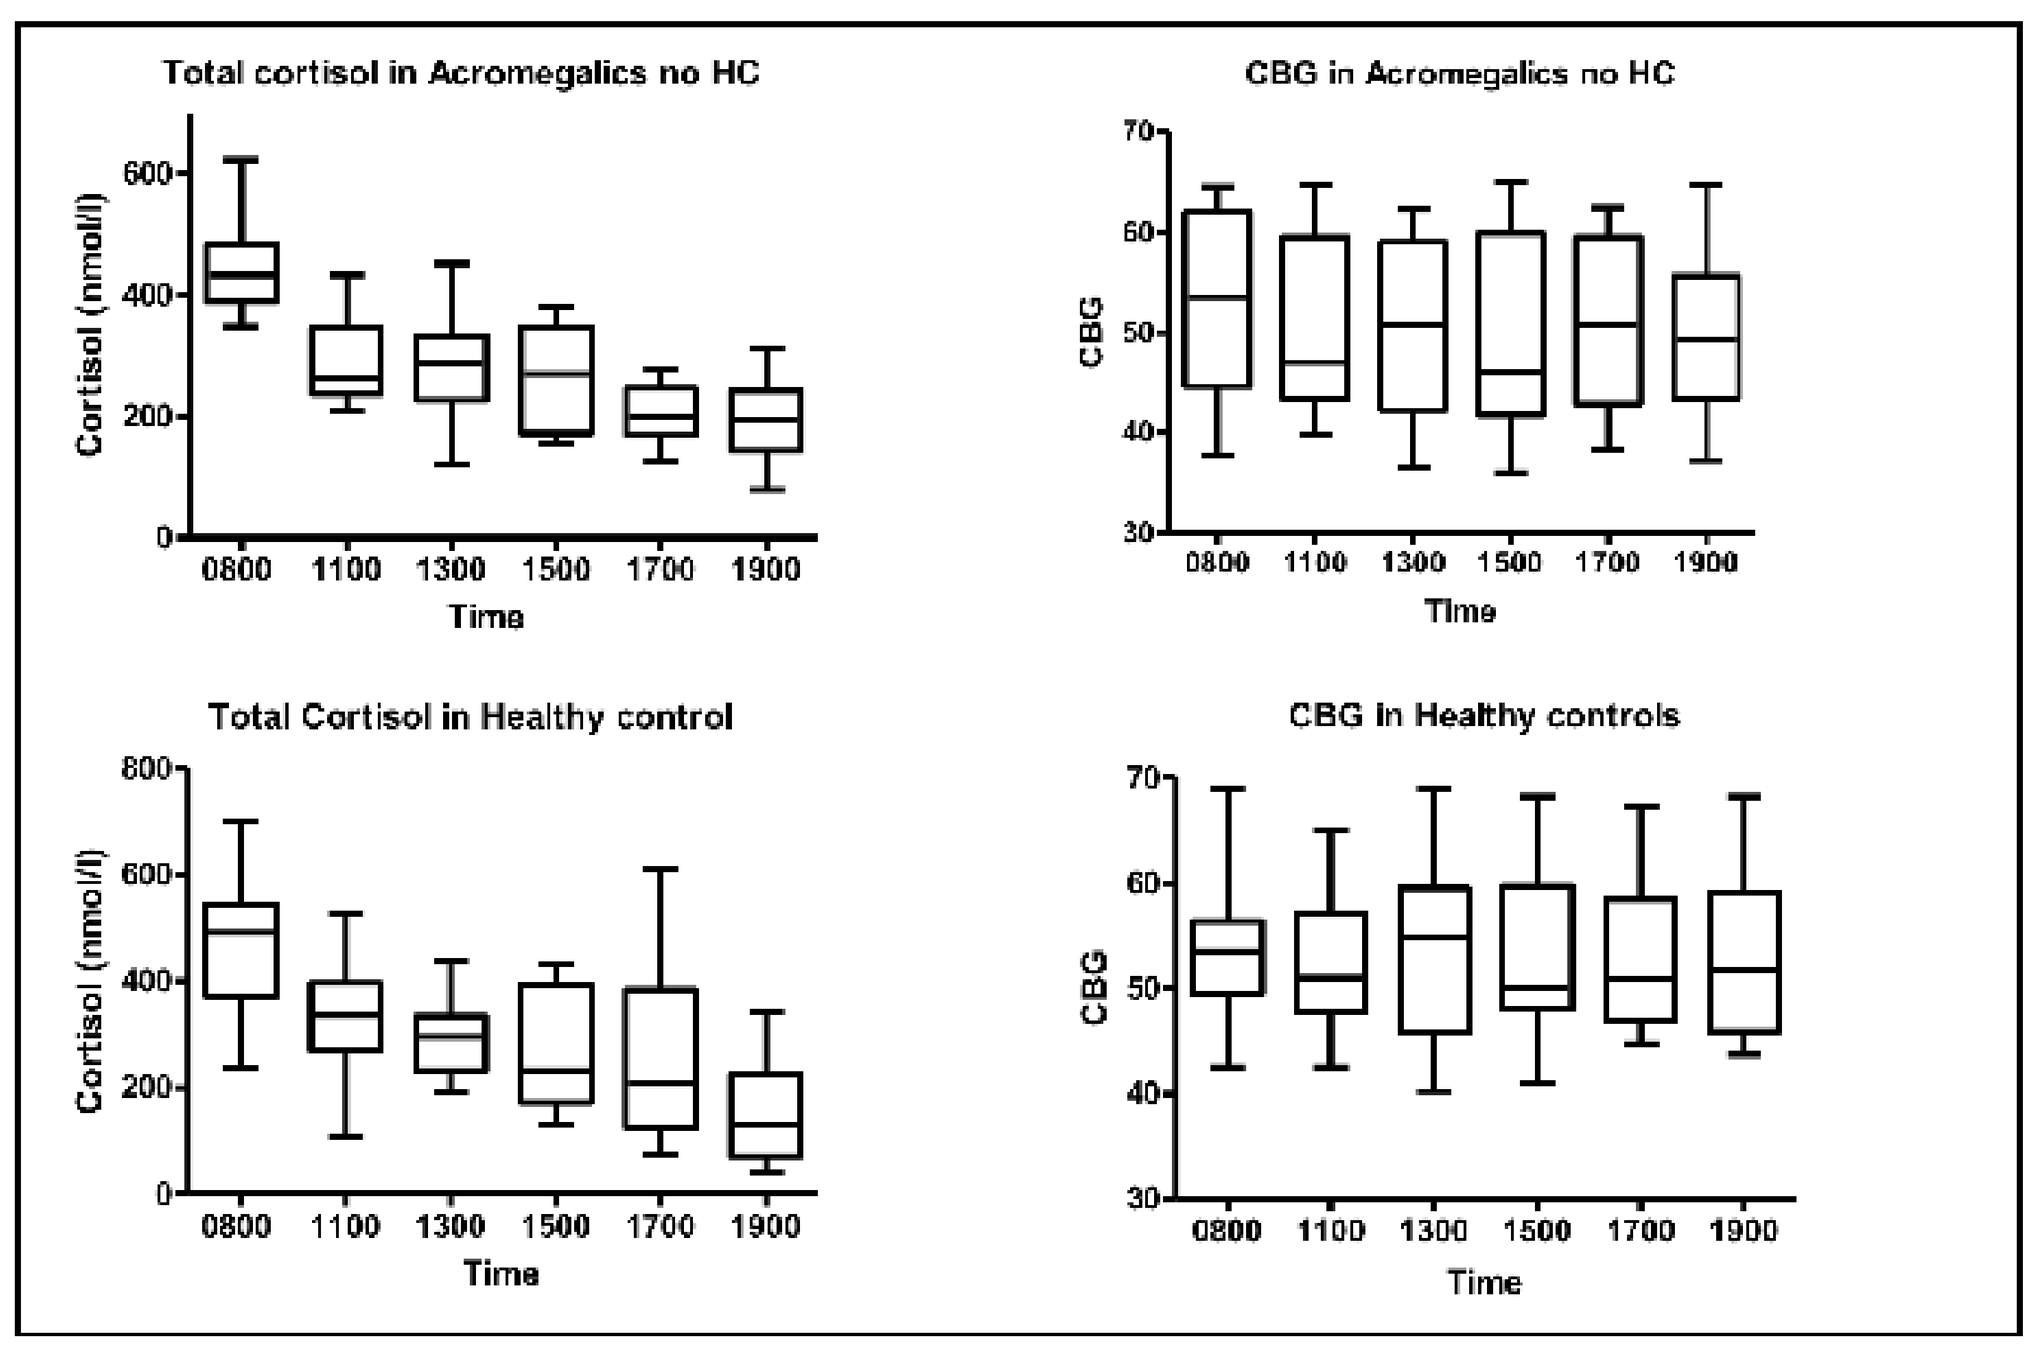
Total cortisol in Acromegalics no HC
Cortisol (nmol/l)
CBG in Acromegalics no HC
CBG
Total Cortisol in Healthy control
CBG in Healthy controls
0800 1100 1300 1500 1700 1900
Time

Figure 2b. Results for the control group (Acromegaly not on HC and healthy controls)

The demographics of each group are summarised in Table 1. Figure 2a and b show the Cortisol and CBG levels at the various time points for all 4 groups (patients and healthy volunteers). Baseline CBG levels were similar in all 4 groups with no statistically significant differences observed. No significant circadian variation in CBG concentration was found in any of the 4 groups.

Conclusion

No statistically significant variation in CBG levels was observed during the day in all patients on hydrocortisone replacement or our control population. Therefore, variation in serum cortisol levels during the day in patients on hydrocortisone replacement cannot be attributed to a circadian variation in CBG. Alternatively, such changes in serum cortisol may be explained by other factors including 11 ®-hydroxysteroid dehydrogenase type 1 activity or circadian changes in the binding capacity of CBG. As novel steroid replacement therapies including once daily or twice daily modified release hydrocortisone (Johannsson et al, 2014; Debono and Ross 2013) or continuous subcutaneous hydrocortisone by pump (Russell et al 2014) become readily available and in the absence of a biomarker for cortisol activity, better understanding of the biology of other factors which may determine serum cortisol concentrations, including CBG and 11 ®-hydroxysteroid dehydrogenase 1, remains important in order to replicate physiological conditions with glucocorticoid replacement.

References

1. Debono, M., Ross, R.J. (2013). What is the best approach to tailoring hydrocortisone dose to meet the patient needs in 2012? Clinical Endocrinology. 78:659-664
2. Johannsson G, Bergthorsdottir R, Nilsson AG, et al., Improving glucocorticoid replacement therapy using a novel modified- release hydrocortisone tablet: a pharmacokinetic study, *Eur J Endocrinol*, 2009;161:119–30.
3. .Russell, G., Durant, C., Ataya, A., Papastathi, C., Bhake, R., Woltersdorf, W., Lightman, S. (2014). Subcutaneous pulsatile glucocorticoid replacement therapy. Clinical Endocrinology. 81(2) ; pp 289-293

Acknowledgement

The expert statistical assistance of Mr JP Bestwick is gratefully acknowledged

This study was supported by an unrestricted investigator-initiated grant from Pfizer inc.

Barts Health NHS
NHS Trust

17 ECE
8--GP-04
Steroids, developmental and paediatric endocrinology
Kirun Gunganah
DOI: 10.3252/pso.eu.17ece.2015
Poster presented at: ECE2015 16-20 May 2015 DUBLIN, IRELAND 17th EUROPEAN CONGRESS OF ENDOCRINOLOGY
PosterSessionOnline.com